Longer-term red blood cell transfusion reduction in the phase 3 MEDALIST study of luspatercept in patients with lower-risk myelodysplastic syndromes with ring sideroblasts

Presentation Number
7518

Scientific Content on Demand
To request a copy of this poster:
Scan QR code via a barcode reader application
QR codes are valid for 180 days after the congress presentation date.

Rami S. Komrokji,[1] Mikkael A. Sekeres,[2] Amer M. Zeidan,[3] Pierre Fenaux,[4] Alan F. List,[5] Amy E. DeZern,[6] Peter L. Greenberg,[7] Michael R. Savona,[8] Joseph G. Jurcic,[9] Amit Verma,[10] Ghulam J. Mufti,[11] Rena Buckstein,[12] Valeria Santini,[13] Abderrahmane Laadem,[14] Rodrigo Ito,[14] Jennie Zhang,[14] Chrystal U. Louis,[14] Peter G. Linde,[15] Guillermo Garcia-Manero[16]

[1]Moffitt Cancer Center, Tampa, FL, USA; [2]Department of Hematology and Medical Oncology, Cleveland Clinic, Cleveland, OH, USA; [3]Department of Internal Medicine, Yale School of Medicine and Yale Cancer Center, Yale University, New Haven, CT, USA; [4]Service d'Hématologie Séniors, Hôpital Saint-Louis, Université Paris 7, Paris, France; [5]Formerly Moffitt Cancer Center, Tampa, FL, USA; [6]The Sidney Kimmel Comprehensive Cancer Center at Johns Hopkins, Baltimore, MD, USA; [7]Stanford University Cancer Center, Stanford, CA, USA; [8]Vanderbilt-Ingram Cancer Center, Vanderbilt University School of Medicine, Nashville, TN, USA; [9]Division of Hematology/Oncology, Herbert Irving Comprehensive Cancer Center, Columbia University Medical Center, New York, NY, USA; [10]Department of Oncology, Albert Einstein College of Medicine, Montefiore Medical Center, New York, NY, USA; [11]Department of Haemato-Oncology, King's College London, London, UK; [12]Odette Cancer Centre, Sunnybrook Health Sciences Centre, Toronto, ON, Canada; [13]MDS Unit, Azienda Ospedaliero Universitaria Careggi, University of Florence, Florence, Italy; [14]Bristol Myers Squibb, Princeton, NJ, USA; [15]Acceleron Pharma, Cambridge, MA, USA; [16]Department of Leukemia, University of Texas MD Anderson Cancer Center, Houston, TX, USA

Introduction

- Lower-risk myelodysplastic syndromes (LR MDS) are characterized by ineffective erythropoiesis, leading to anemia, red blood cell (RBC) transfusion dependence, and reduced quality of life[1,2]
- There is a need for effective treatments for anemia in transfusion-dependent patients with LR MDS[1,2]
- Luspatercept is a first-in-class erythroid maturation agent that binds to select transforming growth factor-β superfamily ligands to decrease Smad2/3 signaling and enhance late-stage erythropoiesis[3]
 - Luspatercept is indicated for the treatment of anemia in adult patients with LR MDS with ring sideroblasts (RS) who require regular RBC transfusions
- MEDALIST is an ongoing phase 3, randomized, double-blind, placebo-controlled trial (NCT02631070) evaluating the efficacy and safety of luspatercept in patients with LR MDS with RS who require RBC transfusions[4]

Objective

- To evaluate long-term transfusion burden reduction in patients treated with luspatercept in the MEDALIST trial

Methods

Study design

- Patients were randomized at 65 sites between March 2016 and June 2017
- Eligible patients were aged ≥ 18 years and had anemia due to Revised International Prognostic Scoring System-defined Very low-, Low-, or Intermediate-risk MDS with RS; were refractory to, intolerant of, or unlikely to respond to (serum erythropoietin > 200 U/L) erythropoiesis-stimulating agents; and required RBC transfusions
- Patients were randomized 2:1 to receive luspatercept (starting dose 1.0 mg/kg, with titration allowed up to 1.75 mg/kg) or placebo subcutaneously every 3 weeks for ≥ 24 weeks (**Figure 1**)
- Patients receiving placebo were not allowed to cross over to the luspatercept arm

Endpoints

Study endpoints

- Primary endpoint: RBC transfusion independence (TI) ≥ 8 weeks (Weeks 1–24)
- Key secondary endpoint: RBC-TI ≥ 12 weeks (Weeks 1–24 and Weeks 1–48)

Additional endpoints

- Reduction in RBC units transfused from baseline
- Achievement of modified hematologic improvement-erythroid (HI-E) (per International Working Group [IWG] 2006 criteria[5])
 - Defined as a reduction of ≥ 4 units RBCs from baseline, sustained for ≥ 8 weeks (for patients with baseline RBC transfusion burden of ≥ 4 units/8 weeks) or a mean hemoglobin increase of ≥ 1.5 g/dL from baseline, sustained for ≥ 8 weeks (for patients with baseline RBC transfusion burden of < 4 units/8 weeks)
- Longest duration of primary response
- Change from baseline in number of transfusion visits
- Change in iron parameters

