Assessment of longer-term efficacy and safety in the phase 3 BELIEVE trial of luspatercept to treat anemia in patients with β-thalassemia

Abstract Number
EP1548

Scientific Content on Demand
To request a copy of this poster:
Scan QR code
via a barcode reader application
This QR code will take you to an external site.
QR codes are valid for 120 days after the congress presentation date.

Ali T. Taher,[1] Vip Viprakasit,[2] Maria Domenica Cappellini,[3] Olivier Hermine,[4,5] Pencho Georgiev,[6,7] Kevin H. M. Kuo,[8] Thomas Coates,[9,10] Ersi Voskaridou,[11] Hong Keng Liew,[12] Idit Pazgal-Kobrowski,[13] Gian Luca Forni,[14] Silverio Perrotta,[15] Abderrahim Khelif,[16] Ashutosh Lal,[17] Antonis Kattamis,[18] Farrukh Shah,[19] John Porter,[20] Abderrahmane Laadem,[21] Jeevan K. Shetty,[22] Wen-Ling Kuo,[21] Jennie Zhang,[21] Dimana Miteva,[22] Tatiana Zinger,[22] Daniel Sinsimer,[21] Chrystal U. Louis,[21] Peter G. Linde,[23] Antonio Piga[24]

[1]Department of Internal Medicine, American University of Beirut Medical Center, Beirut, Lebanon; [2]Siriraj Hospital, Mahidol University, Bangkok, Thailand; [3]Fondazione IRCCS Ca' Granda Policlinico Hospital, University of Milan, Milan, Italy; [4]Imagine Institute, INSERM U1163, University of Paris, Paris, France; [5]Department of Hematology, Necker Hospital, Assistance Publique-Hôpitaux de Paris, Paris, France; [6]St George University Hospital for Active Treatment, Plovdiv, Bulgaria; [7]Medical University of Plovdiv, Plovdiv, Bulgaria; [8]Division of Medical Oncology and Hematology, Department of Medicine, University Health Network and Division of Hematology, Department of Medicine, University of Toronto, Toronto, ON, Canada; [9]Children's Center for Cancer and Blood Diseases, Children's Hospital Los Angeles, Los Angeles, CA, USA; [10]USC Keck School of Medicine, Los Angeles, CA, USA; [11]Thalassemia and Sickle Cell Center of Laiko General Hospital, Athens, Greece; [12]Hospital Sultanah Bahiyah, Alor Setar, Malaysia; [13]Comprehensive Center of Thalassemia, Rabin Medical Center, Beilinson Hospital, Petah Tikva, Israel; [14]Centro della Microcitemia e Anemie Congenite e del Dismetabolismo del Ferro, Ospedale Galliera, Genoa, Italy; [15]Università della Campania, Luigi Vanvitelli, Caserta, Italy; [16]Farhat Hached Teaching Hospital, Sousse University, Tunisia; [17]University of California San Francisco Benioff Children's Hospital, Oakland, CA, USA; [18]First Department of Pediatrics, National and Kapodistrian University of Athens, Athens, Greece; [19]Department of Haematology, Whittington Health NHS Trust, London, UK; [20]University College London, University College London Hospitals, London, UK; [21]Bristol Myers Squibb, Princeton, NJ, USA; [22]Celgene International, a Bristol-Myers Squibb Company, Boudry, Switzerland; [23]Acceleron Pharma, Cambridge, MA, USA; [24]Department of Clinical and Biological Sciences, University of Turin, Turin, Italy

Introduction

- β-thalassemia is a genetic blood disorder associated with ineffective erythropoiesis and anemia[1]
- Patients with β-thalassemia who are transfusion dependent require frequent and lifelong red blood cell (RBC) transfusions and iron chelation therapy (ICT)[2]
- Luspatercept is a first-in-class erythroid maturation agent approved by the US Food and Drug Administration for the treatment of anemia in adult patients with β-thalassemia who require regular RBC transfusions[3,4]
 - Luspatercept binds to select transforming growth factor-β superfamily ligands to diminish Smad2/3 signaling and enhance late-stage erythropoiesis[5]
- The BELIEVE study is an ongoing phase 3, randomized, double-blind, placebo-controlled, multicenter trial of luspatercept in adults who require regular RBC transfusions due to β-thalassemia[6]
 - The primary results of the BELIEVE trial showed that a statistically significant proportion of patients treated with luspatercept achieved ≥ 33% reduction in RBC transfusion burden, with a reduction of ≥ 2 RBC units from Weeks 13–24 versus patients treated with placebo

