Sotatercept Improves Anemia, Vascular Calcification, and Bone Loss in Patients With End-Stage Kidney Disease on Hemodialysis

TH-P0038

John Havill[1]; Nelson Kopyt[2]; Daniel Coyne[3]; Michael Weiswasser[4]; William T. Smith[4]

[1]Kidney Specialists of Southern Nevada, Las Vegas, NV; [2]Lehigh Valley Health Network, Allentown, PA; [3]Washington University, St Louis, MO; [4]Celgene Corporation, Warren, NJ

INTRODUCTION

- Patients with end-stage kidney disease (ESKD) exhibit anemia, primarily caused by decreased renal biosynthesis of erythropoietin (EPO).[1]
- ESKD also gives rise to renal osteodystrophy (ROD) and vascular calcification (VC), integral components of chronic kidney disease-mineral and bone disorder (CKD-MBD), which is strongly associated with unacceptable morbidity and mortality.[2]
- Sotatercept (ACE-011) is a type II activin A receptor (ActRIIA)-IgG1 fusion protein that binds with high affinity to activin A and other members of the transforming growth factor-β superfamily; by blocking signaling through the ActRIIA receptor, sotatercept stimulates erythropoiesis[3,4] and decreases CKD-MBD–related bone resorption and VC.[5]
- In an ongoing 2-part, randomized, single-blind, placebo-controlled trial, sotatercept is being studied for the correction of anemia and improvement of measures of CKD-MBD in subjects with ESKD on hemodialysis (HD).
- In Part 1 of the study, a single subcutaneous (SC) dose of sotatercept 0.1 mg/kg in subjects with ESKD demonstrated a long half-life (21 days) and good tolerability, with no observed changes in blood pressure (BP).[6]
- The current interim analysis of Part 2 describes the pharmacokinetics (PK), safety, and hemoglobin (Hb) effects of sotatercept, explores its effects on VC and bone mineral density (BMD) using quantitative computed tomography (QCT), and builds on a previous report.[7]

METHODS

- In an ongoing study of sotatercept in HD subjects for the correction of anemia, subjects responding to an erythropoietin-stimulating agent (ESA) were washed out of their ESA effect until Hb was <10 g/dL, and then randomized (3:1) to receive escalating dose levels of sotatercept (0.3 mg/kg, 0.5 mg/kg, or 0.7 mg/kg SC) or placebo every 28 days for up to 8 dose cycles; subjects randomized to a 14-day dose group received sotatercept 0.7 mg/kg given every 14 days for 2 doses, then 0.4 mg/kg (0.7/0.4 mg/kg) every 14 days or placebo for up to 13 doses (**Figure 1**). Treatment failures (Hb <9 g/dL) were administered ESA and/or red blood cell transfusion (rescue therapy).
- QCT scans of the hip, lumbar spine, and abdominal aorta were obtained at baseline and after the 225-day treatment phase.
- Interim results are reported for PK, safety, home BP, and Hb in the placebo and sotatercept 0.3 mg/kg, 0.5 mg/kg, 0.7 mg/kg, and 0.7/0.4 mg/kg groups; data from the 0.7/0.4 mg/kg dose group is incomplete, and evaluation of this group is ongoing. VC and BMD effects at Day 225 are also described for the placebo and sotatercept 0.3 mg/kg, 0.5 mg/kg, and 0.7 mg/kg groups; such data are not yet available for the 0.7/0.4 mg/kg dose group.