Figure 1. MEDALIST trial study design

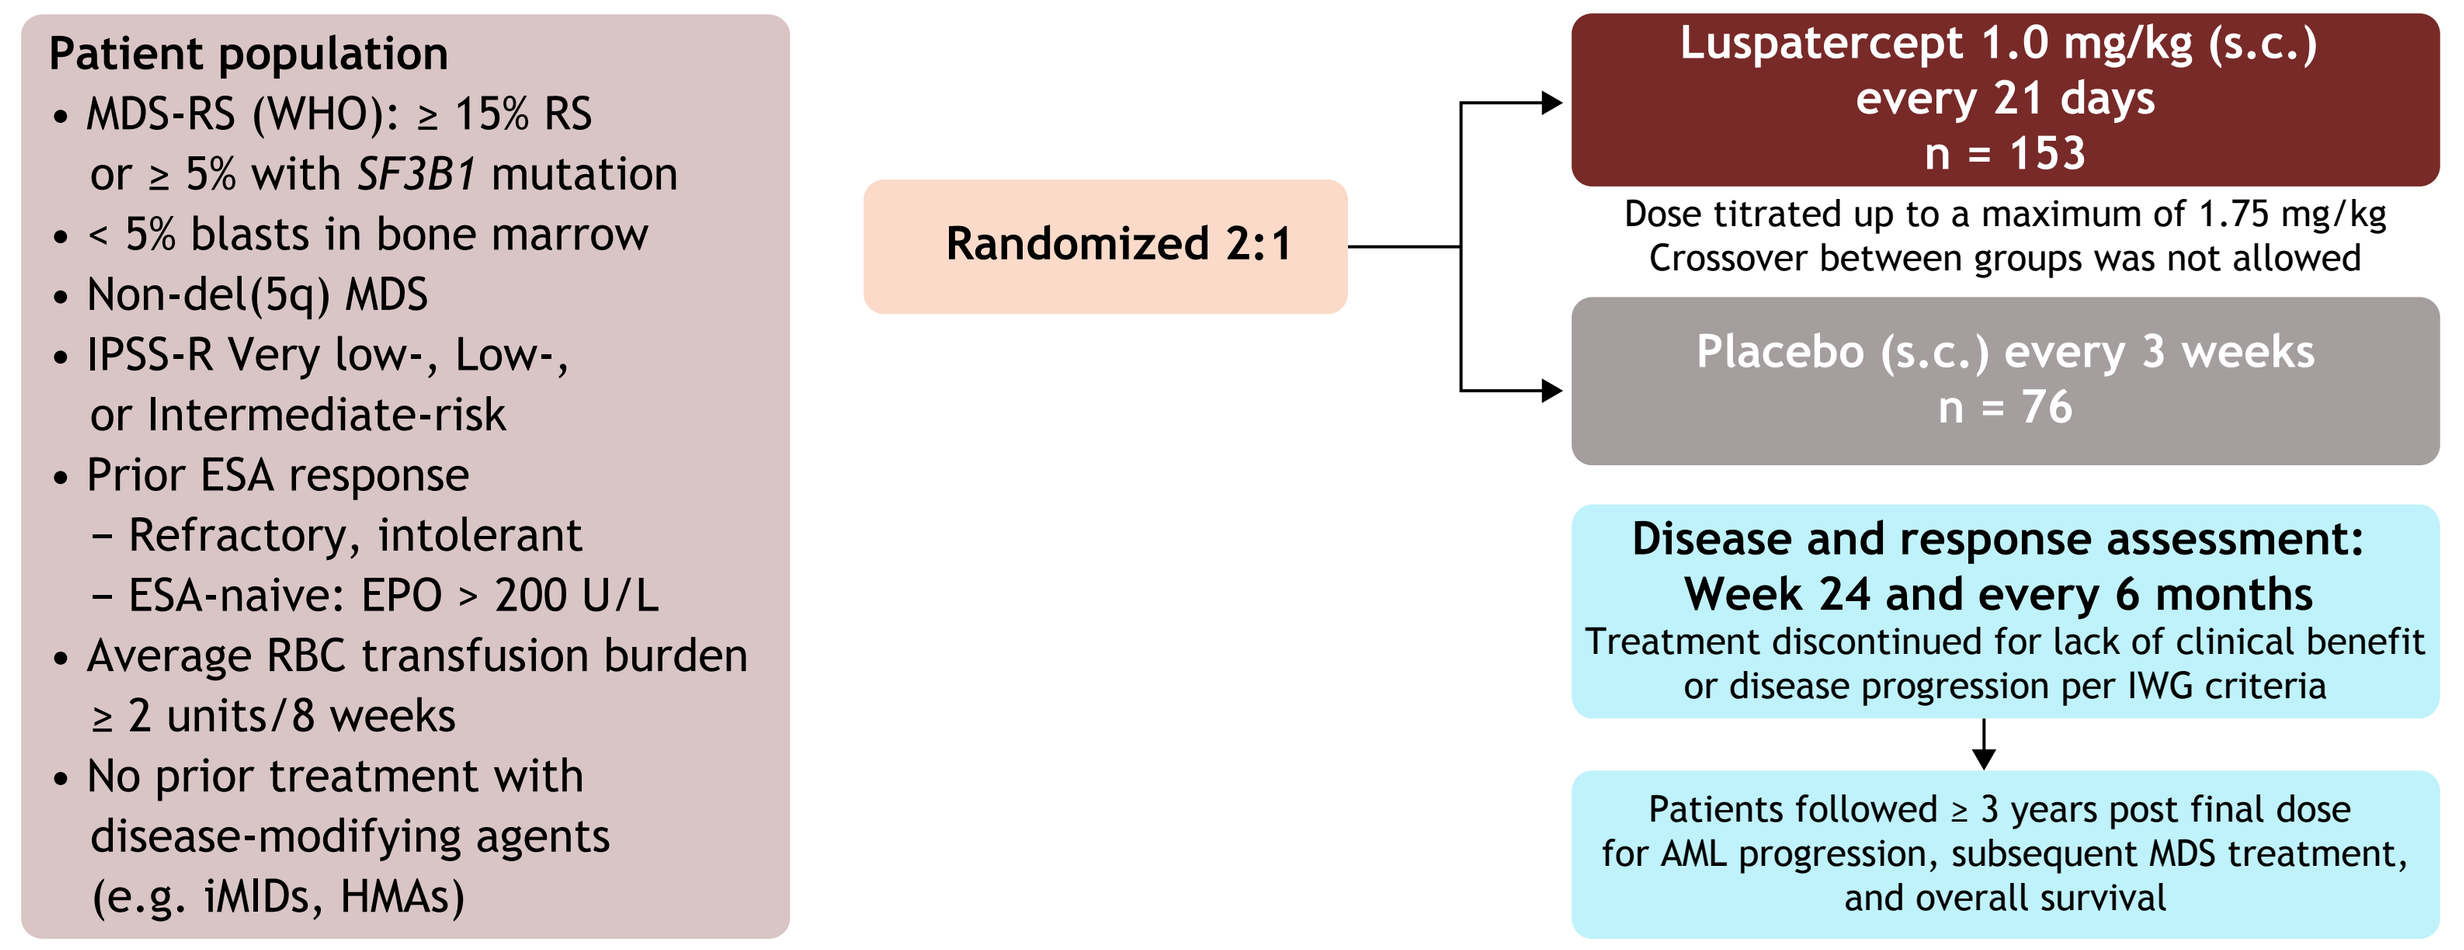
Patient population
• MDS-RS (WHO): ≥ 15% RS or ≥ 5% with SF3B1 mutation
• < 5% blasts in bone marrow
• Non-del(5q) MDS
• IPSS-R Very low-, Low-, or Intermediate-risk
• Prior ESA response
– Refractory, intolerant
– ESA-naive: EPO > 200 U/L
• Average RBC transfusion burden ≥ 2 units/8 weeks
• No prior treatment with disease-modifying agents (e.g. iMIDs, HMAs)
Randomized 2:1
Luspatercept 1.0 mg/kg (s.c.) every 21 days n = 153
Dose titrated up to a maximum of 1.75 mg/kg
Crossover between groups was not allowed
Placebo (s.c.) every 3 weeks n = 76
Disease and response assessment: Week 24 and every 6 months
Treatment discontinued for lack of clinical benefit or disease progression per IWG criteria
Patients followed ≥ 3 years post final dose for AML progression, subsequent MDS treatment, and overall survival

