Sotatercept: Initial Signal-Seeking Quantitative Computed Tomography Results for Bone Mass and Vascular Calcification in Hemodialysis Subjects Treated With Escalating Doses: Interim Analysis of ACE-011-REN-001

Hartmut Malluche[1]; Keith Hruska[2]; Hem N. Singh[3]; William T. Smith[3]

[1]University of Kentucky, Lexington, KY; [2]Washington University St Louis, St Louis, MO; [3]Celgene Corporation, Warren, NJ

INTRODUCTION

- Renal osteodystrophy (ROD) and vascular calcification (VC) are integral components of chronic kidney disease-mineral/bone disorder (CKD-MBD). CKD-MBD is strongly associated with unacceptable morbidity and mortality in end-stage kidney disease (ESKD).[1]
- Activin A is a TGF-β superfamily protein that is found at high levels in bone; its signaling is through the type II activin A receptor (ActRIIA).[2-4] Inhibin, an inhibitor of activin A, is produced in the ovaries and stimulates bone growth.[2,5] Decreased inhibin expression is associated with post-menopausal bone loss.[5]
 - Non-clinical studies suggest that RAP-011 (the murine ActRIIA-IgG1 ligand trap that inhibits ActRIIA signaling) modulates the balance between bone formation and bone resorption activity by blocking signaling through the ActRIIA receptor.[2,6,7]
 - In a 5/6 nephrectomy mouse model of CKD that exhibits bone loss, bone mass measurements were significantly improved with 8 weeks of RAP-011 treatment, compared with the control mice.[6]
- In a mouse model of *ldlr*-/- high fat fed, 5/6 nephrectomy for VC, RAP-011 inhibited bone resorption and increased bone volume. RAP-011 also inhibited Smad-dependent signaling, blocked aortic osteoblastic transition, increased vascular smooth muscle protein levels, and decreased CKD-stimulated VC.[8] These effects appear to be mediated by RAP-011 inhibition of CKD-stimulated endothelial to mesenchymal transition.[9]
- Sotatercept (ACE-011), a human ActRIIA-IgG1 ligand trap that inhibits ActRIIA signaling, improved total hip bone mineral density (BMD) with a 2.4% increase compared with a 0.7% decrease with placebo after 4 months in a post-menopausal healthy volunteer clinical trial.[10]
- Sotatercept is being studied for the correction of anemia and improvement of measures of CKD-MBD in hemodialysis (HD) subjects. Sotatercept blocks activin A signaling and may reduce osteoclastogenesis, promote osteoblast maturation in bone, and reduce arterial osteoblastic transition.
- The current exploratory analysis in HD subjects evaluated the effect of sotatercept on BMD and VC using quantitative computed tomography (QCT).[11-13]
- Enrollment in the subcutaneous (SC) sotatercept 0.7 mg/kg dose group is ongoing. Interim results for pharmacokinetic (PK), safety, and hemoglobin (Hb) effects are presented elsewhere.[14]

METHODS

- In an ongoing study of sotatercept in HD subjects for the correction of anemia, subjects responding to an erythropoietin-stimulating agent (ESA) were washed out of their ESA effect until Hb was <10 g/dL, and then randomized (3:1) to receive escalating dose levels of SC sotatercept (0.3 mg/kg, 0.5 mg/kg, or 0.7 mg/kg) or placebo every 28 days for up to 8 dose cycles. The study design and dosing are described in detail elsewhere.[14] Treatment failures (Hb <9 g/dL) were treated with ESA and/or red blood cell transfusion (rescue therapy).
- QCT of the hip, lumbar spine, and abdominal aorta was obtained at baseline and after the 225-day treatment phase.
 - Trabecular volumetric BMD (vBMD) (mg/cm^3) was determined for 2 vertebrae within L1-4 (typically L1-2).
 - The left proximal femurs were analyzed for vBMD of the cortical, trabecular, and integral bone compartments of the total hip, femoral neck, and trochanteric regions.
 - VC of the abdominal aorta was assessed using proprietary software that semi-automatically segmented the area and volume of calcifications within the region adjacent to the top of L1 through the bottom of L4. The number and location of slices was maintained across visits per subject. Agatston and square root transformed volumetric scores were determined as described elsewhere.[11,12] Lower total Agatston and square root transformed total volume scores (mm^3) indicate lower levels of VC.
 - All image quality control and blinded analyses were performed centrally by PAREXEL imaging (PAREXEL International Corp, Waltham, MA).
- Biomarkers, including bone-specific alkaline phosphatase (BSAP), procollagen type 1 N-terminal propeptide (P1NP), and C-terminal type 1 collagen telopeptide (CTX), were measured at baseline and after dose cycles 1, 3, 5, and 7.

RESULTS

Subjects

- A total of 31 subjects were randomized and received ≥1 dose of study medication (**Table 1**).

Table 1. Randomized Subjects and QCT Analysis Subset

Subjects, n	Placebo	Sotatercept 0.3 mg/kg	Sotatercept 0.5 mg/kg	Sotatercept 0.7 mg/kg
Randomized and received ≥1 dose of study medication	8	9	8	6
QCT measures at baseline and Day 225	3	6	5	2

- The subject disposition is shown in **Figure 1**, including the subjects with paired QCT measurements at baseline and Day 225.
 - Most subjects who were discontinued from study treatment had treatment failure requiring rescue (generally because of Hb <9 g/dL; most common in the placebo and sotatercept 0.3 mg/kg groups); no subject discontinued treatment because of an adverse event (AE).
 - Of the 16 subjects with paired QCT measurements, 9 required rescue therapy during the treatment phase, 8 of whom required rescue within the first 3 dose cycles.
- Among subjects with paired QCT measurements, baseline demographic and clinical characteristics were generally similar across treatment groups (**Table 2**); however, there was a substantially longer time on dialysis in the placebo group, which was also the youngest group. There were also differences between groups in baseline biomarker and Agatston scores (**Table 3**).