Figure 1. Part 2 Study Design

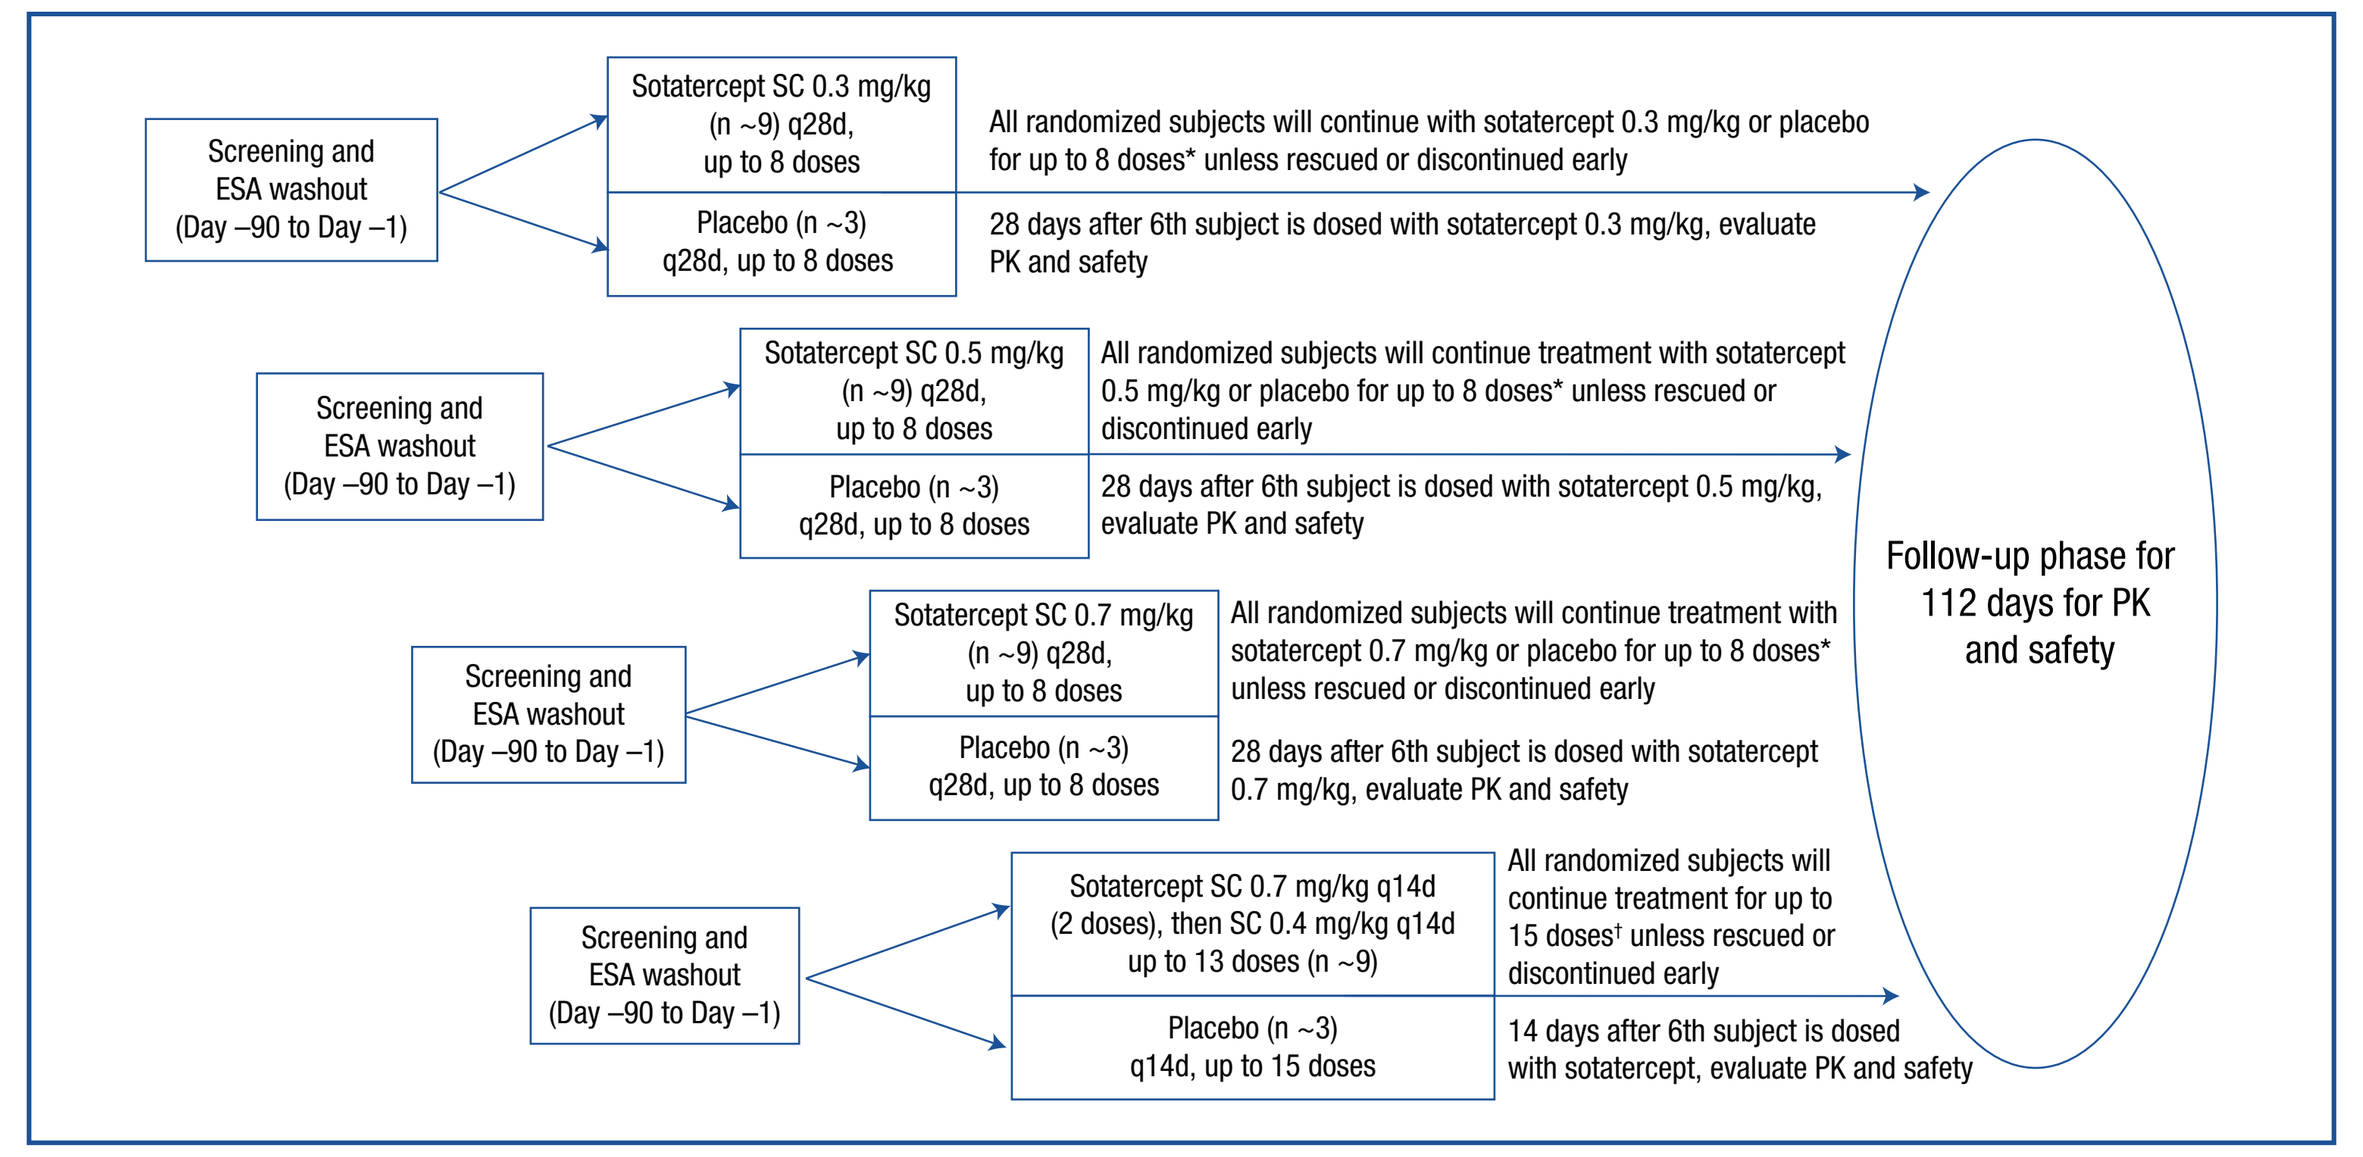
Screening and ESA washout (Day –90 to Day –1)
Sotatercept SC 0.3 mg/kg (n ~9) q28d, up to 8 doses
Placebo (n ~3) q28d, up to 8 doses
All randomized subjects will continue with sotatercept 0.3 mg/kg or placebo for up to 8 doses* unless rescued or discontinued early
28 days after 6th subject is dosed with sotatercept 0.3 mg/kg, evaluate PK and safety
Screening and ESA washout (Day –90 to Day –1)
Sotatercept SC 0.5 mg/kg (n ~9) q28d, up to 8 doses
Placebo (n ~3) q28d, up to 8 doses
All randomized subjects will continue treatment with sotatercept 0.5 mg/kg or placebo for up to 8 doses* unless rescued or discontinued early
28 days after 6th subject is dosed with sotatercept 0.5 mg/kg, evaluate PK and safety
Screening and ESA washout (Day –90 to Day –1)
Sotatercept SC 0.7 mg/kg (n ~9) q28d, up to 8 doses
Placebo (n ~3) q28d, up to 8 doses
All randomized subjects will continue treatment with sotatercept 0.7 mg/kg or placebo for up to 8 doses* unless rescued or discontinued early
28 days after 6th subject is dosed with sotatercept 0.7 mg/kg, evaluate PK and safety
Screening and ESA washout (Day –90 to Day –1)
Sotatercept SC 0.7 mg/kg q14d (2 doses), then SC 0.4 mg/kg q14d up to 13 doses (n ~9)
Placebo (n ~3) q14d, up to 15 doses
All randomized subjects will continue treatment for up to 15 doses† unless rescued or discontinued early
14 days after 6th subject is dosed with sotatercept, evaluate PK and safety
Follow-up phase for 112 days for PK and safety

*If Hb levels are >11 g/dL and have not returned to <11 g/dL by Day 36 (Day 29 + 7 days) of a dosing cycle, sotatercept/placebo doses 2 to 8 may be delayed until Hb is <11 g/dL. Subject will be dosed when Hb is <11 g/dL.
†If Hb levels are >11 g/dL and have not returned to <11 g/dL by Day 19 (Day 15 + 4 days) of a dosing cycle, sotatercept/placebo doses 2 to 15 may be delayed until Hb is <11 g/dL. Subject will be dosed when Hb is <11 g/dL.

RESULTS

Subjects

- A total of 43 subjects were randomized and received ≥1 dose of study medication and comprise the full analysis set (FAS) for the efficacy and safety analyses; subject disposition is summarized in **Table 1**.
- A total of 28 subjects discontinued the study drug and received rescue therapy (**Table 1**); among these, 1 placebo subject was discontinued from the study drug because of an adverse event (AE) of fatigue and was subsequently rescued. The most frequent reasons for early termination from the study were withdrawal of consent in 5 subjects (all but 1 [placebo] occurred after discontinuation of study drug due to rescue) and kidney transplant in 2 subjects.
- Baseline subject demographics and clinical characteristics were generally similar across the treatment groups (**Table 2**).

Table 1. Disposition of Randomized Subjects (FAS, N=43)

		Sotatercept			
	Placebo n=11	0.3 mg/kg n=9	0.5 mg/kg n=8	0.7 mg/kg n=9	0.7/0.4 mg/kg n=6
Days on therapy, mean (range)	56.6 (13–213)	85.2 (22–213)	108.4 (22–226)	152.2 (45–225)	98.6 (38–205)
Subjects requiring rescue, n (%)	9 (81.8)	7 (77.8)	4 (50.0)	5 (55.6)	3 (50.0)
Subject withdrawals due to an AE, n (%)	1 (9.1)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)

Table 2. Baseline Demographics and Clinical Characteristics of Randomized Subjects (FAS, N=43)

		Sotatercept			
	Placebo n=11	0.3 mg/kg n=9	0.5 mg/kg n=8	0.7 mg/kg n=9	0.7/0.4 mg/kg n=6
Age, mean (range), years	55.2 (35–76)	59.9 (36–79)	56.9 (42–76)	61.7 (39–76)	55.0 (29–72)
Female, n (%)	4 (36.4)	6 (66.7)	1 (12.5)	7 (77.8)	1 (16.7)
Race, n (%)					
White	3 (27.3)	3 (33.3)	4 (50.0)	7 (77.8)	1 (16.7)
Black	6 (54.5)	6 (66.7)	4 (50.0)	1 (11.1)	4 (66.7)
Asian	2 (18.2)	0 (0.0)	0 (0.0)	0 (0.0)	1 (16.7)
Other	0 (0.0)	0 (0.0)	0 (0.0)	1 (11.1)	0 (0.0)
Ethnicity, n (%)					
Hispanic	1 (9.1)	2 (22.2)	4 (50.0)	2 (22.2)	1 (16.7)
Non-Hispanic	10 (90.9)	7 (77.8)	4 (50.0)	7 (77.8)	5 (83.3)
Postdialysis weight, mean, kg	80.9	78.6	79.1	80.8	81.0
Body mass index, mean, kg/m²	28.1	27.8	26.5	28.7	30.3
Baseline Hb, mean (range), g/dL	9.5 (8.9–9.9)	9.3 (7.3–10.5)	8.9 (7.1–10.0)	9.3 (8.2–10.0)	9.0 (8.2–9.7)

- Among subjects with paired QCT measurements at baseline and Day 225 (n=21), differences in baseline bone biomarker and Agatston scores were observed between groups, with values for these indicating more severe ROD and VC, respectively, among the placebo subjects (**Table 3**).