Objective

- To evaluate long-term efficacy and safety of luspatercept treatment in adult patients with β-thalassemia in the BELIEVE trial

Methods

Study design

- BELIEVE is an ongoing phase 3, randomized, double-blind, placebo-controlled, multicenter trial (NCT02604433) (**Figure 1**)

Figure 1. Study design of the BELIEVE trial

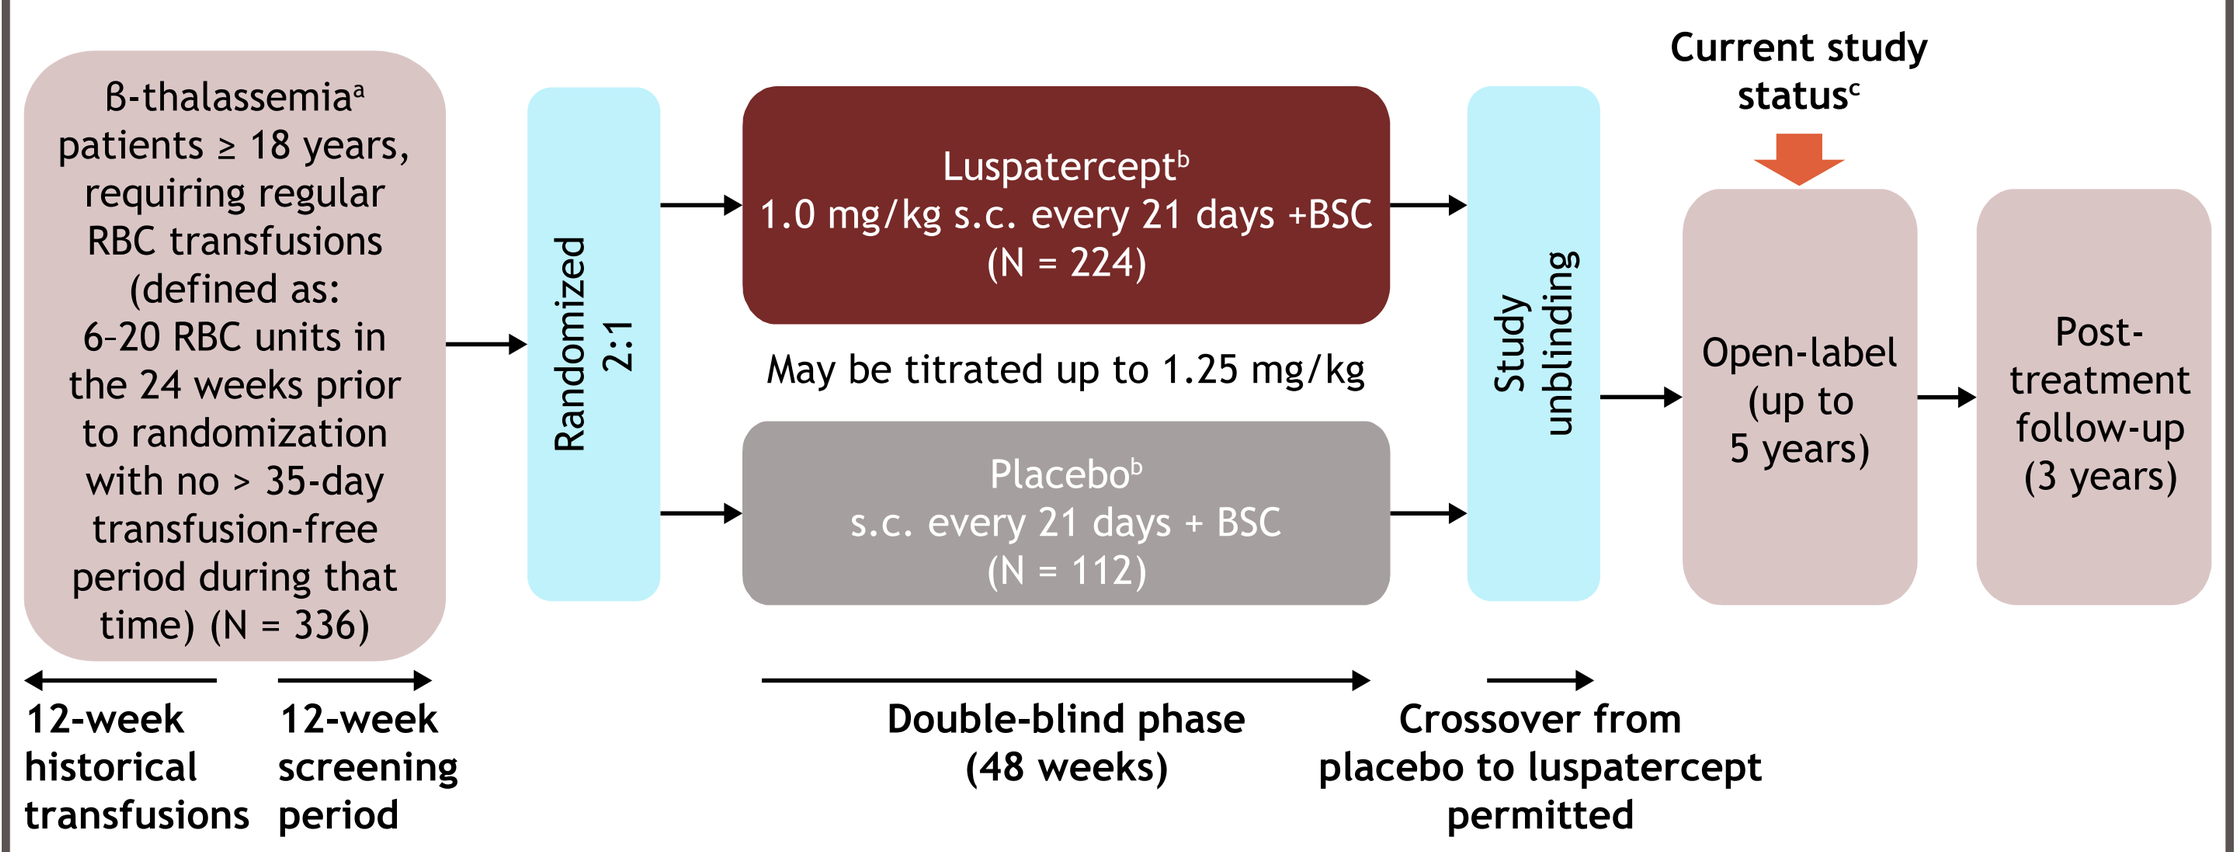
β-thalassemia[a] patients ≥ 18 years, requiring regular RBC transfusions (defined as: 6-20 RBC units in the 24 weeks prior to randomization with no > 35-day transfusion-free period during that time) (N = 336)
Randomized 2:1
Luspatercept[b] 1.0 mg/kg s.c. every 21 days +BSC (N = 224)
May be titrated up to 1.25 mg/kg
Placebo[b] s.c. every 21 days + BSC (N = 112)
Study unblinding
Current study status[c]
Open-label (up to 5 years)
Post-treatment follow-up (3 years)
12-week historical transfusions
12-week screening period
Double-blind phase (48 weeks)
Crossover from placebo to luspatercept permitted

Data cutoff used for this analysis was July 1, 2019.
[a]β-thalassemia or Hb E/β-thalassemia (compound β-thalassemia with mutation and/or multiplication of α-globin genes was allowed). [b]Patients could receive RBC transfusions and ICT to maintain their baseline Hb level. [c]The trial is fully enrolled and patients continue to receive treatment or follow-up.
BSC, best supportive care; Hb, hemoglobin; ICT, iron chelation therapy; RBC, red blood cell; s.c., subcutaneously.