Figure 1. Subject Disposition

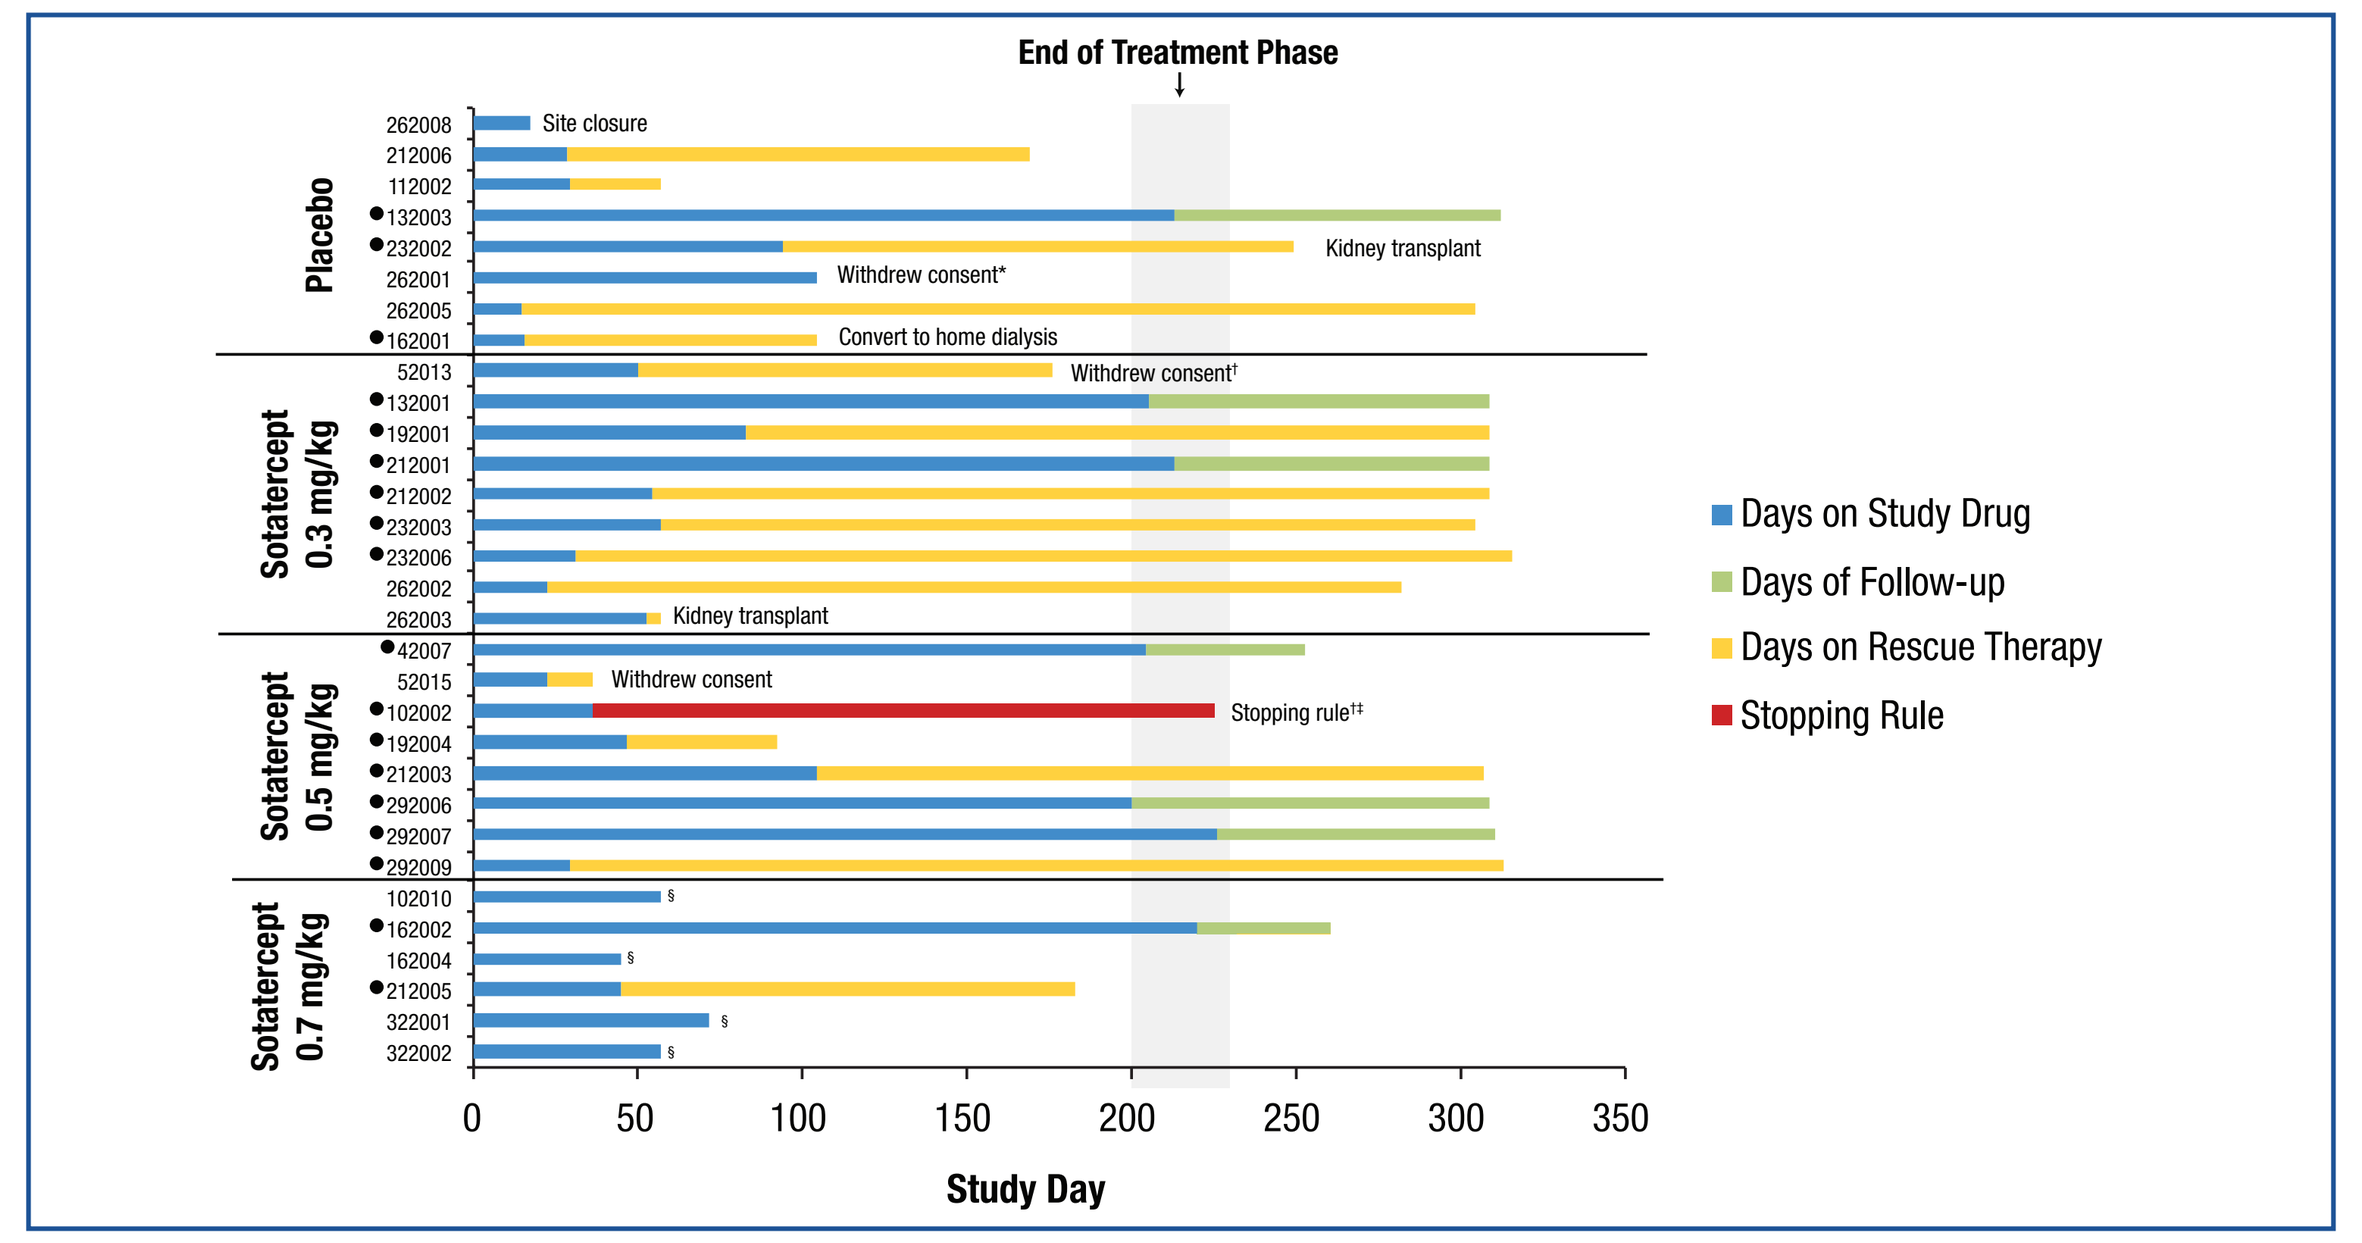
End of Treatment Phase
Placebo
Sotatercept 0.3 mg/kg
Sotatercept 0.5 mg/kg
Sotatercept 0.7 mg/kg
262008
212006
112002
132003
232002
262001
262005
162001
52013
132001
192001
212001
212002
232003
232006
262002
262003
42007
52015
102002
192004
212003
292006
292007
292009
102010
162002
162004
212005
322001
322002
Site closure
Kidney transplant
Withdrew consent*
Convert to home dialysis
Withdrew consent†
Kidney transplant
Withdrew consent
Stopping rule†‡
§
Days on Study Drug
Days of Follow-up
Days on Rescue Therapy
Stopping Rule
0
50
100
150
200
250
300
350
Study Day