Table 3. Mean Baseline Biomarker, Volumetric BMD, and Agatston Scores of Subjects With Paired QCT Measurements

		Sotatercept		
	Placebo n=4	0.3 mg/kg n=6	0.5 mg/kg n=5	0.7 mg/kg n=6
Baseline whole PTH, pg/mL	209.2	104.8	135.5	100.4
BSAP, μg/L	24.0	18.0	13.2	15.5
P1NP, ng/mL	548.3	376.2	468.4	437.5
CTX, pg/mL	3,152.3	2,062.5	2,266.8	2,246.5
Total hip integral BMD, mg/cm³	276.8	286.6	280.0	281.0
Femoral neck cortical BMD, mg/cm³	640.5	667.0	593.1	594.8
Lumbar spine (L1, L2) BMD, mg/cm³	140.1	125.6	149.1	118.7
VC total Agatston score*	6,498.8	9,472.7	3,618.5	1,862.1

PTH=parathyroid hormone; BSAP=bone-specific alkaline phosphatase; P1NP=pro-collagen type 1 N-terminal propeptide; CTX=C-terminal type 1 collagen telopeptide.

Safety Assessments

- Among subjects who received ≥1 dose of study medication, AEs were mostly mild to moderate in severity, unrelated to study drug, relatively similar in type/severity between groups, and generally consistent with subject medical histories (**Table 4**).
- Serious AEs were generally considered unrelated to the study drug, did not lead to discontinuation, and resolved with continued therapy.
- No deaths were reported in any of the sotatercept dose groups.
- Two deaths occurred in the placebo group; neither was considered related to study treatment (n=1, Day 148, cardiomyopathy; n=1, Day 186, worsening coronary artery disease).
- Eleven subjects had AEs of hypertension. The distribution and severity of events were not dose-dependent. All events were mild to moderate in severity, and most resolved, with transient, non-persistent increases in BP. Many events occurred weeks after the last dose of study drug and/or after rescue with epoetin was initiated.
- Persistent but minor clinical laboratory changes were observed with some sotatercept doses, including:
 - Decreased calcium levels, without AEs of hypocalcemia (sotatercept 0.3 mg/kg, 0.7 mg/kg, and 0.7/0.4 mg/kg).
 - Decreased follicle-stimulating hormone (expected pharmacodynamic effect, sotatercept 0.7 mg/kg and 0.7/0.4 mg/kg).
 - Increased lactate dehydrogenase (during the first 5 dose cycles for sotatercept 0.3 mg/kg, 0.5 mg/kg, and 0.7 mg/kg, and generally first 7 cycles for 0.7/0.4 mg/kg).
- No other trends in laboratory, electrocardiogram, or vital sign parameters were observed, including study visit and intra-dialytic BP, in any sotatercept dose group during long-term follow-up.
- One subject developed low-level anti-drug antibody titers without consequence to PK, Hb effect, or safety. No injection site or hypersensitivity reactions were observed in any subjects.

Table 4. Overview of AEs

		Sotatercept			
	Placebo n=11	0.3 mg/kg n=9	0.5 mg/kg n=8	0.7 mg/kg n=9	0.7/0.4 mg/kg n=6
Subjects, n (%)					
Any AE	9 (81.8)	8 (88.9)	6 (75.0)	9 (100.0)	3 (50.0)
Any AE related to study drug	3 (27.3)	2 (22.2)	0 (0.0)	4 (44.4)	1 (16.7)
Any severe AE	3 (27.3)	2 (22.2)	2 (25.0)	2 (22.2)	0 (0.0)
Any severe AE related to study drug	0 (0.0)	1 (11.1)	0 (0.0)	0 (0.0)	0 (0.0)
≥1 serious AE	4 (36.4)	4 (44.4)	1 (12.5)	7 (77.8)	1 (16.7)
Death	2 (18.2)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)
AEs in ≥2 subjects in any treatment group n (%)					
Fatigue	2 (18.2)	2 (22.2)	0 (0.0)	1 (11.1)	1 (16.7)
Pain	1 (9.1)	2 (22.2)	0 (0.0)	0 (0.0)	0 (0.0)
Constipation	1 (9.1)	2 (22.2)	1 (12.5)	0 (0.0)	0 (0.0)
Nausea	1 (9.1)	1 (11.1)	1 (12.5)	3 (33.3)	0 (0.0)
Hypertension	2 (18.2)	4 (44.4)	0 (0.0)	5 (55.6)	0 (0.0)
Fall	2 (18.2)	1 (11.1)	0 (0.0)	1 (11.1)	0 (0.0)
Type 2 diabetes mellitus	0 (0.0)	0 (0.0)	0 (0.0)	2 (22.2)	0 (0.0)
Dizziness	0 (0.0)	0 (0.0)	0 (0.0)	2 (22.2)	0 (0.0)
Increased blood phosphorous	0 (0.0)	0 (0.0)	0 (0.0)	2 (22.2)	0 (0.0)

Home BP Measurements

- At the end of the first dose cycle, small changes from baseline in home systolic BP (SBP) and diastolic BP (DBP) measurements were generally similar among subjects receiving placebo or sotatercept 0.3 mg/kg, 0.5 mg/kg, 0.7 mg/kg, and 0.7/0.4 mg/kg in the safety population (mean change in SBP: –0.1, 5.2, 4.7, –1.4, and –4.0 mm Hg, respectively; mean change in DBP: 2.4, 3.6, 4.0, 2.4, and 0.3 mm Hg, respectively).
- Proportions of subjects with ≥20 mm Hg increase in SBP at any time during the study were 46%, 11%, 25%, 67%, and 33% in the placebo or sotatercept 0.3 mg/kg, 0.5 mg/kg, 0.7 mg/kg, and 0.7/0.4 mg/kg groups, respectively, and with ≥20 mm Hg increase in DBP at any time during study were 18%, 0%, 13%, 0%, and 0%, respectively.
- During the long-term (225-day) treatment phase, home BP measurements showed no consistent or dose-dependent change from baseline among subjects in any of the treatment groups.

Pharmacokinetics

- Sotatercept exhibited dose-dependent increases in the serum drug exposure, as measured by maximum plasma concentration and area under the concentration-vs.-time curve to infinity, with a mean elimination half-life of 21 to 29 days.

Efficacy

Hb Effects

- Mean changes from baseline to peak Hb response in the first 28 days for the placebo or sotatercept 0.3 mg/kg, 0.5 mg/kg, 0.7 mg/kg, and 0.7/0.4 mg/kg groups were –0.1, 0.5, 0.8, 0.8, and 0.5 g/dL, respectively.
- During the 225-day treatment phase, the proportions of subjects achieving Hb increase ≥1 g/dL, Hb >10 g/dL, or both Hb increase ≥1 g/dL and Hb >10 g/dL, or requiring rescue varied in relation to dose for the sotatercept 0.3 mg/kg, 0.5 mg/kg, and 0.7 mg/kg dose groups and the associated placebo-control subjects for which there are available long-term study data (**Figure 2**).
- The sotatercept 0.7/0.4 mg/kg dose group is ongoing and not included in this analysis; however, the mean time to rescue was similar between the 0.7/0.4 mg/kg dose group (4 subjects rescued, 100.3 days) and the 0.7 mg/kg dose group (5 subjects rescued, 99.4 days).