Data cutoff: July 1, 2019.
AML, acute myeloid leukemia; EPO, erythropoietin; ESA, erythropoiesis-stimulating agent; HMA, hypomethylating agent; iMID, immunomodulatory drug; IPSS-R, Revised International Prognostic Scoring System; IWG, International Working Group; MDS, myelodysplastic syndromes; RBC, red blood cell; RS, ring sideroblast; s.c., subcutaneously; *SF3B1*, splicing factor 3b subunit 1; WHO, World Health Organization.

Results

Patients

- 229 patients were randomized; 153 to receive luspatercept and 76 to receive placebo (**Table 1**)
- As of data cutoff (July 1, 2019), the median follow-up time in the luspatercept and placebo arms was 26.4 and 26.1 months, respectively
- The median (range) transfusion burden at baseline was 5 (1-15) and 5 (2-20) RBC units over 8 weeks in the luspatercept and placebo arms
- The mean (standard deviation) transfusion burden at baseline was 5.5 (2.76) and 5.8 (2.95) RBC units over 8 weeks in the luspatercept and placebo arms

Table 1. Baseline characteristics

Characteristic	Luspatercept (N = 153)	Placebo (N = 76)
Age, median (range), years	71 (40–95)	72 (26–91)
Male, n (%)	94 (61.4)	50 (65.8)
RBC transfusion burden category, n (%)[a]		
≥ 6 U/8 weeks	66 (43.1)	33 (43.4)
≥ 4 to < 6 U/8 weeks	41 (26.8)	23 (30.3)
< 4 U/8 weeks	46 (30.1)	20 (26.3)
IPSS-R, n (%)[b]		
Very low	18 (11.8)	6 (7.9)
Low	109 (71.2)	57 (75.0)
Intermediate	25 (16.3)	13 (17.1)
***SF3B1* mutation, n (%)[c]**	138 (93.2)[c]	64 (86.5)[c]
Serum EPO, n (%)[d]		
< 200 U/L	88 (57.5)	50 (65.8)
≥ 200 U/L	64 (41.8)	26 (34.2)
Previously received ESAs, n (%)	148 (96.7)	70 (92.1)
Baseline Hb, median (range), g/dL[e]	7.6 (6–10)	7.6 (5-9)
Serum ferritin, mean (SD), μg/L	1,348.0 (971.2)	1,503.8 (1,242.9)

[a]In the 16 weeks prior to randomization. [b]1 patient in the luspatercept arm was classified as IPSS-R High risk. [c]Of patients with available baseline gene mutation data: n = 148 in the luspatercept arm; and n = 74 in the placebo arm; no patients with *SF3B1* mutation had RS <15%. [d]Data were missing for 1 patient in the luspatercept arm. [e]Baseline Hb was defined as the last value measured on or before the date and time of first dose. EPO, erythropoietin; ESA, erythropoiesis-stimulating agent; Hb, hemoglobin; IPSS-R, Revised International Prognostic Scoring System; RBC, red blood cell; RS, ring sideroblast; SD, standard deviation; *SF3B1*, splicing factor 3b subunit 1.