Note: "•" indicates the subjects with paired QCT measurements at baseline and Day 225.
*This subject receiving placebo had elevated serum erythropoietin levels, suggesting off-protocol erythropoietin administration.
†Protocol violation.
‡Subject met stopping rule criteria for elevated blood pressure on Day 29; study treatment was discontinued, and rescue therapy was administered on Day 36, with continued follow-up.
Subject was randomized in error with non-qualifying blood pressure, based on incomplete evaluation, at baseline.
§Subject remains in the study, still ongoing.

Table 2. Baseline Demographic and Clinical Characteristics of Subjects With Paired QCT Measurements

	Placebo n=3	Sotatercept 0.3 mg/kg n=6	Sotatercept 0.5 mg/kg n=5	Sotatercept 0.7 mg/kg n=2
Age, mean, years	54.0	57.3	60.8	69.0
Female, n (%)	0 (0.0)	4 (66.7)	0 (0.0)	1 (50.0)
Race, n (%)				
White	1 (33.0)	3 (50.0)	3 (60.0)	1 (50.0)
Black	1 (33.0)	3 (50.0)	2 (40.0)	1 (50.0)
Asian	1 (33.0)	0 (0.0)	0 (0.0)	0 (0.0)
Ethnicity, n (%)				
Hispanic	0 (0.0)	2 (33.0)	3 (60.0)	1 (50.0)
Non-Hispanic	3 (100.0)	4 (66.7)	2 (40.0)	1 (50.0)
Postdialysis weight, mean, kg	70.4	80.3	79.4	84.4
Body mass index, mean, kg/m^2	25.4	27.7	26.9	29.2
Diabetes, n (%)	2 (66.7)	5 (83.3)	5 (100.0)	2 (100.0)
Parathyroidectomy, n (%)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)
Time on dialysis, mean, months	101.2	43.3	22.4	11.3
Non-calcium phosphate binder, n (%)*	3 (100.0)	3 (50.0)	3 (60.0)	0 (0.0)
Calcium-based phosphate binder, n (%)*	1 (33.3)	4 (66.7)	4 (80.0)	2 (100.0)
Calcimimetic, n (%)*	1 (33.3)	2 (33.3)	0 (0.0)	0 (0.0)
1,25-OH vitamin D analog, n (%)	2 (66.7)	5 (83.3)	2 (40.0)	2 (100.0)

*Subjects could be receiving multiple types of binders.

Table 3. Mean Baseline Biomarker, Volumetric BMD, and Agatston Scores of Subjects With Paired QCT Measurements

	Placebo n=3	Sotatercept 0.3 mg/kg n=6	Sotatercept 0.5 mg/kg n=5	Sotatercept 0.7 mg/kg n=2
Baseline whole PTH, pg/mL	261.4	104.8	135.5	100.4
BSAP*, μg/L	28.3	18.0	13.2	7.5
P1NP†, ng/mL	623.7	376.2	468.4	308.0
CTX‡, pg/mL	3,417.7	2,062.5	2,266.8	1,377.0
Femoral neck cortical BMD, mg/cm^3	653.1	666.9	593.1	566.5
Lumbar spine (L1, L2) trabecular BMD, mg/cm^3	123.1	125.6	149.1	150.6
VC total Agatston score§	8,665.0	9,472.7	3,618.5	823.2

PTH=parathyroid hormone.
*Bone-specific alkaline phosphatase (BSAP) reference range: males, 6-30; females (premenopausal), 3-19; females (postmenopausal), 6-26.
†Procollagen type 1 N-propeptide (P1NP) reference range: males, 30-110; females, 20-108.
‡C-terminal telopeptide (CTX) reference range: males, 0-854; females (premenopausal), 26-573; females (postmenopausal), 104-1,008.
§Lower Agatston scores indicate lower levels of VC.

Safety Assessments

- The safety findings from this study are described in detail elsewhere.[14]
- Serious AEs were generally considered unrelated to the study drug, did not lead to discontinuation, and resolved with continued therapy. No deaths were reported in the sotatercept groups.
- AEs were mostly mild or moderate in severity, unrelated to the study drug, relatively similar between treatment groups, and generally consistent with subjects' medical histories.
- In the first 28-day dose cycle and during the 225-day treatment phase, home blood pressure measurements showed no consistent or dose-dependent changes from baseline among subjects in any of the treatment groups.

QCT Volumetric BMD Measurements

- **Table 4** provides the percent change from baseline in femoral neck cortical, hip cortical, and lumbar spine trabecular volumetric BMD measurements.

Table 4. Baseline and Percent Change From Baseline in Femoral Neck Cortical, Total Hip Cortical, and Lumbar Spine Trabecular Volumetric BMD

	Placebo n=3	Sotatercept 0.3 mg/kg n=6	Sotatercept 0.5 mg/kg n=5	Sotatercept 0.7 mg/kg n=2
Femoral neck cortical				
Baseline BMD, mg/cm^3	653.1	666.9	593.1	565.5
% Change from baseline BMD (n)	–0.9 (3)	–1.4 (5)	1.6 (5)	3.0 (1)
Total hip cortical				
Baseline BMD, mg/cm^3	708.6	668.4	647.3	635.6
% Change from baseline BMD (n)	–0.1 (3)	–1.1 (5)	0.5 (5)	2.7 (1)
Mean lumbar spine (L1, L2) trabecular				
Baseline BMD, mg/cm^3	123.1	125.6	149.1	150 .6
% Change from baseline BMD (n)	12.6 (3)	8.0 (6)	0.5 (5)	–2.7 (2)

- Proportions of subjects with >2% loss, >2% gain, or no change in cortical BMD were determined[15]; sotatercept treatment is associated with a dose-dependent increase in the proportion of subjects with a ≥2% increase in femoral neck cortical bone (**Table 4** and **Figure 2A**).
- In high-turnover ROD, trabecular bone mass increases, as measured by lumbar spine trabecular BMD, without a reduction in vertebral fracture rates in ESKD compared with the general population.[13,16] In this setting, the increased trabecular bone mass is of poor quality.[17] Sotatercept appears to prevent the increase in trabecular bone mass in the lumbar spine (**Table 4** and **Figure 2B**).