- Patients were randomized at 65 sites between July 2016 and June 2017
- Eligible patients were aged ≥ 18 years and:
 - Had β-thalassemia or hemoglobin (Hb) E/β-thalassemia (compound β-thalassemia mutation and/or multiplication of α-globin genes was allowed)
 - Required regular transfusions (6–20 RBC units in the 24 weeks prior to randomization with no transfusion-free period > 35 days)
- Patients were randomized 2:1 to luspatercept 1.0 mg/kg (titration up to 1.25 mg/kg allowed) or placebo, administered subcutaneously every 21 days for ≥ 48 weeks; all patients continued to receive best supportive care, including RBC transfusions to maintain target pre-transfusion Hb levels and ICT
- After study unblinding, patients randomized to receive placebo were eligible to cross over to receive luspatercept in an open-label phase

Longer-term efficacy analysis endpoints

- Achievement of ≥ 33% or ≥ 50% reduction in RBC transfusion burden during any 12, 24, or 48 weeks up to the updated data cutoff of July 1, 2019
- Median duration of the longest single period of ≥ 33% and ≥ 50% reduction in RBC transfusion burden

Crossover patients analysis endpoints

- Achievement of ≥ 33% or ≥ 50% reduction in RBC transfusion burden during Weeks 13–24 after crossover
- Achievement of ≥ 33% or ≥ 50% reduction in RBC transfusion burden during any 12 or 24 weeks after crossover up to data cutoff (July 1, 2019)

Safety endpoints

- Treatment-emergent adverse events (TEAEs) of interest
- Serious AEs
- Thromboembolic/thrombophlebitis events

Results

Patients

- The intent-to-treat population (N = 336) comprised all randomized patients, regardless of whether or not the patient received the study drug, and included 224 patients in the luspatercept arm and 112 patients in the placebo arm (**Table 1**)

Table 1. Baseline characteristics of the BELIEVE ITT population

Characteristic[a]	Luspatercept (N = 224)	Placebo[b] (N = 112)
Age, median (range), years	30 (18-66)	30 (18-59)
Female, n (%)	132 (58.9)	63 (56.3)
Hb (24 weeks), median (range), g/dL[c]	9.31 (4.5-11.4)	9.15 (5.8-11.7)
RBC transfusion burden, median (range), units/12 weeks	6.1 (3-14)	6.3 (3-12)
RBC transfusion burden, median (range), units/24 weeks	14 (6-24)	15 (6-26)
Splenectomy, n (%)	129 (57.6)	65 (58.0)
Serum ferritin, mean (SD), µg/L	2,096.9 (1,756.6)	1,845.1 (1,669.1)
LIC, mean (SD), mg/g dw	12.0 (14.8)	10.1 (11.5)
> 7 mg/g dw, n (%)	103 (46.0)	45 (40.2)
Myocardial iron by T2* MRI, mean (SD), ms	33.5 (16.2)	34.8 (10.7)

[a]Data on endocrine function were not collected. [b]Patients in the placebo arm were evaluated only prior to open-label phase. [c]Defined as the mean of all documented pre-transfusion Hb values during the 24 weeks prior to first dose for each patient. dw, dry weight; Hb, hemoglobin; ITT, intent to treat; LIC, liver iron concentration; RBC, red blood cell; SD, standard deviation; T2* MRI, T2-weighted magnetic resonance imaging.