Long-term transfusion burden reduction

- The primary endpoint (RBC-TI ≥ 8 weeks during Weeks 1-24) was achieved by 58 of 153 (37.9%) and 10 of 76 (13.2%) patients receiving luspatercept and placebo, respectively ($P < 0.0001$)
- 73 of 153 (47.7%) and 12 of 76 (15.8%) patients receiving luspatercept and placebo, respectively, achieved RBC-TI ≥ 8 weeks any time during the treatment period ($P < 0.0001$)
- 77 of 153 (50.3%) patients receiving luspatercept and 11 of 76 (14.5%) patients receiving placebo achieved ≥ 50% reduction in RBC transfusion burden from baseline for ≥ 24 weeks ($P < 0.0001$)
 - The median longest episode of RBC transfusion burden reduction ≥ 50% was 131.6 weeks in the luspatercept arm and not estimable in the placebo arm

Change in RBC transfusions on treatment

- In Weeks 9–24, mean change from baseline in RBC units transfused was −3.0 (95% confidence interval [CI] −3.9, −2.1) versus +0.4 (95% CI −0.6, +1.4) in the luspatercept versus placebo arms (**Figure 2**)
 - In Weeks 33–48, mean change from baseline in RBC units transfused was −4.9 (95% CI −5.9, −3.9) in patients receiving luspatercept
- In Weeks 1–24, mean number of transfusion visits was 5.9 versus 9.5 in the luspatercept versus placebo arms (**Figure 3**)

Total transfusions over 48 weeks

- The mean number of RBC units transfused during Weeks 1–48 was 22.89 versus 35.98 in luspatercept versus placebo arms ($P < 0.0001$) (**Table 2**)
- The mean number of RBC transfusion visits during Weeks 1–48 was 12.95 versus 19.54 in the luspatercept versus placebo arms ($P < 0.0001$) (**Table 2**)