Figure 2. Change in Volumetric BMD in (A) Femoral Neck Cortical Bone and (B) Lumbar Spine Trabecular Bone

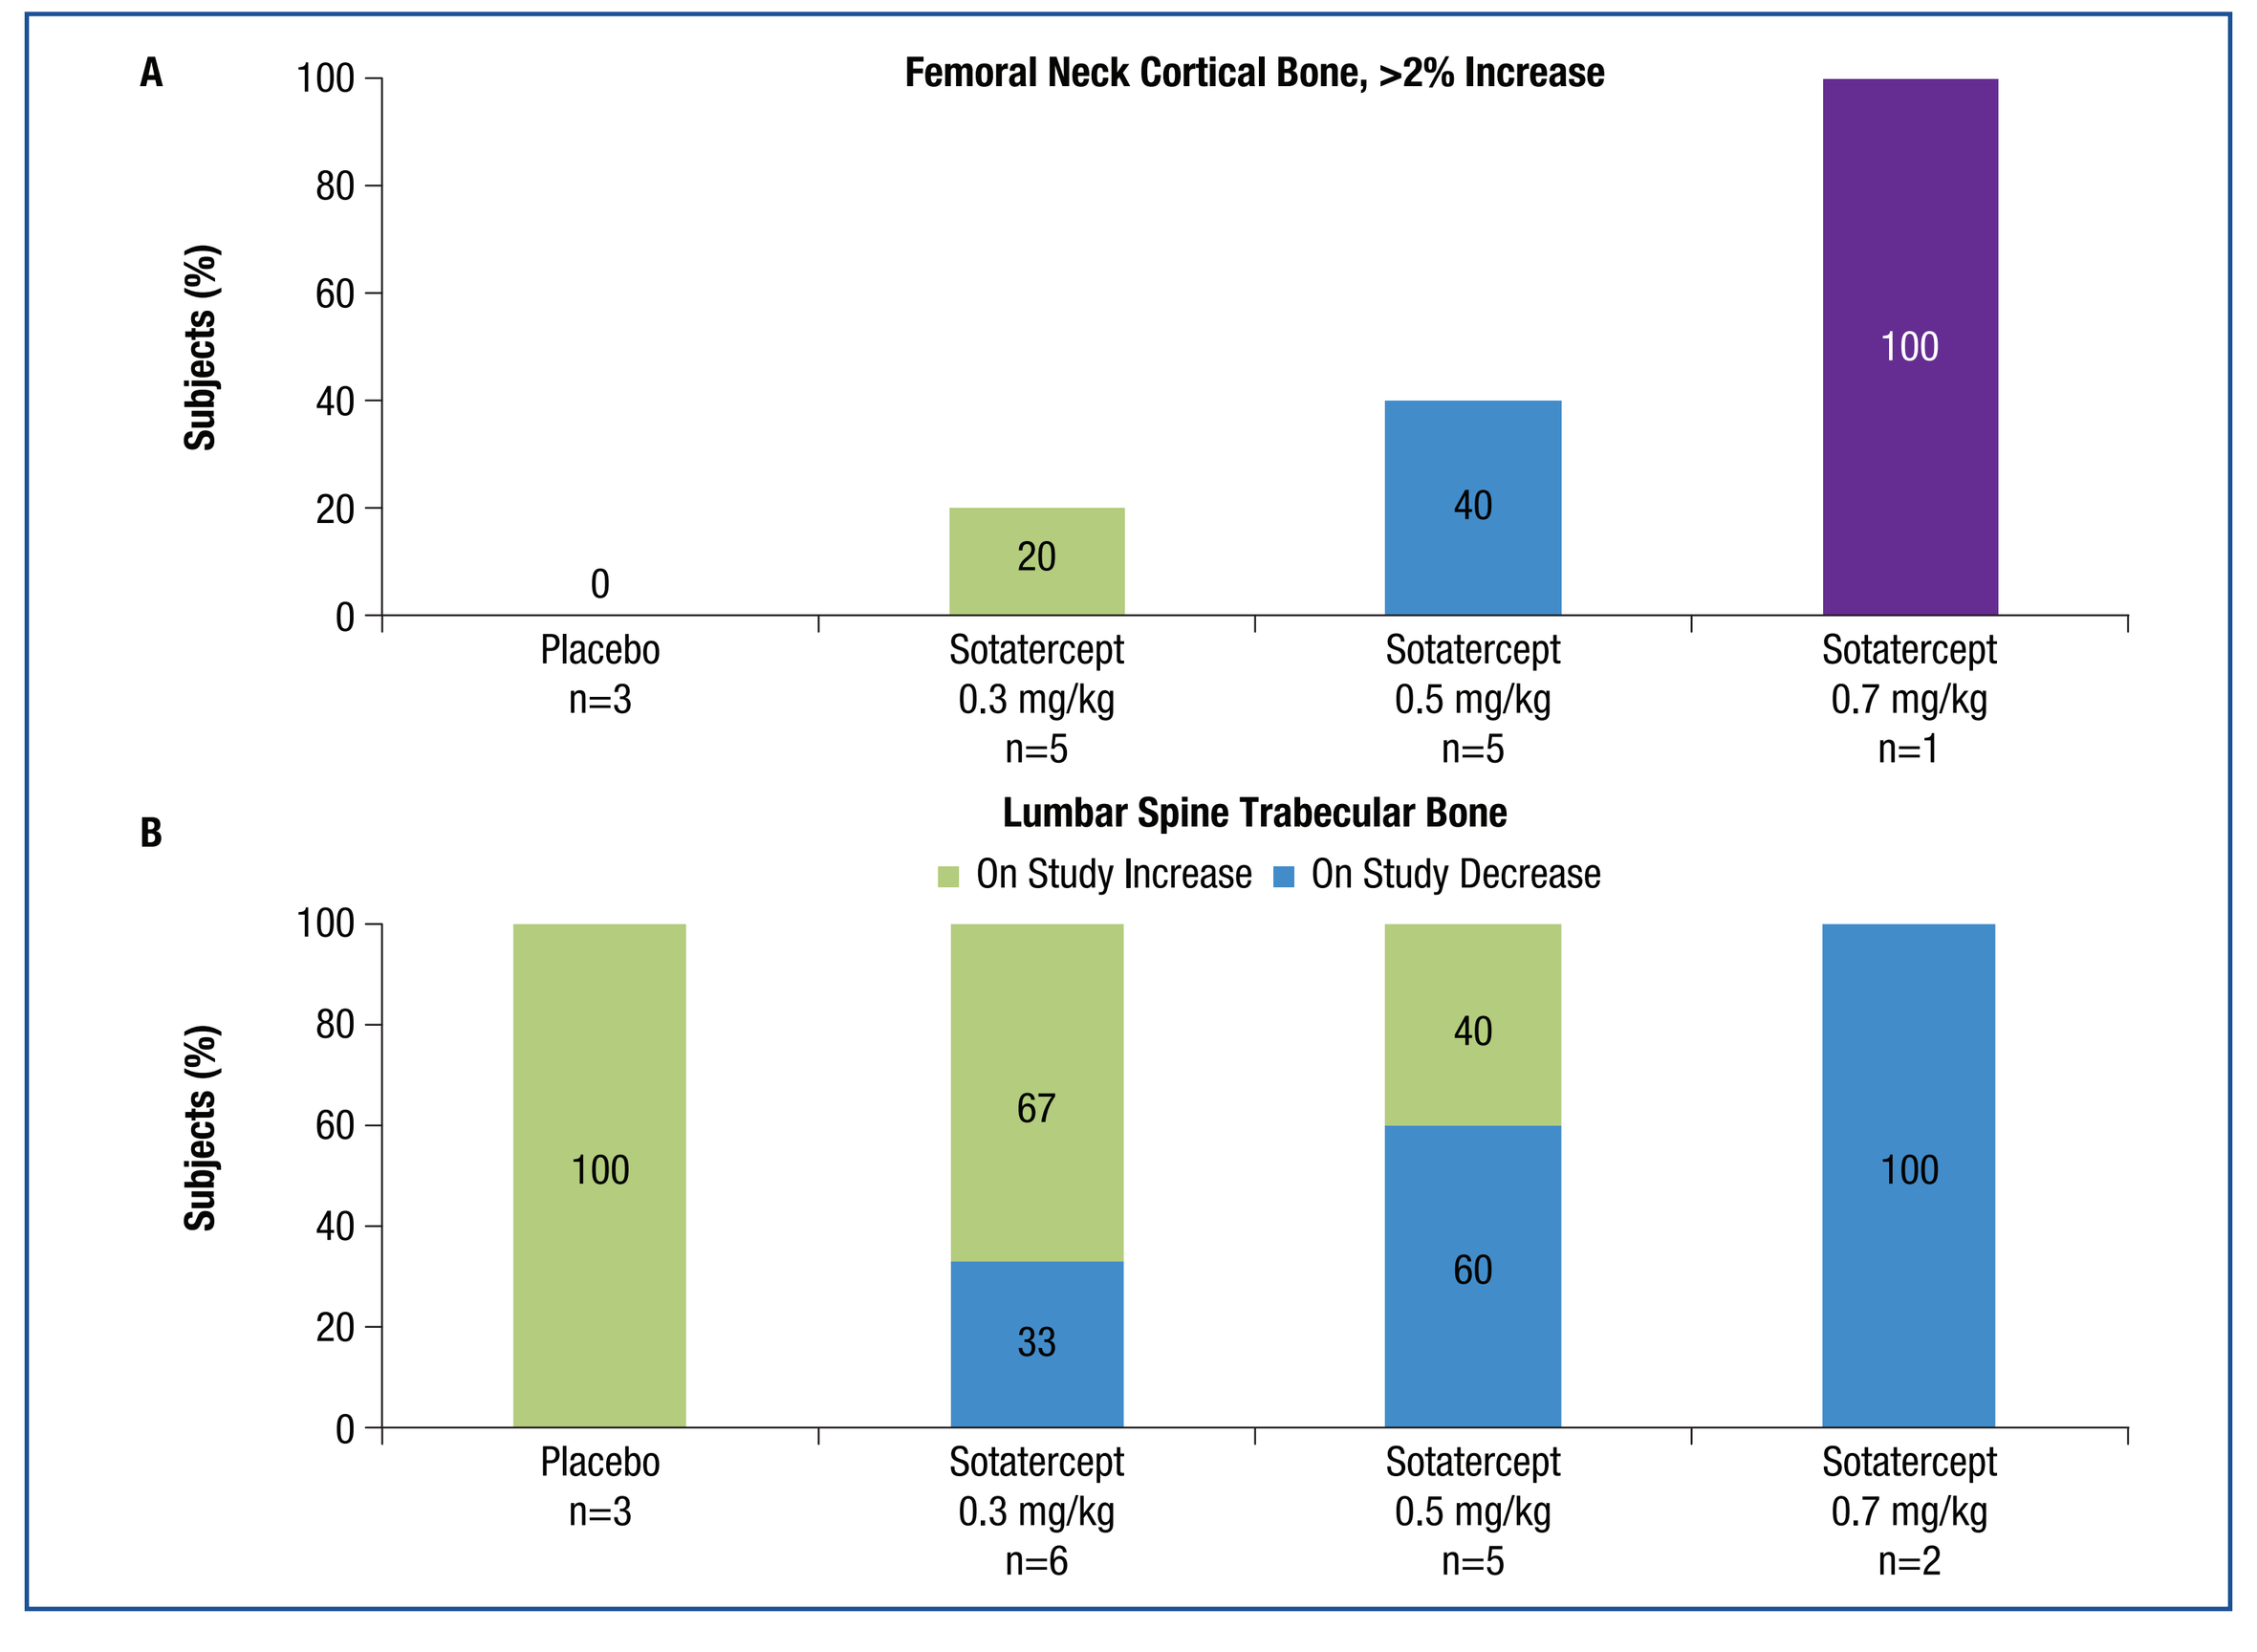
A
Femoral Neck Cortical Bone, >2% Increase
Subjects (%)
0
20
40
100
Placebo n=3
Sotatercept 0.3 mg/kg n=5
Sotatercept 0.5 mg/kg n=5
Sotatercept 0.7 mg/kg n=1
B
Lumbar Spine Trabecular Bone
On Study Increase
On Study Decrease
Subjects (%)
100
67
33
40
60
100
Placebo n=3
Sotatercept 0.3 mg/kg n=6
Sotatercept 0.5 mg/kg n=5
Sotatercept 0.7 mg/kg n=2