Figure 2. Proportions of Subjects With Hb Response or Rescue During the 225-Day Treatment Phase (FAS)*

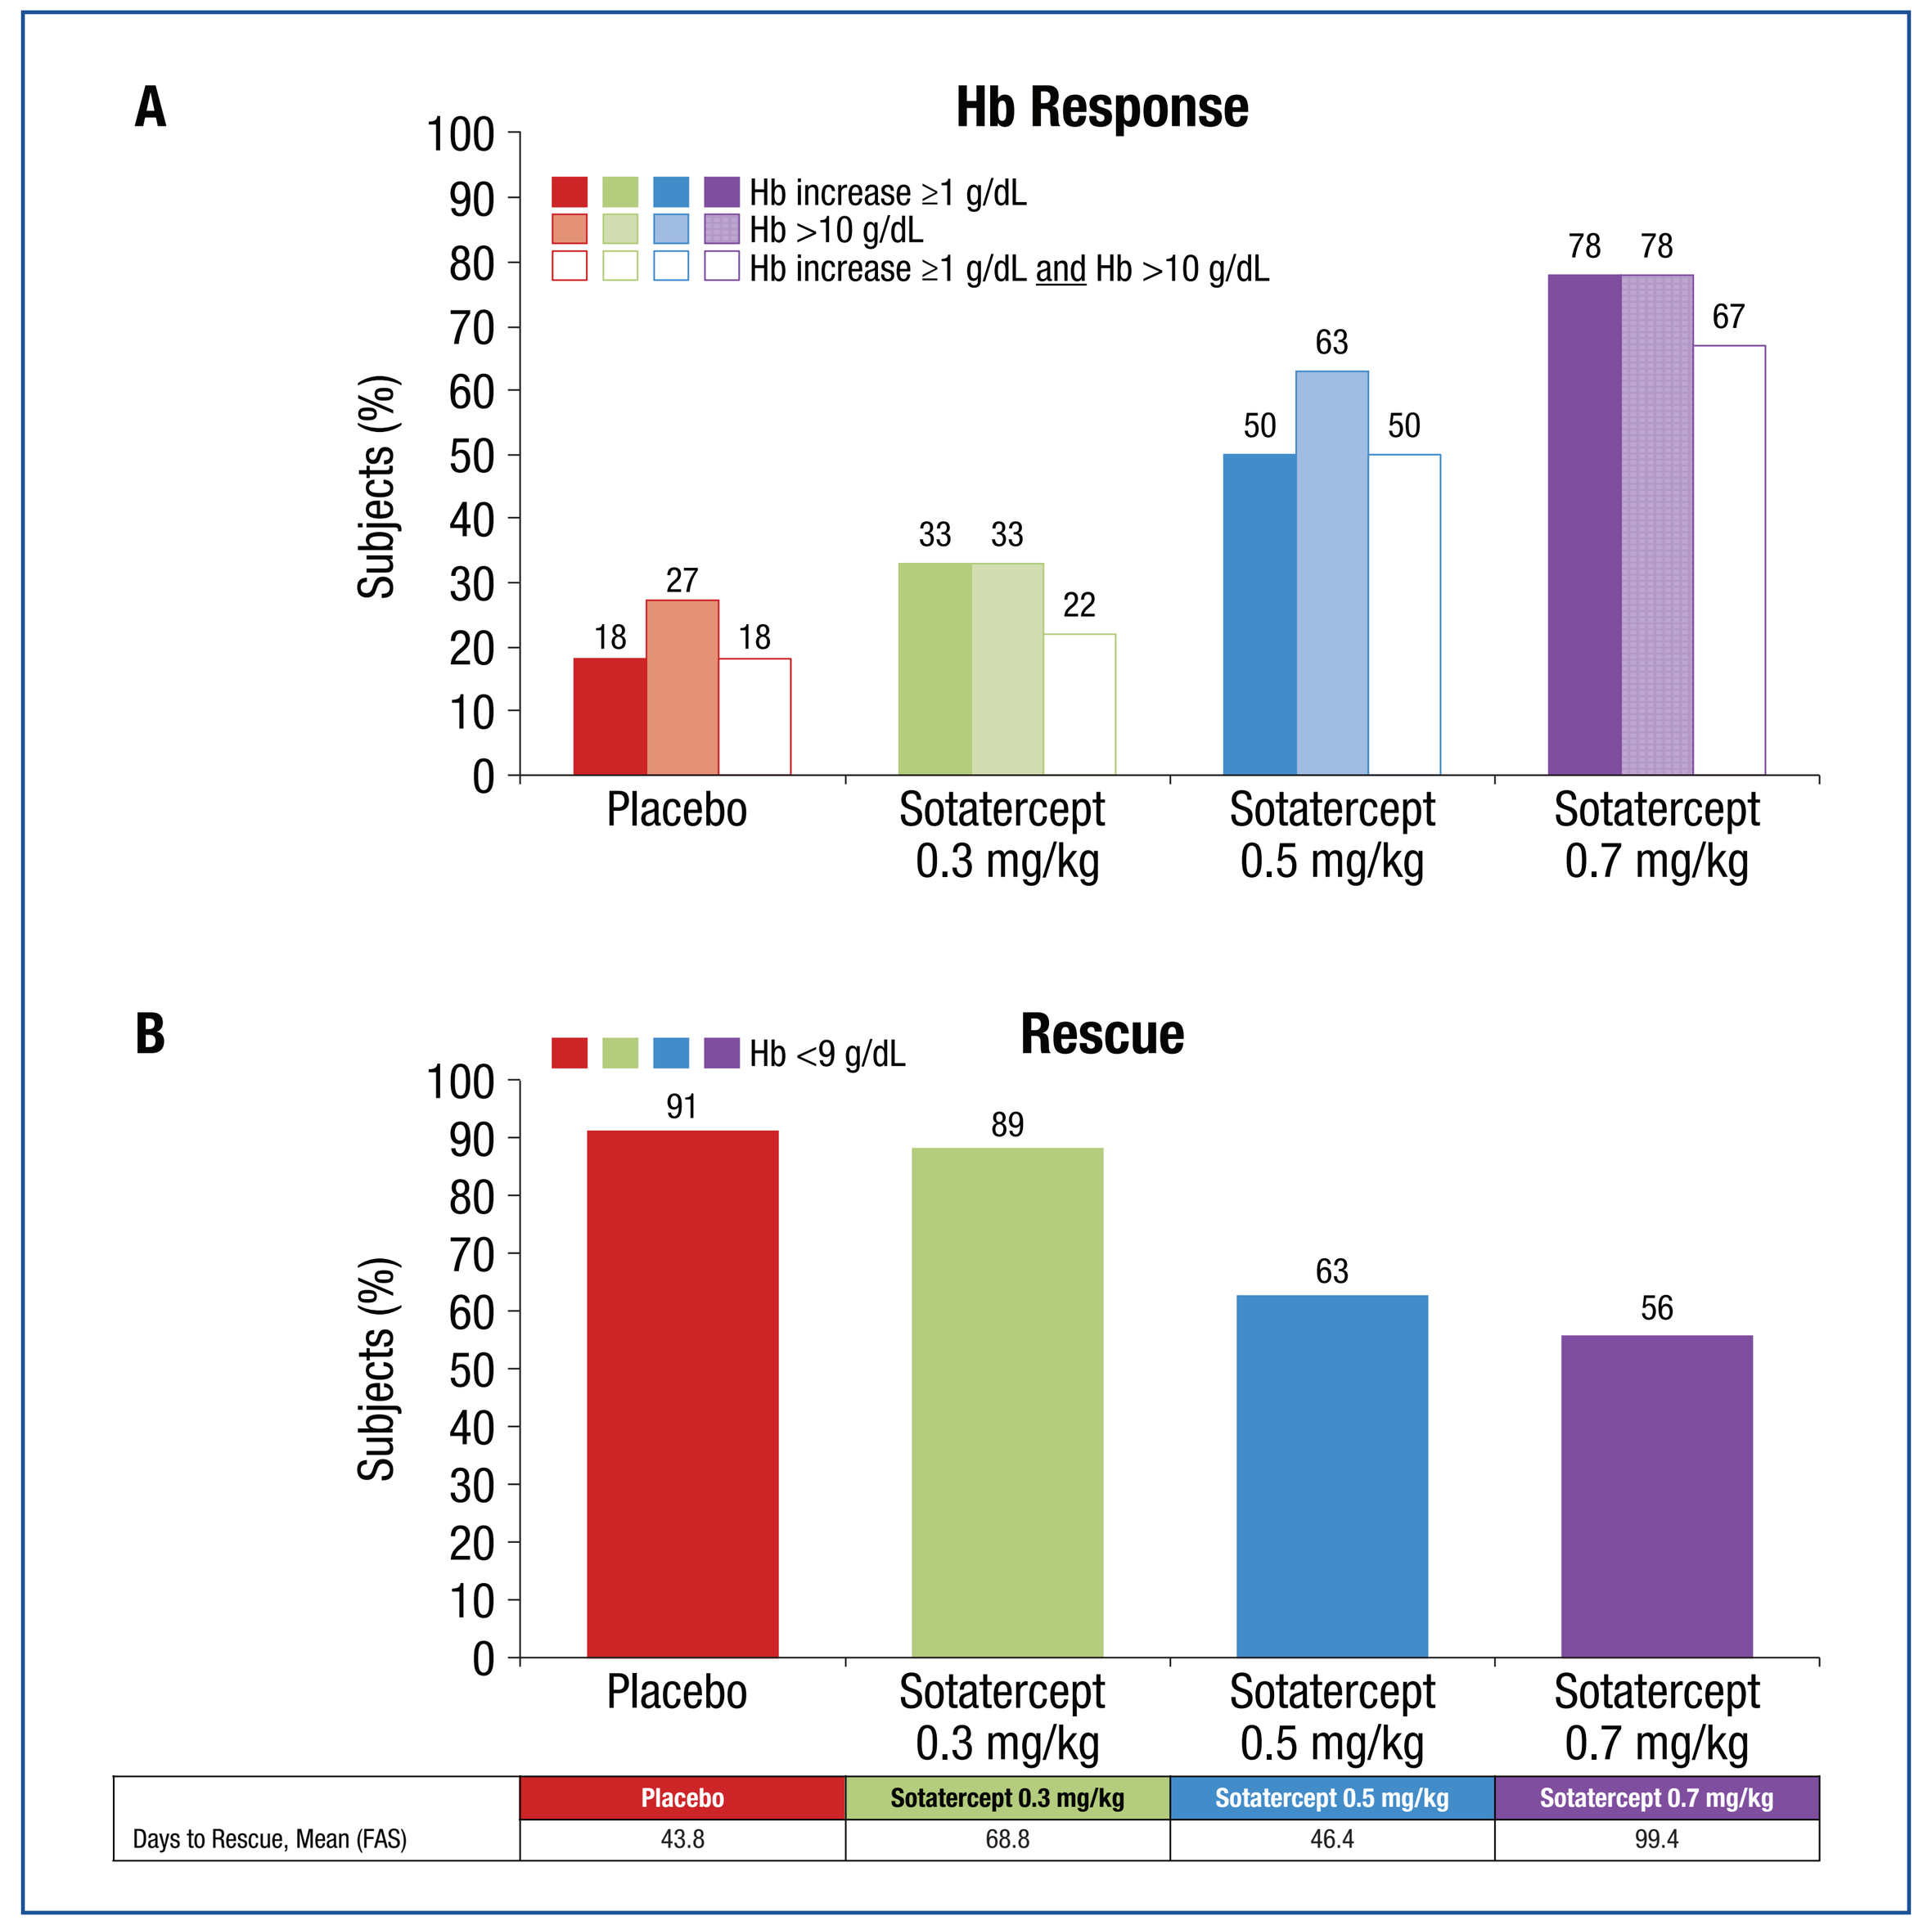
A
Hb Response
Hb increase ≥1 g/dL
Hb >10 g/dL
Hb increase ≥1 g/dL and Hb >10 g/dL
Subjects (%)
100 90 80 70 60 50 40 30 20 10 0
18 27 18
33 33 22
50 63 50
78 78 67
Placebo
Sotatercept 0.3 mg/kg
Sotatercept 0.5 mg/kg
Sotatercept 0.7 mg/kg
B
Rescue
Hb <9 g/dL
Subjects (%)
91 89 63 56
Placebo
Sotatercept 0.3 mg/kg
Sotatercept 0.5 mg/kg
Sotatercept 0.7 mg/kg
Days to Rescue, Mean (FAS)
Placebo 43.8
Sotatercept 0.3 mg/kg 68.8
Sotatercept 0.5 mg/kg 46.4
Sotatercept 0.7 mg/kg 99.4