Figure 2. Mean change from baseline in RBC units transfused

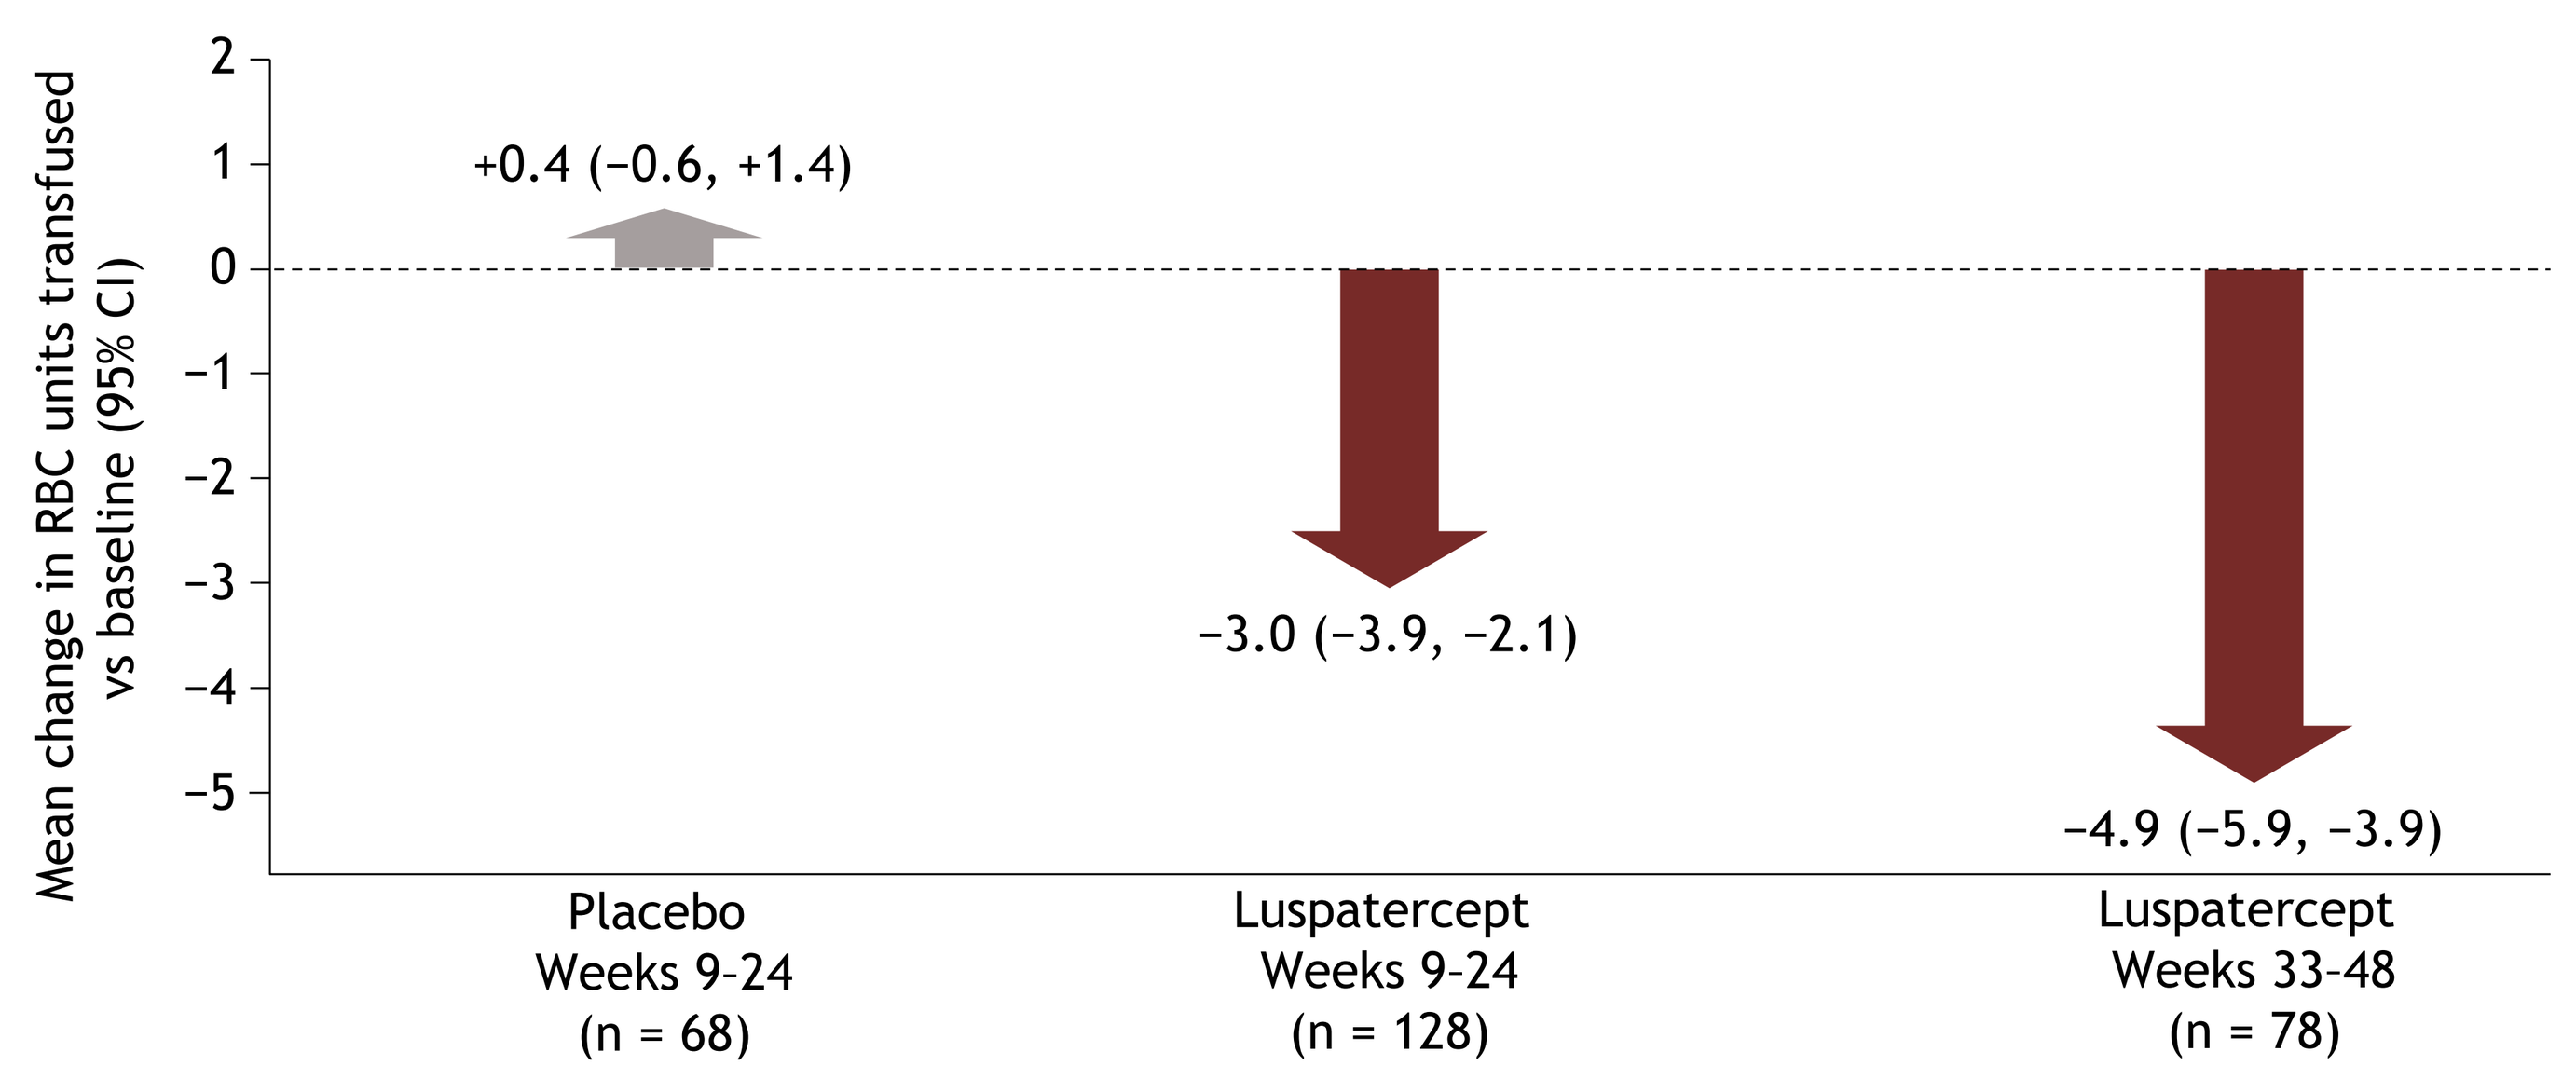
Mean change in RBC units transfused vs baseline (95% CI)
2
1
0
−1
−2
−3
−4
−5
+0.4 (−0.6, +1.4)
−3.0 (−3.9, −2.1)
−4.9 (−5.9, −3.9)
Placebo Weeks 9-24 (n = 68)
Luspatercept Weeks 9-24 (n = 128)
Luspatercept Weeks 33-48 (n = 78)