QCT VC Measurements

- The changes from baseline in abdominal aorta total Agatston scores are provided in **Table 5**. Categories of change in the total Agatston score and square root transformed total volume score are shown in **Figure 3** and **Table 6**.

Table 5. Baseline and Change From Baseline in Abdominal Aorta Total Agatston Score and Square Root Transformed Total Volume Score*

	Placebo n=3	Sotatercept 0.3 mg/kg n=6	Sotatercept 0.5 mg/kg n=5	Sotatercept 0.7 mg/kg n=2
Abdominal aorta total Agatston score				
Baseline total Agatston score	8,665	9,473	3,619	823
Change from baseline total Agatston score	1,050.4	8,578.9	225.7	43.4
% Change from baseline total Agatston score	58.4	24.9	17.3	3.4
Square root transformed total volume score				
Baseline square root of total volume, mm^3	52.0	46.2	33.1	16.9
Change from baseline square root of total volume, mm^3	4.5	11.1	1.2	0.4

*Lower total Agatston and square root transformed total volume scores indicate lower levels of VC.

Figure 3. Categories of Change in (A) Abdominal Aorta Total Agatston Score and (B) Square Root Transformed Total Volume Score[18]

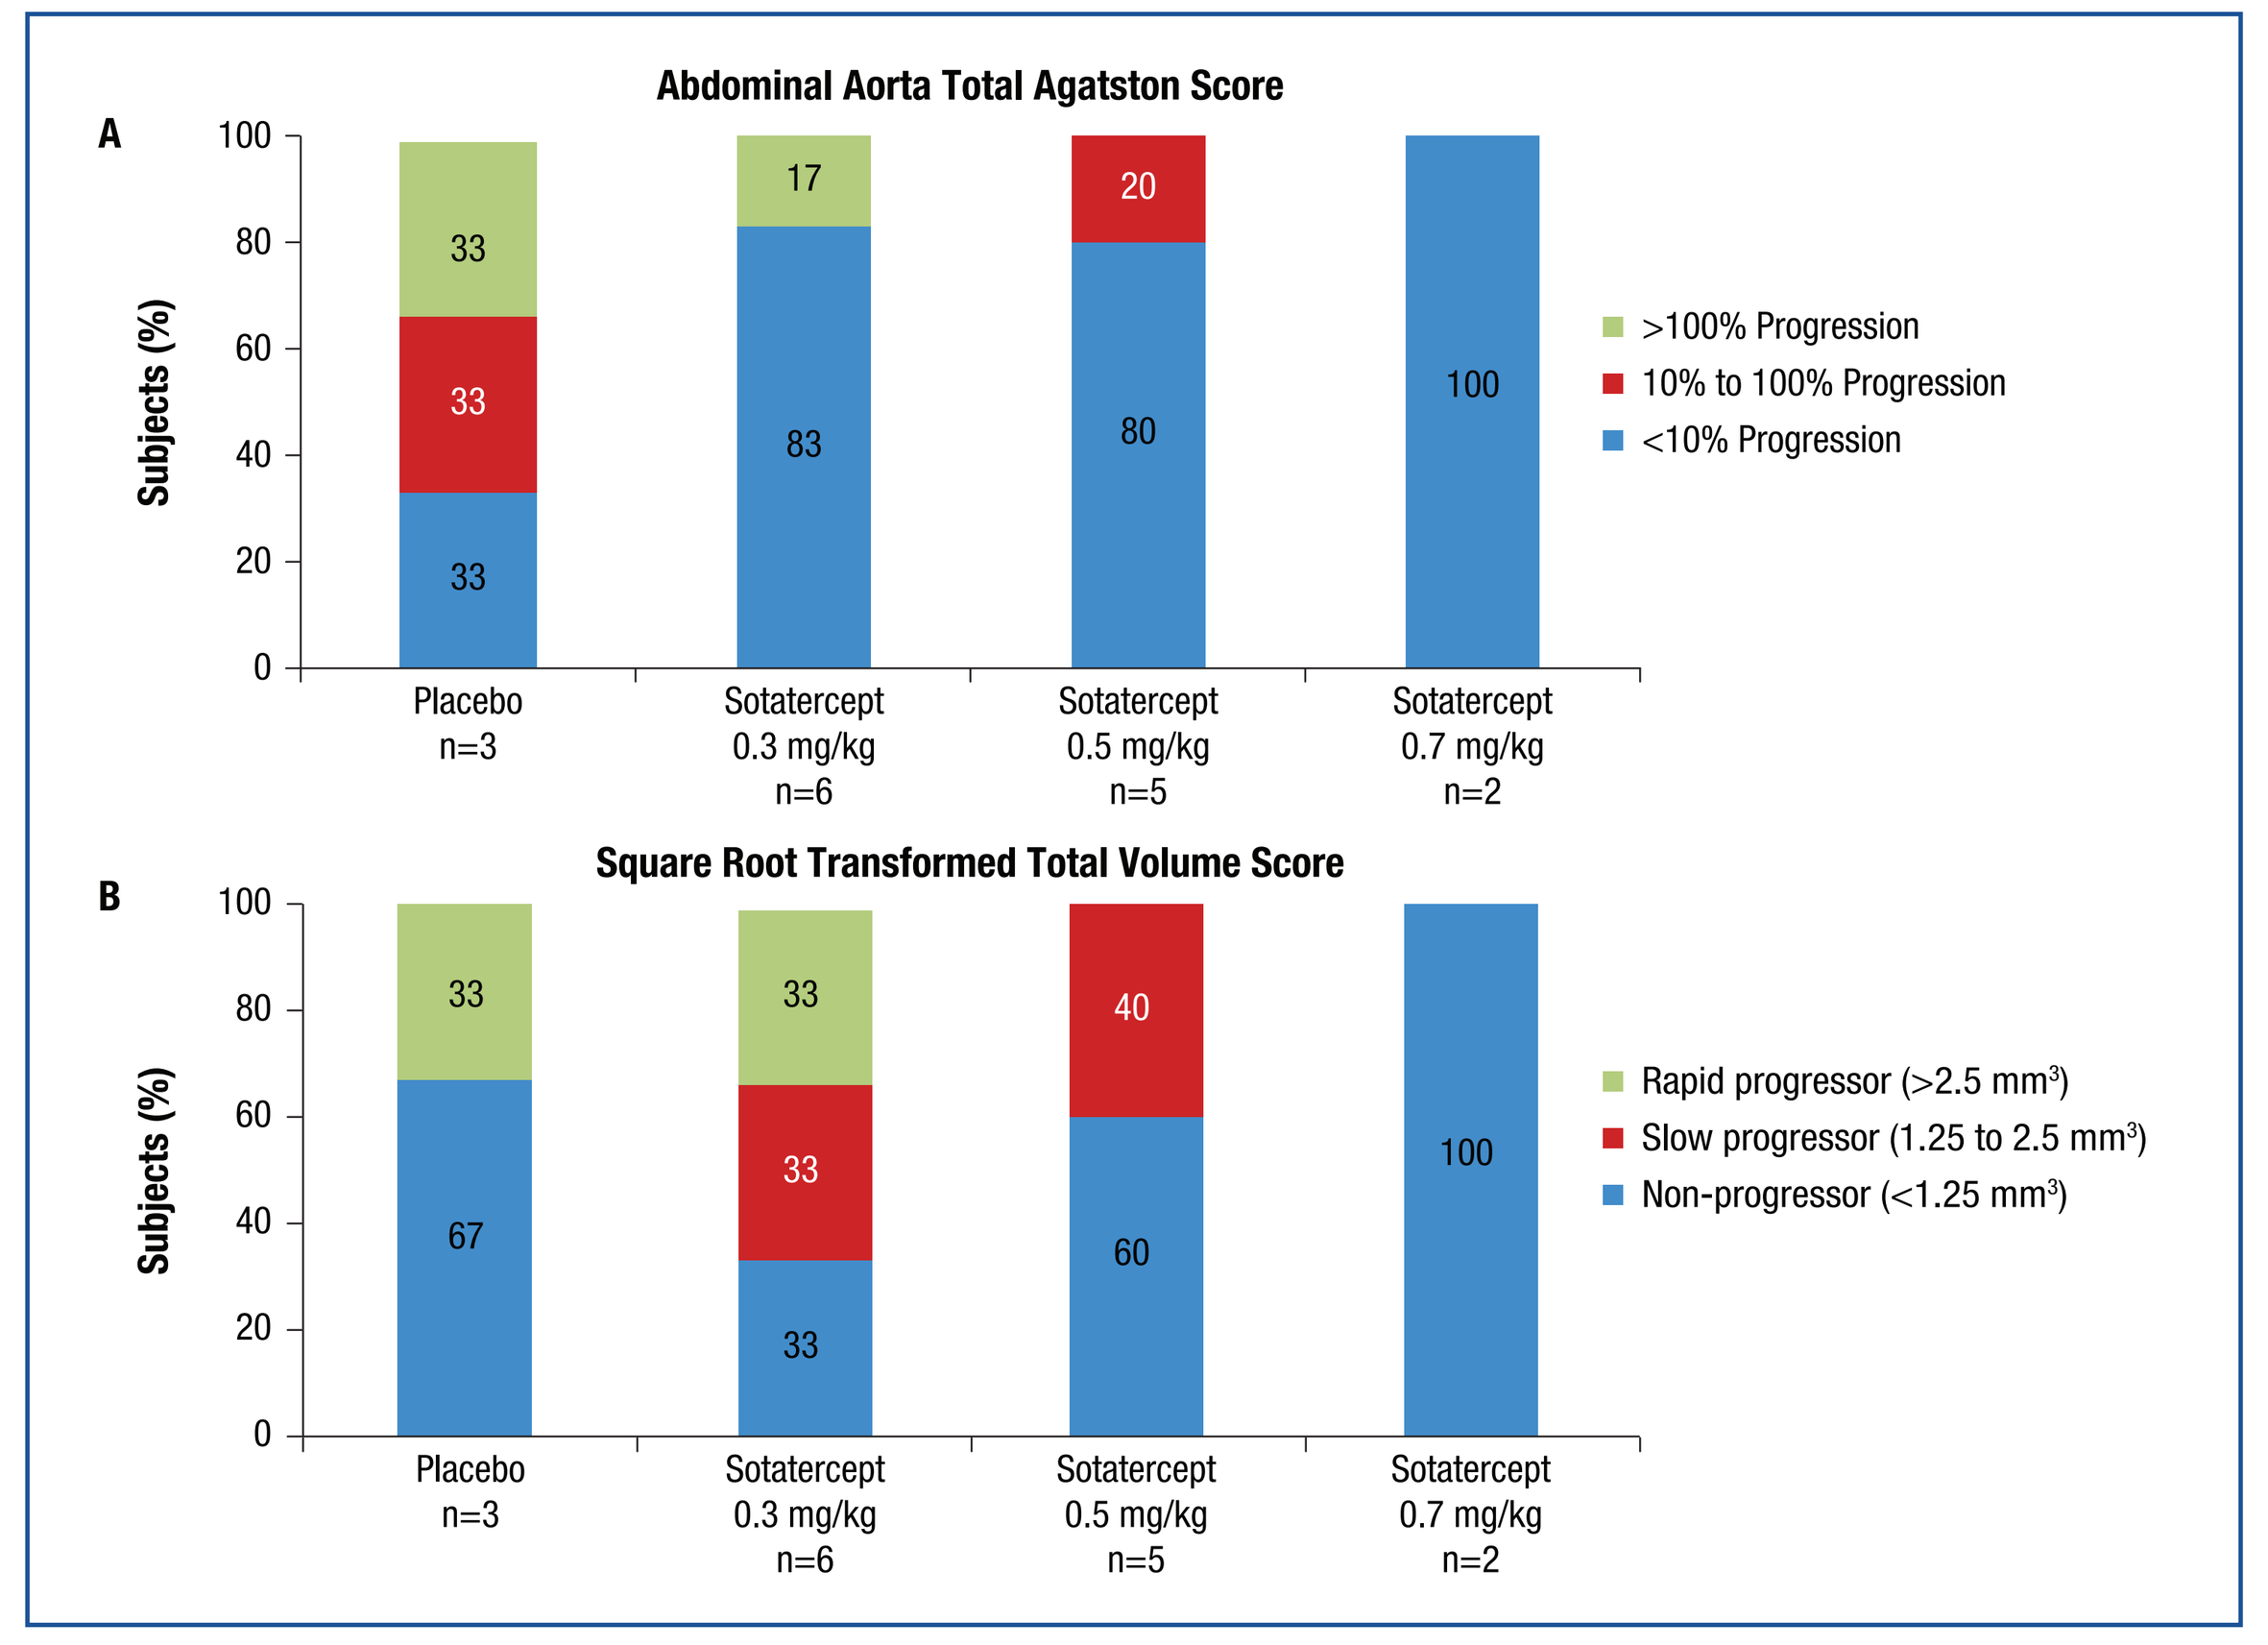
A
Abdominal Aorta Total Agatston Score
Subjects (%)
33
33
33
17
83
20
80
100
>100% Progression
10% to 100% Progression
<10% Progression
Placebo n=3
Sotatercept 0.3 mg/kg n=6
Sotatercept 0.5 mg/kg n=5
Sotatercept 0.7 mg/kg n=2
B
Square Root Transformed Total Volume Score
Subjects (%)
33
67
33
33
33
40
60
100
Rapid progressor (>2.5 mm³)
Slow progressor (1.25 to 2.5 mm³)
Non-progressor (<1.25 mm³)
Placebo n=3
Sotatercept 0.3 mg/kg n=6
Sotatercept 0.5 mg/kg n=5
Sotatercept 0.7 mg/kg n=2