*Values for Hb increase ≥1 g/dL, Hb >10 g/dL, or both Hb ≥1 g/dL and Hb >10 g/dL are censored for those who required rescue treatment. One placebo subject had elevated serum EPO levels, suggesting off-protocol EPO administration.

QCT VC Measurements

- The changes from baseline in abdominal aorta total Agatston scores are provided in **Table 5**. The proportion of subjects with <15% progression in abdominal aorta total Agatston scores is shown in **Figure 3**.

Table 5. Mean Baseline and Change From Baseline in Abdominal Aorta Total Agatston Scores and Square Root Transformed Total Volume Scores*

		Sotatercept		
	Placebo n=4	0.3 mg/kg n=6	0.5 mg/kg n=5	0.7 mg/kg n=6
Abdominal aorta total Agatston score				
Baseline total Agatston score	6,498.8	9,472.7	3,618.5	1,862.1
% Change from baseline total Agatston score	58.4	29.9	17.3	7.4
Square root transformed total volume score				
Baseline square root of total volume, mm³	39.0	46.2	33.1	27.4
Change from baseline square root of total volume, mm³	3.4	11.1	1.2	1.3

Note: The n reflects the number of randomized subjects with paired QCT assessments; actual number of subjects available for each parameter may vary.
*Lower total Agatston and square root transformed total volume scores indicate lower levels of VC.

Figure 3. Proportion of Subjects With <15% Progression in Abdominal Aorta Total Agatston Scores