CI, confidence interval; RBC, red blood cell.

Figure 3. Mean number of transfusion visits in Weeks 1-24

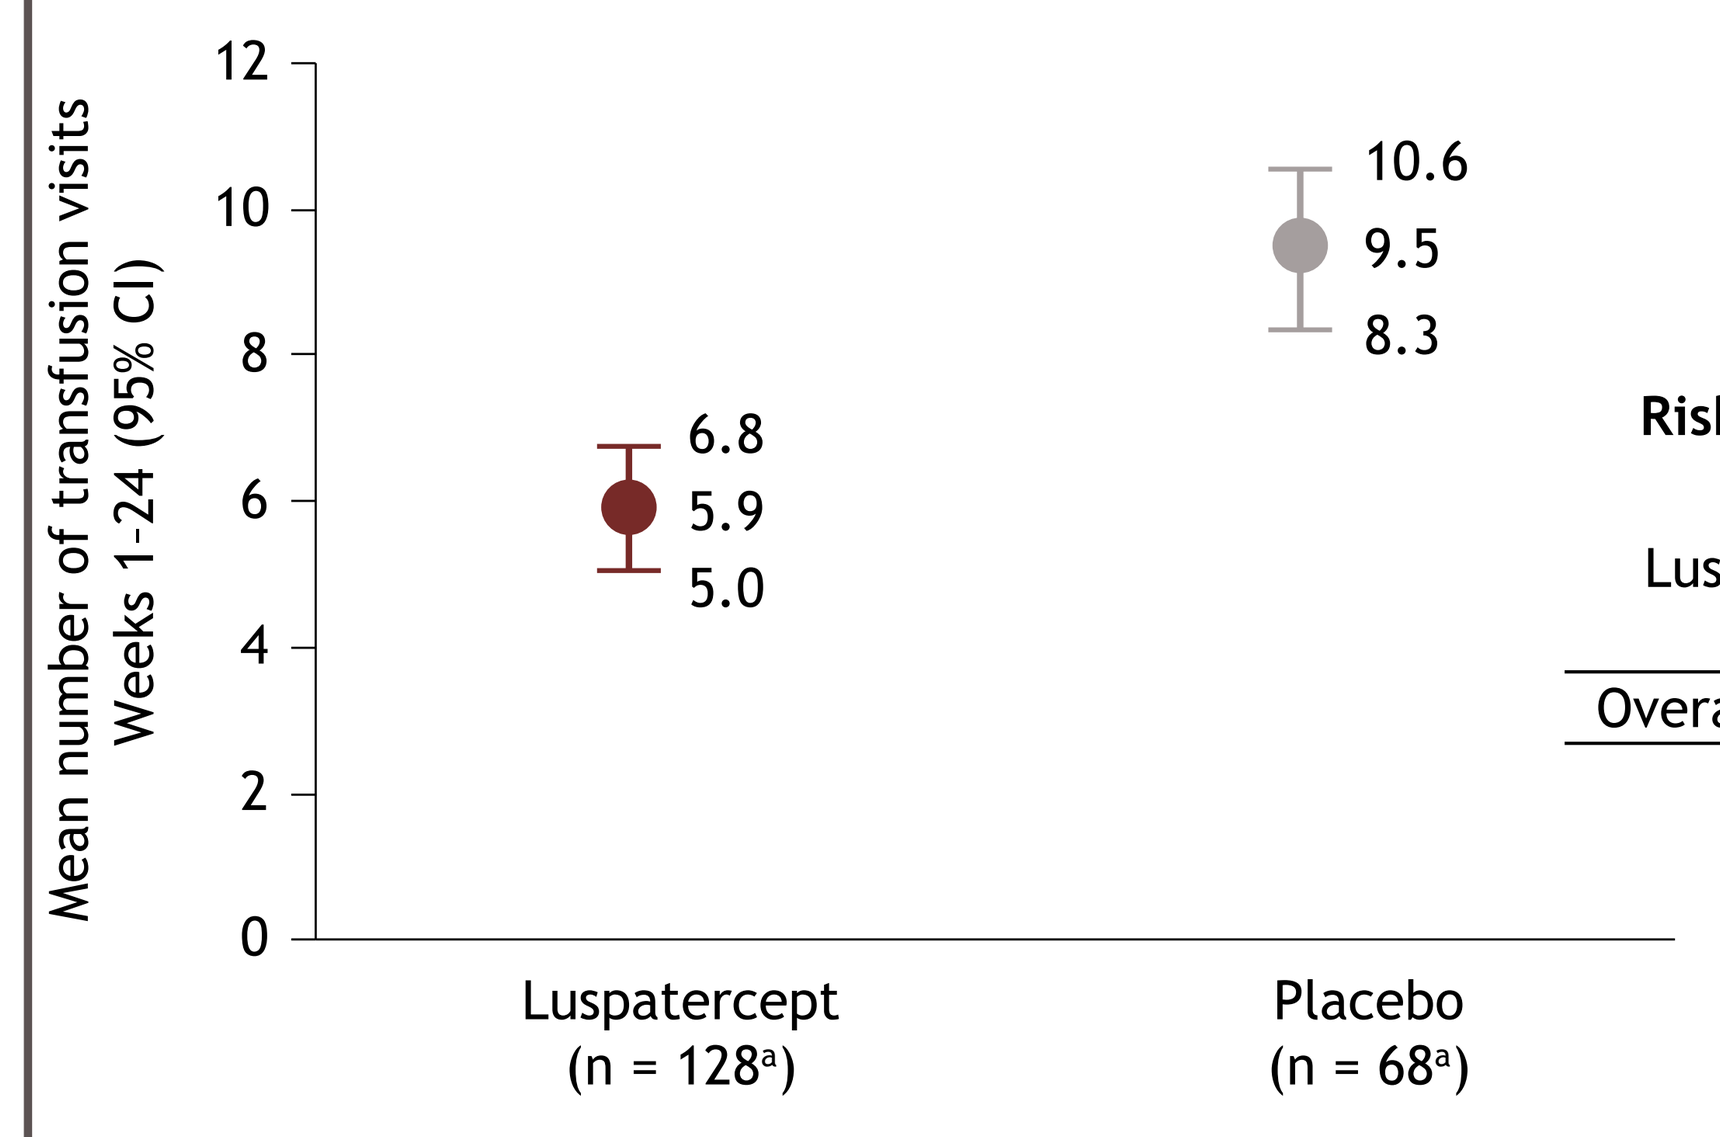
Mean number of transfusion visits Weeks 1-24 (95% CI)
12
10
8
6
4
2
0
6.8
5.9
5.0
10.6
9.5
8.3
Luspatercept (n = 128a)
Placebo (n = 68a)