- Eleven subjects had baseline Agatston scores that were distributed between 10 and 10,000 (72–9,833). Five subjects may be considered outliers at baseline (low: 0.0, 1.1, and 1.4; high: 21,033 and 44,356).
- An analysis was also performed excluding the outliers to determine if this influenced the dose-dependent trend in the Agatston scores (**Table 6**).

Table 6. Baseline and Change in Total Agatston Score Excluding 5 Outliers*

	Placebo n=1	Sotatercept 0.3 mg/kg n=4	Sotatercept 0.5 mg/kg n=4	Sotatercept 0.7 mg/kg n=2
Baseline total Agatston score, excluding 5 outliers	4,960	3,120	4,523	823
Change from baseline total Agatston score, excluding 5 outliers	2,711.3	287.7	282.0	43.4
% Change from baseline, excluding 5 outliers	54.7	9.0	5.0	3.4

*Lower total Agatston and square root transformed total volume scores indicate lower levels of VC.

Analysis of Subjects by Duration of Exposure to Sotatercept

- Subjects were analyzed by exposure status, regardless of dose level: sotatercept subjects receiving ≤3 doses (n=6) and sotatercept subjects receiving ≥4 doses (n=7). The placebo group (n=3) was unchanged.
- Other than gender (female 66.7% vs. 14.3% in the ≤3 doses and ≥4 doses groups, respectively) and Agatston score (**Table 7**), key baseline demographic and disease characteristics were similar between those receiving ≤3 doses of sotatercept and those receiving ≥4 doses.
- An exposure effect on VC remains intact in all sotatercept subjects (**Table 7**), as well as when excluding outliers (**Table 8**).

Table 7. Baseline and Change From Baseline in Total Agatston Score and Square Root Transformed Total Volume Score, by Sotatercept Exposure*

	Placebo n=3	Sotatercept ≤3 Doses n=6	Sotatercept ≥4 Doses n=7
Baseline total Agatston score	8,665	8,350	3,782
Change from baseline total Agatston score	1,050.4	8,461.4	274.3
% Change from baseline total Agatston score	58.4	33.7	5.8
Baseline square root of total volume, mm^3	52.0	38.2	35.3
Change from baseline square root of total volume, mm^3	4.5	10.6	1.4

*Lower total Agatston scores and square root transformed total volume scores indicate lower levels of VC.

Table 8. Baseline and Change From Baseline in Total Agatston Score, Excluding 5 Outliers, by Sotatercept Exposure*

	Placebo n=1	Sotatercept ≤3 Doses n=3	Sotatercept ≥4 Doses n=7
Baseline total Agatston score, excluding 5 outliers	4,960	1,914	3,782
Change from baseline total Agatston score, excluding 5 outliers	2,711.3	148.4	274.3
% Change from baseline, excluding 5 outliers	54.7	7.5	5.8

*Lower total Agatston scores and square root transformed total volume scores indicate lower levels of VC.