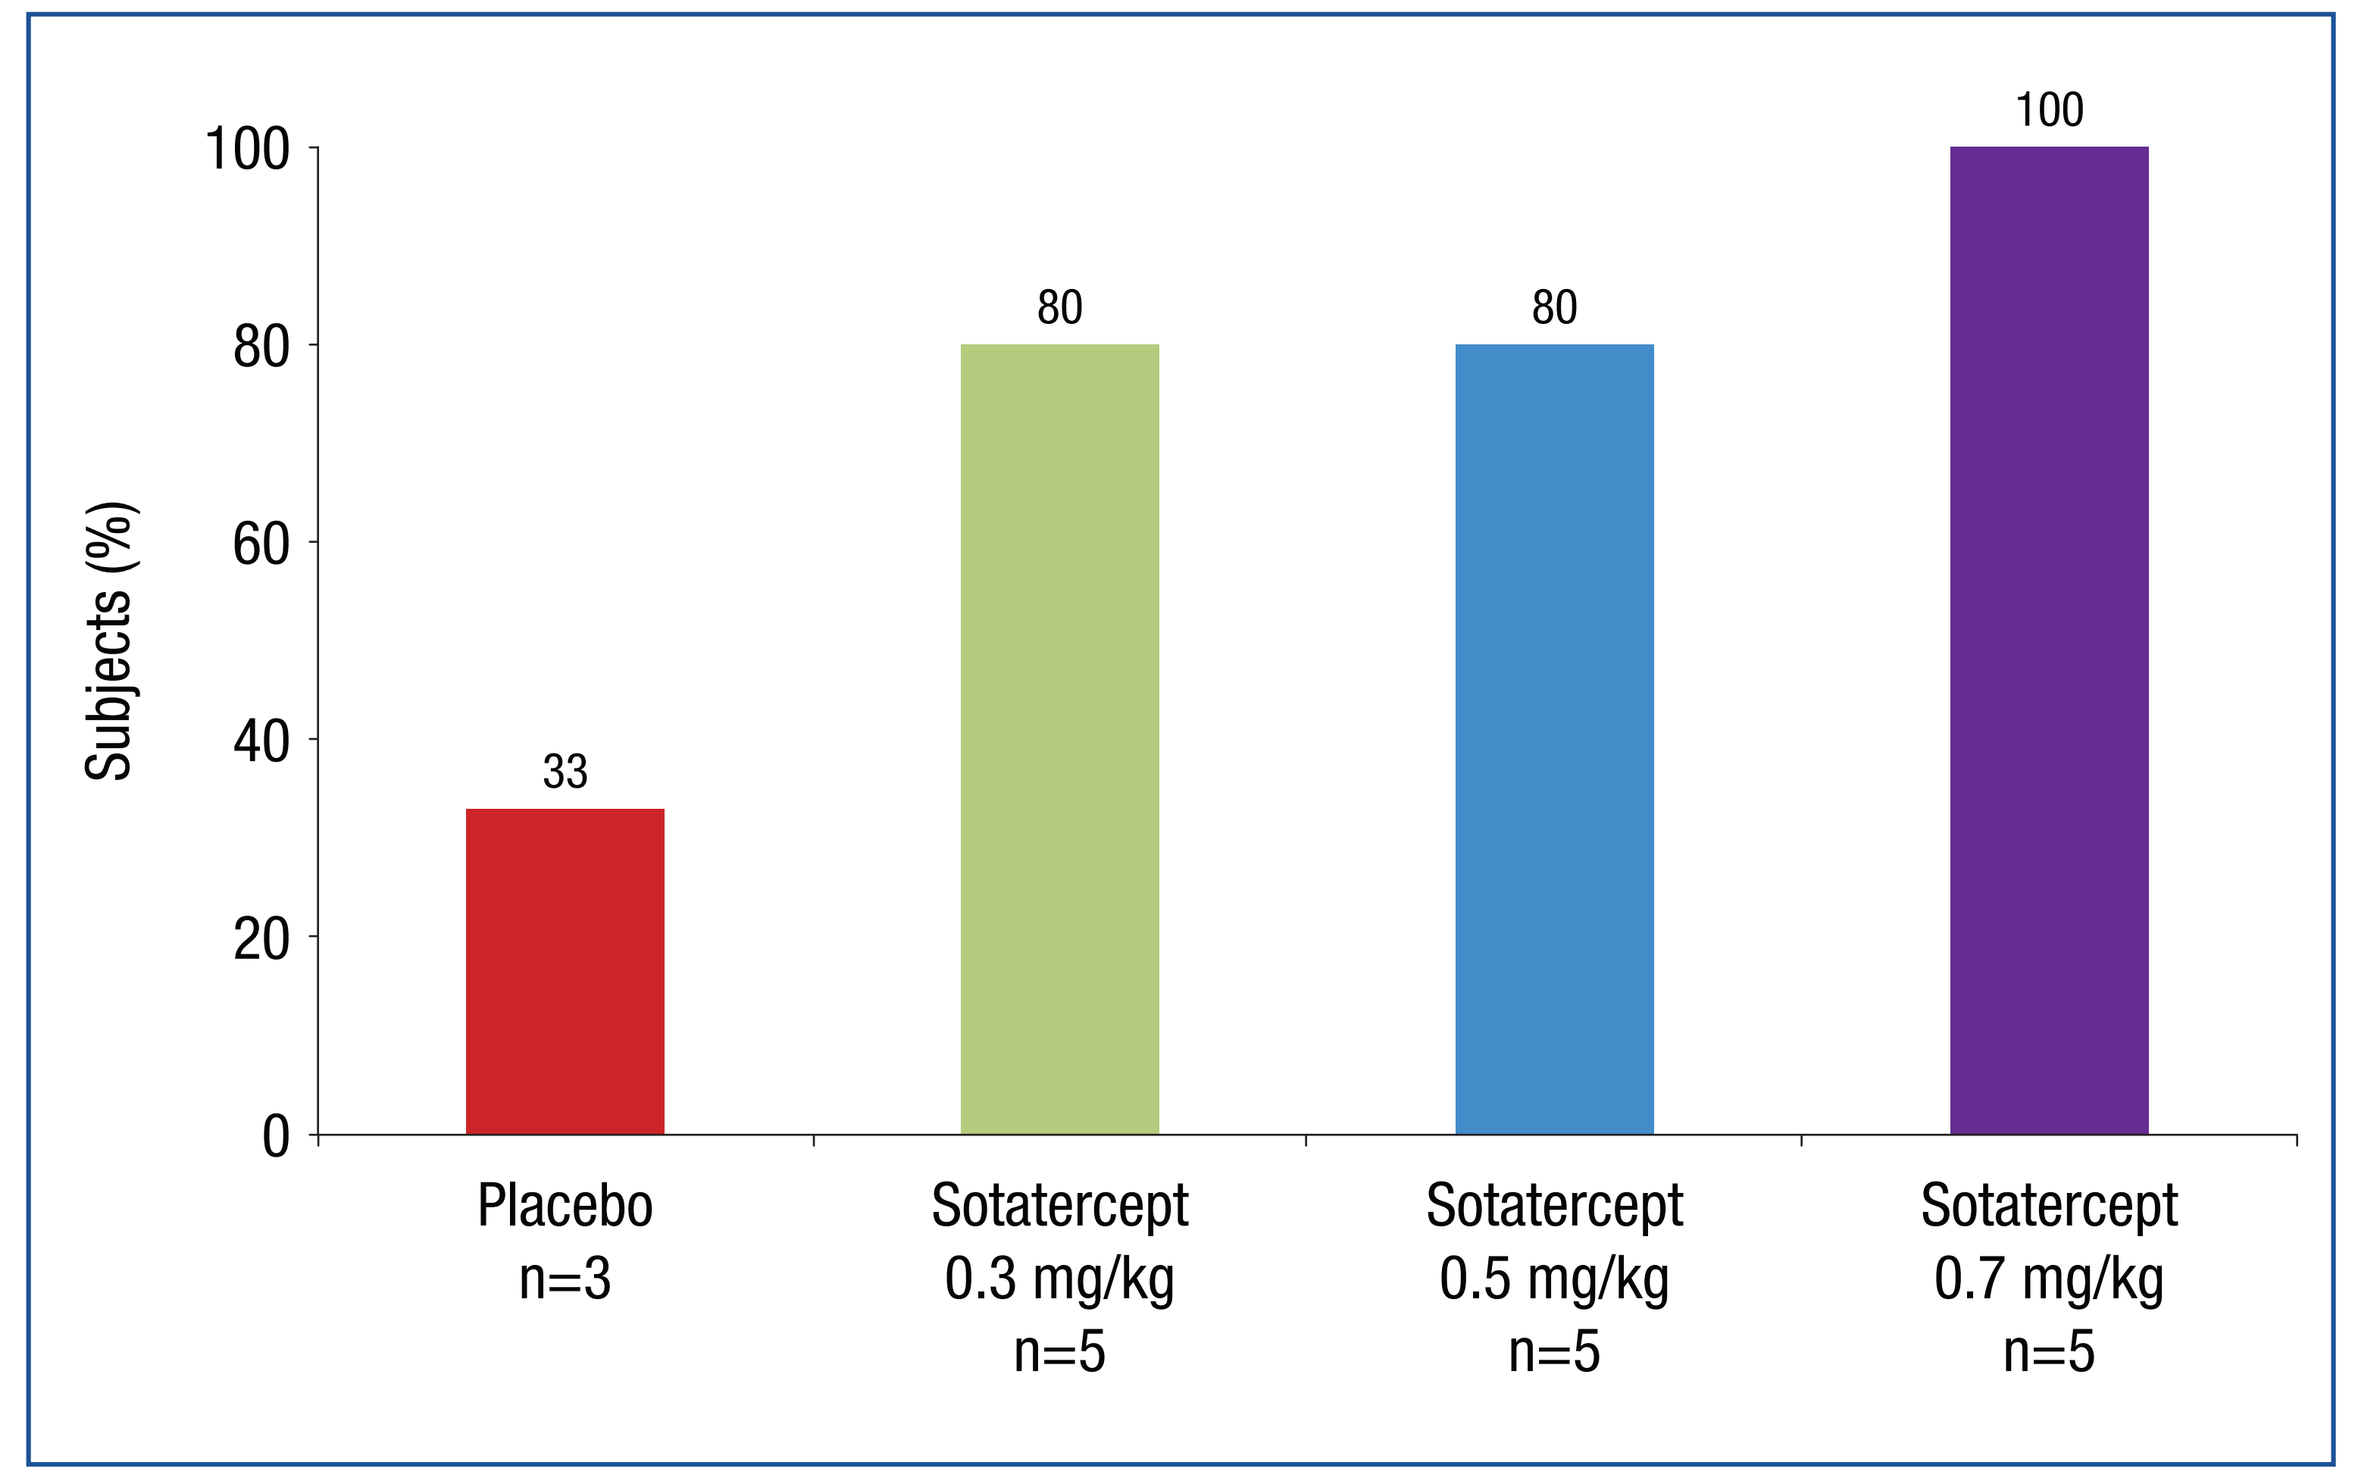
Subjects (%)
100 80 60 40 20 0
33 80 80 100
Placebo n=3
Sotatercept 0.3 mg/kg n=5
Sotatercept 0.5 mg/kg n=5
Sotatercept 0.7 mg/kg n=5

QCT Volumetric BMD Measurements

- **Table 6** provides the percent change from baseline in femoral neck cortical and mean lumbar spine trabecular volumetric BMD measurements.
- The proportion of subjects with >2% gain in femoral neck cortical BMD was determined.[8] Sotatercept treatment was associated with increases in the proportion of subjects with a >2% increase in femoral neck cortical BMD (**Table 6**; **Figure 4**).
- In high-turnover ROD, trabecular bone mass increases, as measured by lumbar spine trabecular BMD, without a reduction in vertebral fracture rates in ESKD compared with the general population.[9,10] In this setting, the increased trabecular bone mass is of poor quality.[11] Sotatercept appears to slow the increase in trabecular bone mass in the lumbar spine (**Table 6**).