Risk of recurrent transfusion visits during Weeks 1-24[a,b]

Luspatercept vs placebo	Hazard ratio (95% CI)
Overall population	0.699 (0.597, 0.819)

[a]83.7% (128/153) patients treated with luspatercept and 89.5% (68/76) placebo patients completed 24 weeks of treatment and were considered having complete data for this analysis. [b]A Cox model using the Andersen-Gill model[6] to assess the recurrence of transfusion visits during the primary treatment phase (Weeks 1–24).
CI, confidence interval.

Table 2. Number of RBC units transfused and transfusion visits over 48 weeks

	Luspatercept (n = 153)	Placebo (n = 76)
Number of RBC units transfused over 48 weeks		
Mean	22.89	35.98
LS mean	23.28	35.20
LS mean difference (95% CI)	−11.92 (−15.55, −8.28)	
P value	< 0.0001	
Number of RBC transfusion visits over 48 weeks		
Mean	12.95	19.54
LS mean	13.14	19.15
LS mean difference (95% CI)	−6.00 (−8.16, −3.85)	
P value	< 0.0001	

CI, confidence interval; LS, least squares; RBC, red blood cell.

Change in serum ferritin

- Least squares (LS) mean change in serum ferritin from baseline to Weeks 9–24 was −2.7 μg/L and +226.5 μg/L in the luspatercept and placebo arms, respectively (LS mean difference −229.1 μg/L; $P = 0.0024$) (**Table 3**)
 - The mean change in serum ferritin in Weeks 33–48 was −72.0 μg/L and +247.4 μg/L in the luspatercept and placebo arms, respectively (LS mean difference −319.5 μg/L; $P = 0.0294$)

Table 3. Mean change from baseline in serum ferritin

	Averaged over Weeks 9-24		Averaged over Weeks 33-48	
	Luspatercept (n = 153)	Placebo (n = 76)	Luspatercept (n = 153)	Placebo (n = 76)
LS mean (SE), μg/L	−2.7 (54.05)	+226.5 (68.02)	−72.0 (74.76)	+247.4 (140.96)
LS mean difference (95% CI), μg/L	−229.1 (74.43)		−319.5 (144.57)	
***P* value**	0.0024		0.0294	

CI, confidence interval; LS, least squares; SE, standard error.

Achievement of HI-E

- In Weeks 1–24, 81 of 153 (52.9%) patients in the luspatercept arm and 9 of 76 (11.8%) patients in the placebo arm achieved modified HI-E response per IWG 2006 criteria

Conclusions

- Luspatercept produced clinically meaningful and durable reductions in RBC transfusions in patients with LR-MDS with RS
- Luspatercept also resulted in statistically significant reductions in serum ferritin in patients with LR-MDS with RS

References

1. Adès L, et al. *Lancet* 2014;383:2239-2252.
2. Fenaux P, Adès L. *Blood* 2013;121:4280-4286.
3. Suragani RN, et al. *Nat Med* 2014;20:408-414.
4. Fenaux P, et al. *N Engl J Med* 2020;382:140-151.
5. Cheson BD, et al. *Blood* 2006;108:419-425.
6. Andersen PK, Gill RD. *Ann Stat* 1982;10:1100-1120.