- The safety population consisted of all patients who were randomized and received ≥ 1 dose of the study drug and included 223 patients in the luspatercept arm and 109 patients in the placebo arm
- The crossover population consisted of 92 of 112 (82.1%) patients randomized to the placebo arm who crossed over to receive luspatercept during the open-label phase

Long-term efficacy

- As of the updated data cutoff of July 1, 2019 the median treatment duration for patients in the luspatercept, crossover, and placebo arms was 119.1, 45.9, and 74.7 weeks, respectively (**Table 2**)
- Patients in the luspatercept, crossover, and placebo arms received, respectively, a mean of 33.1, 13.6, and 22.4 doses of study drug (**Table 2**)
- 171 (76.3%), 109 (48.7%), and 62 (27.7%) of 224 luspatercept-treated patients achieved ≥ 33% reduction in RBC transfusion burden during any 12, 24, and 48 weeks, respectively (**Figure 2A**)
- 107 (47.8%), 48 (21.4%), and 28 (12.5%) luspatercept-treated patients achieved ≥ 50% reduction in RBC transfusion burden during any 12, 24, and 48 weeks, respectively (**Figure 2B**)
- The median longest single period of ≥ 33% reduction in RBC transfusion was 110 and 90 days in the luspatercept and placebo arms, respectively (**Table 3**)
- The median longest single period of ≥ 50% reduction in RBC transfusion was 99 and 86 days in the luspatercept and placebo arms, respectively (**Table 3**)

Table 2. Treatment exposure in the BELIEVE safety population

Characteristic	Luspatercept (N = 223)	Placebo crossover to luspatercept (N = 92)	Placebo[a] (N = 109)
Treatment duration, median (range), weeks[b]	119.1 (1.7-153.1)	45.9 (6.1-47.7)	74.7 (8.9-104.0)
Number of doses received per patient, mean (range)	33.1 (1.0-51.0)	13.6 (2.0-17.0)	22.4 (3.0-34.0)
Maximum dose received, n (%)			
1.0 mg/kg	109 (48.9)	49 (53.3)	37 (33.9)
1.25 mg/kg	114 (51.1)	43 (46.7)	72 (66.1)

[a]Patients in the placebo arm are evaluated only prior to the open-label phase. [b]Duration is measured in total person years (defined as the sum of treatment duration in years for all patients in the treatment arm).

Figure 2. Patients achieving ≥ 33% (A) or ≥ 50% (B) reduction in RBC transfusion burden from baseline during any 12, 24, or 48 weeks