Table 6. Mean Baseline and Percent Change From Baseline in Femoral Neck Cortical and Mean Lumbar Spine (L1, L2) Trabecular BMD

		Sotatercept		
	Placebo n=4	0.3 mg/kg n=6	0.5 mg/kg n=5	0.7 mg/kg n=6
Femoral neck cortical BMD				
Baseline BMD, mg/cm³	640.5	667.0	593.1	594.8
% Change from baseline BMD (n)	–1.4 (4)	–1.4 (5)	1.6 (5)	–0.1 (4)
Lumbar spine (L1, L2) trabecular BMD				
Baseline BMD, mg/cm³	140.1	125.6	149.1	118.7
% Change from baseline BMD (n)	10.9 (4)	8.0 (6)	0.5 (5)	4.5 (6)

Note: The n reflects the number of randomized subjects with paired QCT assessments; actual number of subjects available for each parameter may vary.

Figure 4. Proportions of Subjects With >2% Increase in Femoral Neck Cortical BMD

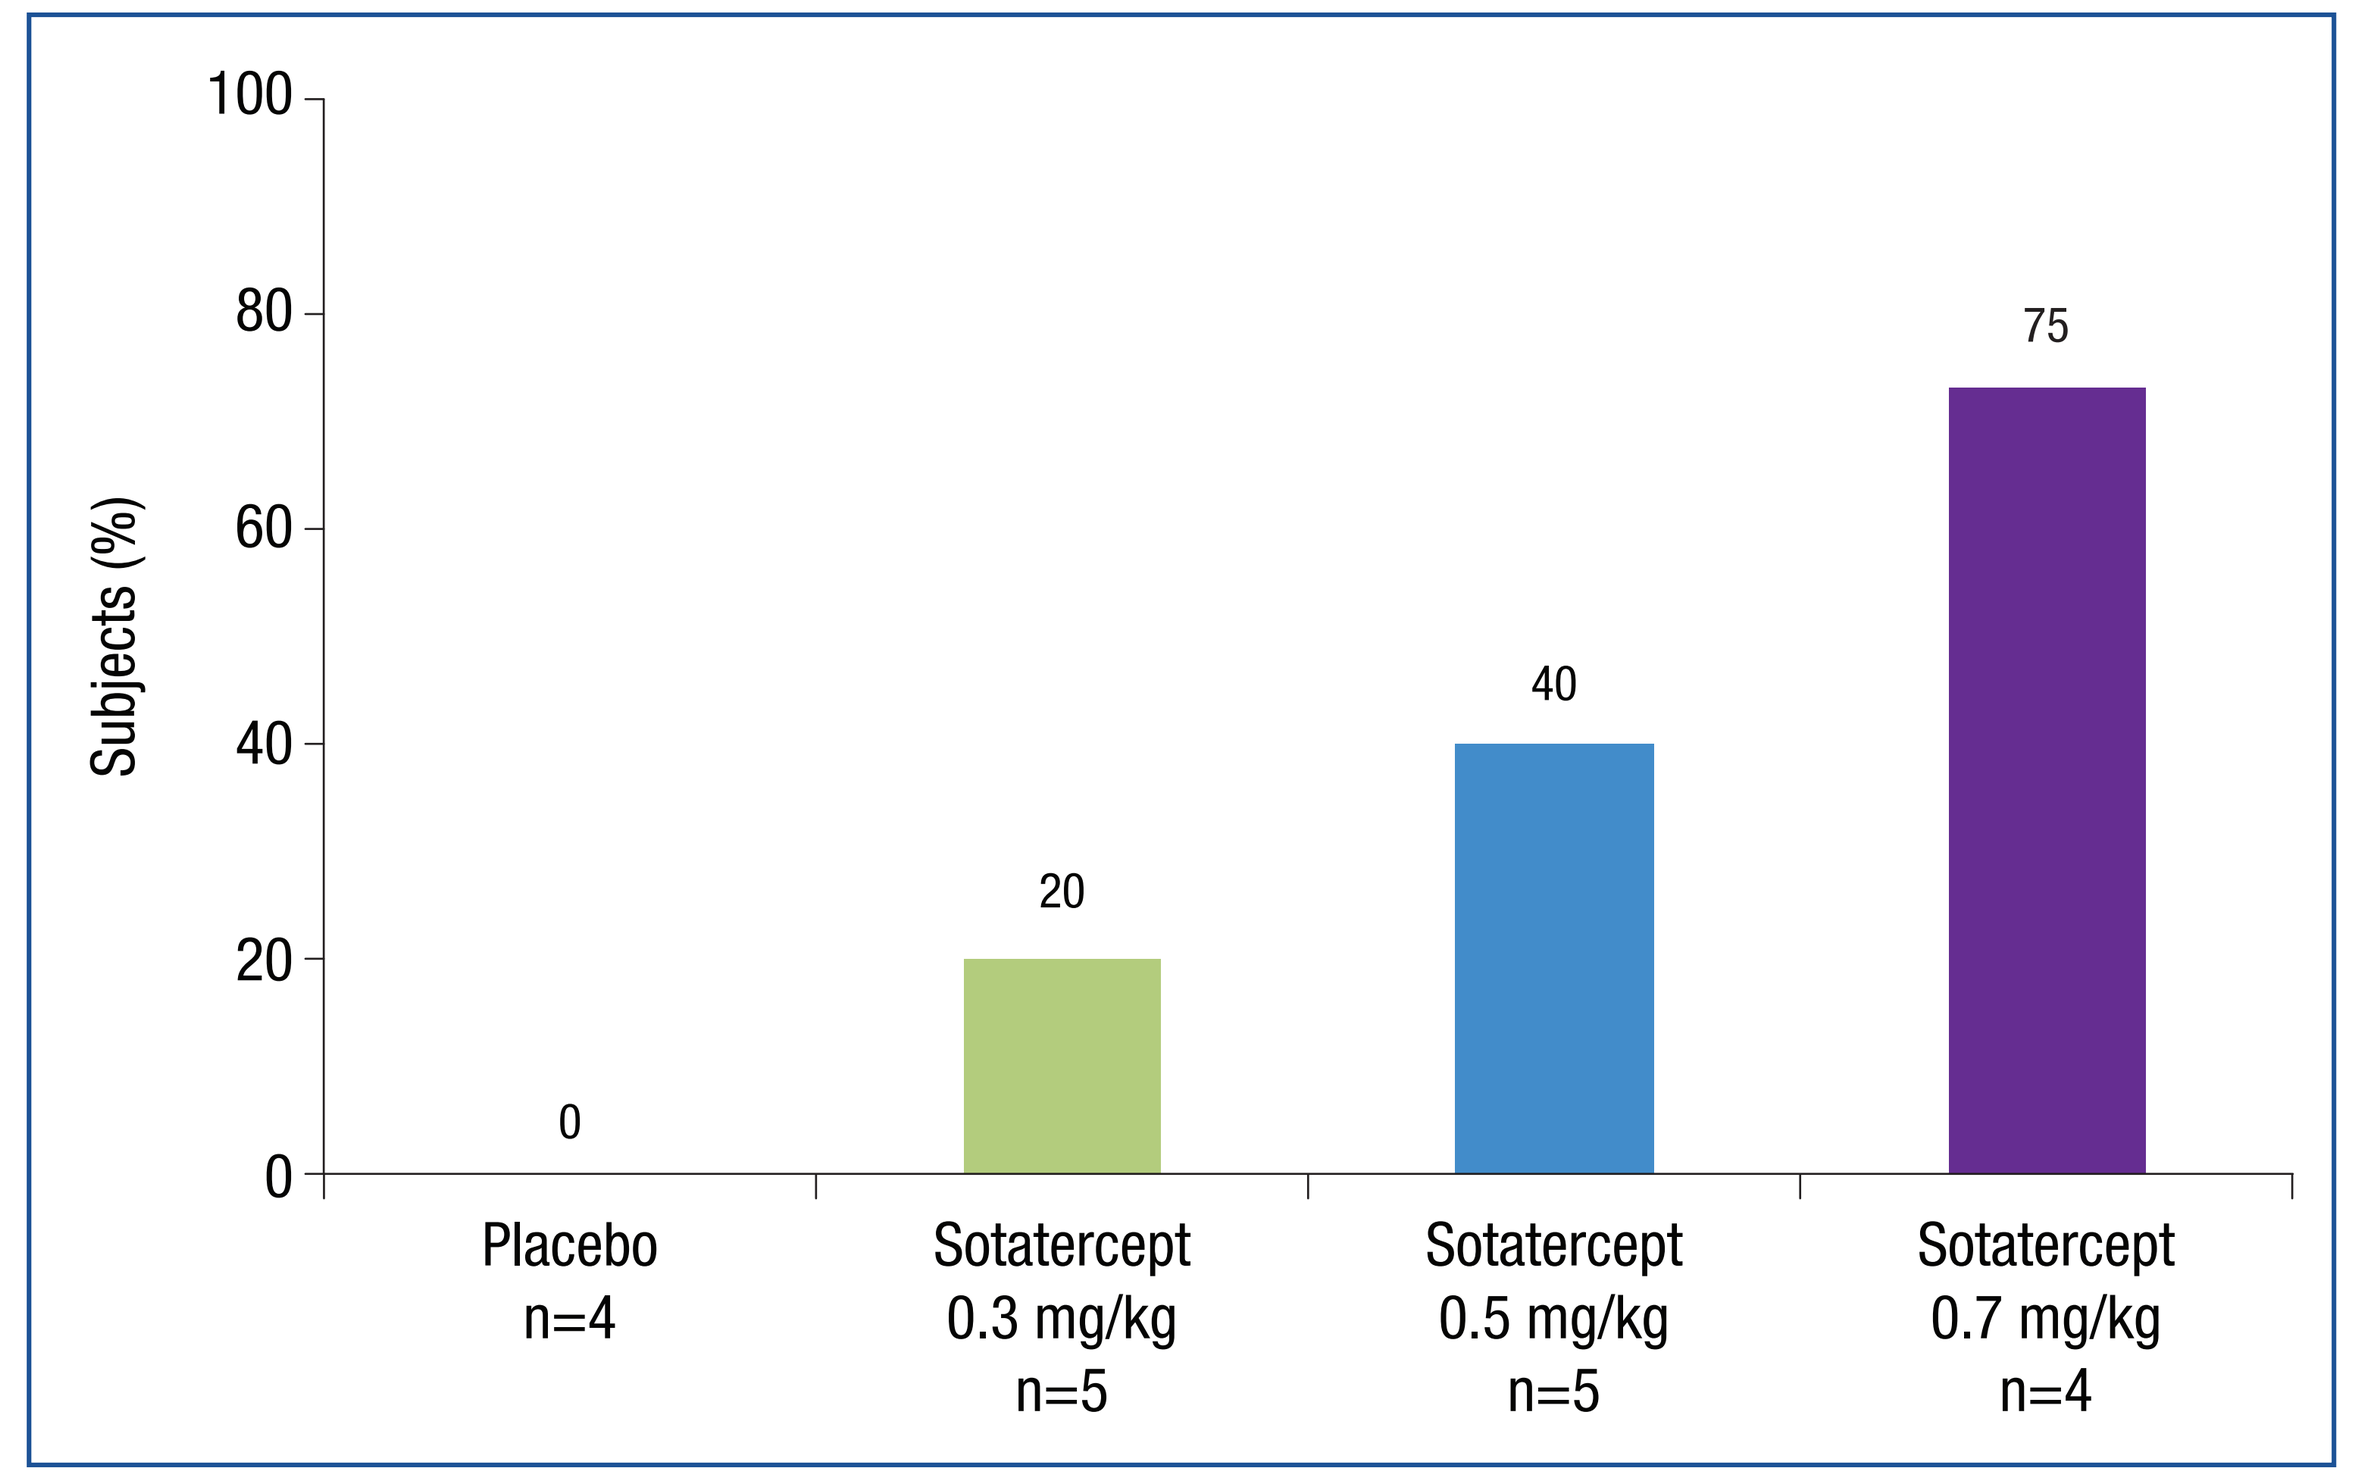
Subjects (%)
100 80 60 40 20 0
0 20 40 75
Placebo n=4
Sotatercept 0.3 mg/kg n=5
Sotatercept 0.5 mg/kg n=5
Sotatercept 0.7 mg/kg n=4