Acknowledgments

- The study was supported by Celgene, a Bristol-Myers Squibb Company
- All authors contributed to and approved the presentation; writing and editorial assistance were provided by Daniel Gilmartin, PhD, of Excerpta Medica, funded by Bristol-Myers Squibb Company

Disclosures

R.S.K.: BMS, Novartis, Daiichi Sankyo, Pfizer, Janssen, Agios, Incyte - consulting or advisory role; Novartis, Alexion Pharmaceuticals, Jazz Pharmaceuticals - speakers' bureau; BMS, Incyte, Alexion Pharmaceuticals, Novartis, Jazz Pharmaceuticals, Daiichi Sankyo - travel, accommodations, expenses; AbbVie - stock and other ownership interests. M.A.S.: BMS, Millennium, Syros Pharmaceuticals - consulting or advisory role; Takeda, Pfizer - research funding. A.M.Z.: AbbVie, Otsuka, Pfizer, BMS, Jazz Pharmaceuticals, Incyte, Agios, Boehringer Ingelheim, Novartis, Acceleron Pharma, Astellas Pharma, Daiichi Sankyo, Cardinal Health, Taiho Pharmaceutical, Seattle Genetics, BeyondSpring Pharmaceuticals, Trovagene, Takeda, Ionis Pharmaceuticals, Epizyme - honoraria, consulting or advisory role; BMS, AbbVie, Astex Pharmaceuticals, Pfizer, AstraZeneca/MedImmune, Boehringer Ingelheim, Trovagene, Incyte, Takeda, Novartis, Aprea Therapeutics, ADC Therapeutics - research funding; Pfizer, Novartis, BMS, Trovagene - travel, accommodations, expenses. P.F.: BMS, Janssen, Jazz Pharmaceuticals, AbbVie - honoraria, consulting or advisory role; Jazz Pharmaceuticals - travel, accommodations, expenses. A.F.L.: BMS, Cellular Biomedicine Group, Aileron Therapeutics, Acceleron Pharma, International Personalized Cancer Center Advisory Committee - consulting or advisory role; BMS, Cellular Biomedicine Group - travel, accommodations, expenses; Thousand Talents Award - other relationship; BMS, Aileron Therapeutics, Cellular Biomedical Group - honoraria; BMS - research funding. A.E.D.: Acceleron Pharma, Syros Pharmaceuticals, Otsuka US - consulting or advisory role. P.L.G.: BMS, Amgen - stock and other ownership interests; Novartis - consulting or advisory role; BMS, Novartis, H3 Biomedicine, Aprea Therapeutics, Notable Labs, Genentech - research funding. M.R.S.: Karyopharm Therapeutics, BMS, TG Therapeutics, Merck, Ryvu - consulting or advisory role; Boehringer Ingelheim - patents, royalties, other intellectual property (property license); Karyopharm Therapeutics - stock and other ownership interests; Sunesis Pharmaceuticals, TG Therapeutics, Astex Pharmaceuticals, Incyte, Takeda - research funding. J.G.J.: Novartis, AstraZeneca, Boston Biomedical, BMS, Daiichi Sankyo, AbbVie - consulting or advisory role; BMS, Astellas Pharma, Actinium Pharmaceuticals, Daiichi Sankyo, FORMA Therapeutics, Seattle Genetics, Kura, Syros Pharmaceuticals, Genentech, AbbVie - research funding. A.V.: Stelexis Therapeutics - stock and other ownership interests; Stelexis Therapeutics, BMS, Acceleron Pharma - honoraria; Janssen Oncology, BMS - research funding. G.J.M.: Novartis, AbbVie - consulting or advisory role; Novartis, BMS - research funding. R.B.: BMS - honoraria, consulting or advisory role, research funding; Otsuka - research funding. V.S.: BMS, Johnson & Johnson, Novartis - honoraria; Acceleron Pharma, Amgen, BMS, Menarini, Novartis - consulting or advisory role. A.L., J.Z.: BMS - employment, stock and other ownership interests. R.I.: BMS - employment; Bayer - employment (immediate family member); BMS - stock and other ownership interests. C.U.L.: BMS - employment, stock and other ownership interests, patents, royalties, other intellectual property; Cell Medica, Merrimack - patents, royalties, other intellectual property. P.G.L.: Acceleron Pharma - employment, stock and other ownership interests, patents, royalties, other intellectual property; FibroGen, Abbott, AbbVie - stock and other ownership interests. G.G.-M.: BMS, Astex Pharmaceuticals, Acceleron Pharma, Jazz Pharmaceuticals, Helsinn Therapeutics - consulting or advisory role; BMS, Astex Pharmaceuticals, Acceleron Pharma, Helsinn Therapeutics, AbbVie - honoraria; BMS, Astex Pharmaceuticals, Amphivena Therapeutics, Helsinn Therapeutics, Novartis, AbbVie, Onconova Therapeutics, H3 Biomedicine, Merck - research funding.

Presented at the American Society of Clinical Oncology (ASCO), May 29–31, 2020.

Contact: http://www.globalbmsmedinfo.com

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.