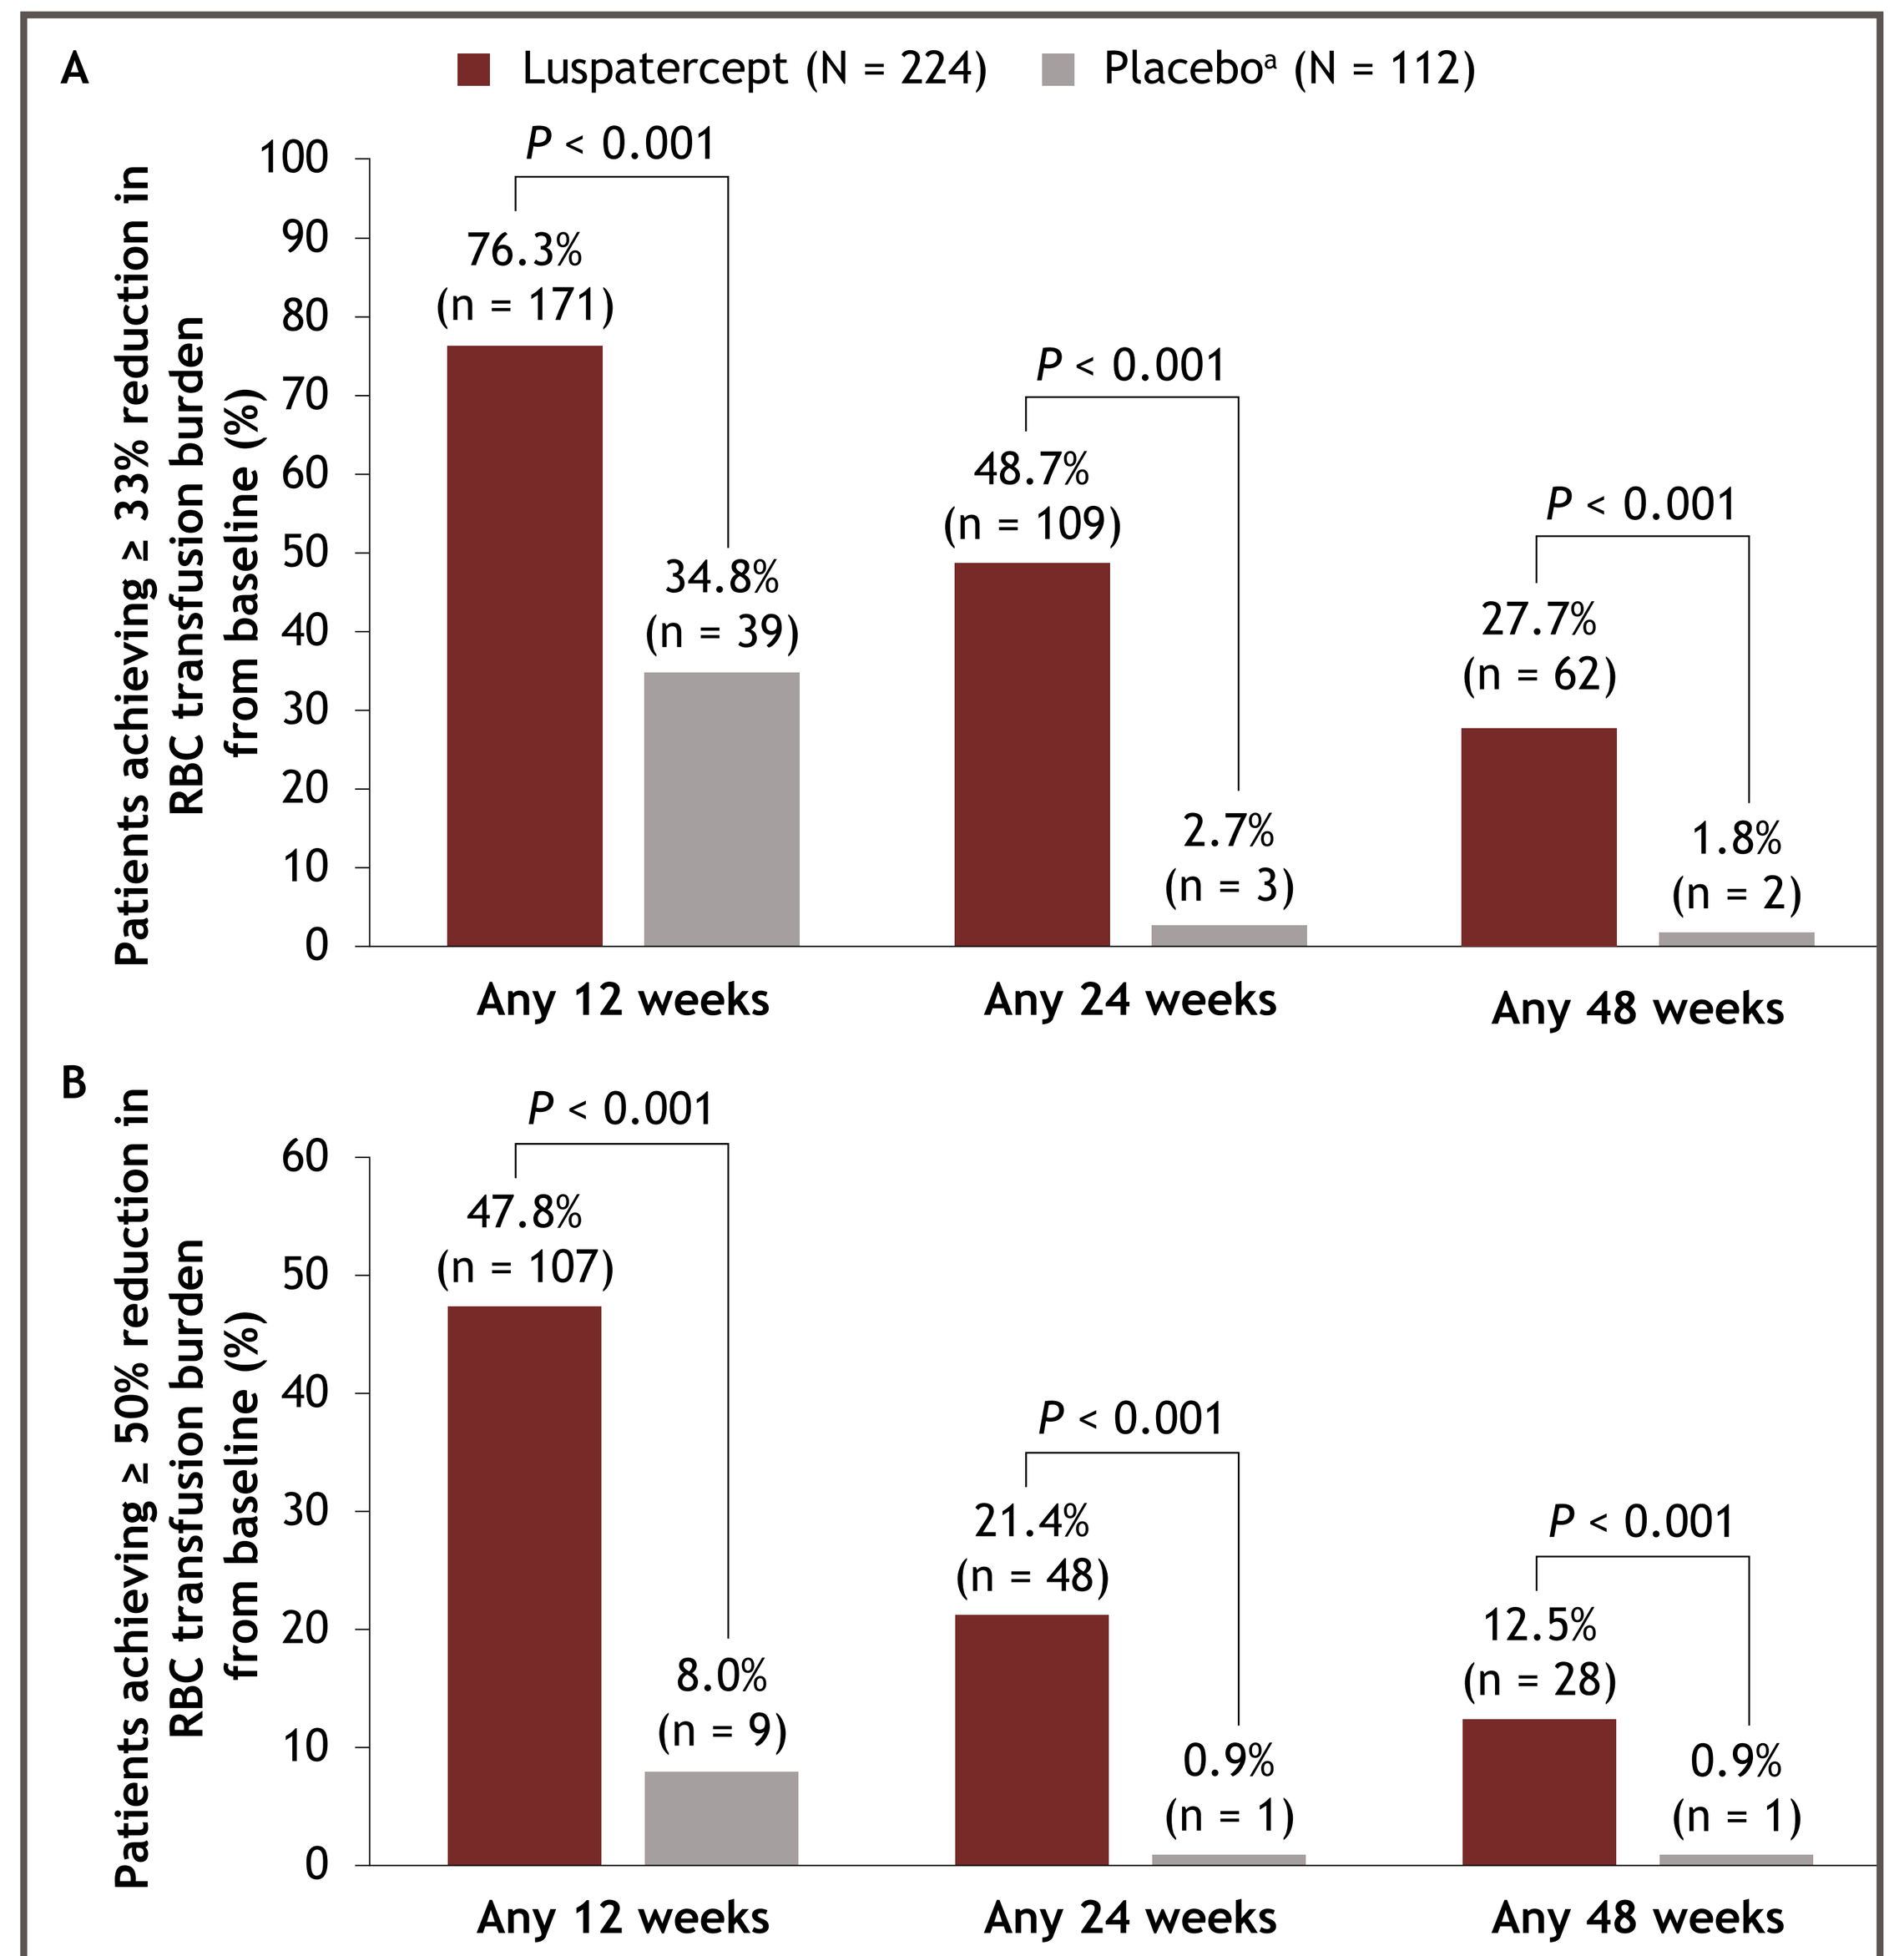
A
Luspatercept (N = 224)
Placebo[a] (N = 112)
Patients achieving ≥ 33% reduction in RBC transfusion burden from baseline (%)
100
90
80
70
60
50
40
30
20
10
0
P < 0.001
76.3% (n = 171)
34.8% (n = 39)
P < 0.001
48.7% (n = 109)
2.7% (n = 3)
P < 0.001
27.7% (n = 62)
1.8% (n = 2)
Any 12 weeks
Any 24 weeks
Any 48 weeks
B
Patients achieving ≥ 50% reduction in RBC transfusion burden from baseline (%)
60
50
40
30
20
10
0
P < 0.001
47.8% (n = 107)
8.0% (n = 9)
P < 0.001
21.4% (n = 48)
0.9% (n = 1)
P < 0.001
12.5% (n = 28)