Bone Turnover Biomarkers

- The placebo group had the highest PTH, BSAP, P1NP, and CTX levels at baseline (**Table 3**).
- No consistent changes from baseline in phosphate, PTH, FGF-23, or sclerostin levels were observed, except that PTH was lower at all time points than at baseline in the placebo group (–15.2 to –100.7 ng/L), decreasing to a level similar to the other groups. For bone biomarkers, no clear changes were seen in percent change in the formation markers BSAP and P1NP.
- More subjects receiving sotatercept 0.5 mg/kg had meaningful, persistent decreases in the resorption marker CTX. There currently are not enough data available for the 0.7 mg/kg group to evaluate effects.

CONCLUSIONS

- Based on baseline biomarker data, subjects in this study tended to have high-turnover ROD. The placebo group had changes as expected for high turnover: decreased cortical bone mass, increased trabecular bone mass, and increased VC.
- In this small interim dataset evaluating exploratory QCT data measured at baseline and Day 225 (after up to eight 28-day dose cycles), there is an apparent dose effect on multiple parameters of CKD-MBD, including increasing femoral neck cortical bone mass, decreasing lumbar spine bone mass, and slowing progression of VC.
- Bone histomorphometry data are required to better characterize the effects on lumbar spine BMD.
- Changes in bone turnover biomarkers suggest that there may be a primarily anti-resorptive effect, most apparent with sotatercept 0.5 mg/kg, currently the highest dose evaluable for these markers. The findings of less VC and changes in bone biomarkers are consistent with the histologic findings in the mouse model of *ldlr*-/- high fat fed, 5/6 nephrectomy for VC.[8]
- Important differences in baseline characteristics may influence these results; for example, the placebo group had the longest length of time on dialysis, the highest bone turnover biomarkers, and the highest baseline Agatston scores. Conversely, the placebo group had the lowest use of calcium-based phosphate binders, and their PTH decreased through the study to levels more similar to the other groups.
- Analyses of sotatercept-treated subjects into 2 groups based on duration of treatment exposure led to comparable groups based on baseline parameters, and demonstrated an exposure effect on the derangements in CKD-MBD similar to that seen in the dose group analyses.
- If these results bear out in larger studies, sotatercept may be shown to address significant unmet needs in ESKD with favorable effects on CKD-MBD, in addition to its ability to raise Hb.

REFERENCES

1. Kidney Disease: Improving Global Outcomes (KDIGO) CKD-MBD Work Group. KDIGO clinical practice guideline for the diagnosis, evaluation, prevention, and treatment of chronic kidney disease-mineral and bone disorder (CKD-MBD). *Kidney Int Suppl.* 2009;S1-S130.
2. Pearsall RS, Canalis E, Cornwall-Brady M, et al. A soluble activin type IIA receptor induces bone formation and improves skeletal integrity. *Proc Natl Acad Sci U S A.* 2008;105:7082-7087.
3. Fuller K, Bayley KE, Chambers TJ. Activin A is an essential cofactor for osteoclast induction. *Biochem Biophys Res Commun.* 2000;268:2-7.
4. Sakai R, Eto Y, Ohtsuka M, Hirafuji M, et al. Activin enhances osteoclast-like cell formation in vitro. *Biochem Biophys Res Commun.* 1993;195:39-46.
5. Perrien DS, Akel NS, Edwards PK, et al. Inhibin A is an endocrine stimulator of bone mass and strength. *Endocrinology.* 2007;148:1654-1665.
6. Wooldridge T, Kaplan M, Alcorn H Jr, et al. The pharmacokinetics and safety of a single-dose of sotatercept (ACE-011) in subjects on hemodialysis and the effects of its murine analog (RAP-011) on anemia and in preventing bone loss in C57BL/6 mice with 5/6 nephrectomy [oral presentation SA-OR087]. Presented at: the American Society of Nephrology (ASN) Kidney Week 2012; October 30-November 4, 2012; San Diego, CA.
7. Chantry AD, Heath D, Mulivor AW, et al. Inhibiting activin-A signaling stimulates bone formation and prevents cancer-induced bone destruction in vivo. *J Bone Miner Res.* 2010;25:2633-2646.
8. Fang Y, Agapova O, Sugatani T, et al. Treatment of the CKD-MBD with a ligand trap for the activin receptor type 2A [abstract]. Presented at: the American Society of Nephrology (ASN) Kidney Week 2014; November 11-16, 2014; Philadelphia, PA.
9. Agapova O, Fang Y, Sugatani T, et al. Chronic kidney disease (CKD) stimulates aortic endothelial to mesenchymal transition (EnMT) which causes vascular calcification and is inhibited by an activin ligand trap [abstract]. Presented at: the American Society of Nephrology (ASN) Kidney Week 2014; November 11-16, 2014; Philadelphia, PA.
10. Sherman ML, Borgstein NG, Mook L, et al. Multiple-dose, safety, pharmacokinetic, and pharmacodynamic study of sotatercept (ActRIIA-IgG1), a novel erythropoietic agent, in healthy postmenopausal women. *J Clin Pharmacol.* 2013;53:1121-1130.
11. Agatston AS, Janowitz WR, Hildner FJ, et al. Quantification of coronary artery calcium using ultrafast computed tomography. *J Am Coll Cardiol.* 1990;15:827-832.
12. Hokanson JE, MacKenzie T, Kinney G, et al. Evaluating changes in coronary artery calcium: an analytic method that accounts for interscan variability. *AJR Am J Roentgenol.* 2004;182:1327-1332.
13. Leonard MB. A structural approach to skeletal fragility in chronic kidney disease. *Semin Nephrol.* 2009;29:133-143.
14. El-Shahawy M, Cotton J, Kaupke J, et al. Safety and hemoglobin effect of the first 28-day dose cycle of sotatercept 0.7 mg/kg compared with lower doses and placebo for correction of anemia in hemodialysis subjects: interim analysis [poster TH-PO985]. Presented at: the American Society of Nephrology (ASN) Kidney Week 2014; November 11-16, 2014; Philadelphia, PA.
15. Malluche HH, Davenport D, Cantor T, et al. Bone mineral density and serum biochemical predictors of bone loss in patients with CKD on dialysis. *Clin J Am Soc Nephrol.* 2014;9:1254-1262.
16. Duan Y, De Luca V, Seeman E. Parathyroid hormone deficiency and excess: similar effects on trabecular bone but differing effects on cortical bone. *J Clin Endocrinol Metab.* 1999;84:718-722.
17. Malluche HH, Porter DS, Moniere-Faugere MC, et al. Differences in bone quality in low- and high-turnover renal osteodystrophy. *J Am Soc Nephrol.* 2012;23:525-532.
18. Jung HH, Kim SW, Han H. Inflammation, mineral metabolism and progressive coronary artery calcification in patients on haemodialysis. *Nephrol Dial Transplant.* 2006;21:1915-1920.

This study was sponsored by Celgene Corporation.

Presented at: the American Society of Nephrology (ASN) Kidney Week 2014; November 11-16, 2014; Philadelphia, PA.

Scan this QR code to receive the PDF of the poster