CONCLUSIONS

- Sotatercept was well tolerated over 225 days (up to eight 28-day dose cycles), with an acceptable safety profile in subjects with ESKD on HD and without increases in home BP. In subjects given sotatercept 0.7/0.4 mg/kg on a 14-day dose cycle, cumulative safety appeared to be consistent with the other dose groups, although data are still accruing.
- Sotatercept treatment with 0.3 mg/kg, 0.5 mg/kg, and 0.7 mg/kg doses every 28 days was associated with dose-dependent Hb improvements (achieving target increase ≥1 g/dL and target range 10–12 g/dL) that were generally sustained over multiple dosing cycles. An ongoing dose group with 14-day dosing (0.7/0.4 mg/kg) did not improve Hb efficacy in the first 28 days; however, time to rescue was similar to that in the 0.7 mg/kg dose group.
- In this small interim dataset evaluating exploratory QCT data measured at baseline and Day 225 (after up to eight 28-day dose cycles), there was an apparent effect of sotatercept on multiple parameters of CKD-MBD, including slowing progression of VC, increasing femoral neck cortical bone mass, and slowing the increase in lumbar spine bone mass.
 - Differences between the groups at baseline may have influenced these results. The placebo group exhibited higher baseline serum levels of bone turnover markers and higher Agatston scores indicative of more severe ROD and VC, potentially increasing the chance of observing differences on these measures between the placebo group and the sotatercept groups.
- Sotatercept may address significant unmet needs in the treatment of anemia and CKD-MBD in patients with ESKD.
 - Additional data from the ongoing sotatercept 0.7/0.4 mg/kg dose group are expected to further substantiate the efficacy and safety findings observed in the other dose groups.

REFERENCES

1. McGonigle RJ, Wallin JD, Shadduck RK, et al. Erythropoietin deficiency and inhibition of erythropoiesis in renal insufficiency. *Kidney Int.* 1984;25:437-444.
2. Kidney Disease: Improving Global Outcomes (KDIGO) CKD-MBD Work Group. KDIGO clinical practice guideline for the diagnosis, evaluation, prevention, and treatment of chronic kidney disease-mineral and bone disorder (CKD-MBD). *Kidney Int Suppl.* 2009:S1-S130.
3. Iancu-Rubin C, Mosoyan G, Wang J, et al. Stromal cell-mediated inhibition of erythropoiesis can be attenuated by sotatercept (ACE-011), an activin receptor type II ligand trap. *Exp Hematol.* 2013;41:155-166.
4. Carrancio S, Markovics J, Wong P, et al. An activin receptor IIA ligand trap promotes erythropoiesis resulting in a rapid induction of red blood cells and haemoglobin. *Br J Haematol.* 2014;165:870-882.
5. Hruska KA, Agapova OA, Fang F, et al. Chronic kidney disease (CKD) stimulates activin and endothelial to mesenchymal transition (EnMT), which causes vascular calcification and is inhibited by an activin ligand trap [abstract SA-OR063]. *J Am Soc Nephrol.* 2014;25(Suppl):95A.
6. Wooldridge T, Kaplan M, Alcorn H Jr, et al. The pharmacokinetics and safety of a single-dose of sotatercept (ACE-011) in subjects on hemodialysis and the effects of its murine analog (RAP-011) on anemia and in preventing bone loss in C57BL/6 mice with 5/6 nephrectomy [oral presentation SA-OR087]. Presented at: the American Society of Nephrology Kidney Week 2012 Annual Meeting; October 30-November 4, 2012; San Diego, CA.
7. Smith WT, Havill J, Kopyt N, et al. Long-term effects of sotatercept compared with placebo for correction of anemia in hemodialysis subjects: interim analysis of ACE-011-REN-001 [poster FP661]. Presented at: the Annual Congress of the European Renal Association - European Dialysis and Transplant Association; May 28-31, 2015; London, UK.
8. Malluche HH, Davenport DL, Cantor T, et al. Bone mineral density and serum biochemical predictors of bone loss in patients with CKD on dialysis. *Clin J Am Soc Nephrol.* 2014;9:1254-1262.
9. Leonard MB. A structural approach to skeletal fragility in chronic kidney disease. *Semin Nephrol.* 2009;29:133-143.
10. Duan Y, De Luca V, Seeman E. Parathyroid hormone deficiency and excess: similar effects on trabecular bone but differing effects on cortical bone. *J Clin Endocrinol Metab.* 1999;84:718-722.
11. Malluche HH, Porter DS, Monier-Faugere MC, et al. Differences in bone quality in low- and high-turnover renal osteodystrophy. *J Am Soc Nephrol.* 2012;23:525-532.

This study was sponsored by Celgene Corporation.

Presented at: the American Society of Nephrology (ASN) Kidney Week 2015 Annual Meeting; November 5–8, 2015; San Diego, CA.

Scan this QR code to receive a PDF of the poster