0.9% (n = 1)
Any 12 weeks
Any 24 weeks
Any 48 weeks

[a]Patients in the placebo arm are evaluated only prior to the open-label phase. RBC, red blood cell.

Table 3. RBC transfusion burden reduction during entire study period

Characteristic	Luspatercept (N = 223)	Placebo[a] (N = 109)
≥ 33% reduction in RBC transfusion burden		
n, (%)	171 (76.3)	39 (38.4)
Duration of longest single period of RBC transfusion burden reduction, days (range)	110 (84–1,004)	90 (84-342)
≥ 50% reduction in RBC transfusion burden		
n, (%)	107 (47.8)	9 (8.0)
Duration of longest single period of RBC transfusion burden reduction, days (range)	99 (84–1,004)	86 (84-342)

[a]Patients in the placebo arm are evaluated only prior to the open-label phase. RBC, red blood cell.

Crossover patients

- 17 (18.5%) and 9 (9.8%) of 92 placebo patients who crossed over to receive luspatercept achieved ≥ 33% or ≥ 50% reduction in RBC transfusion burden, respectively in Weeks 13-24 after crossover (**Figure 3**)

Figure 3. Patients achieving ≥ 33% or ≥ 50% reduction in RBC transfusion burden from baseline during Weeks 13-24

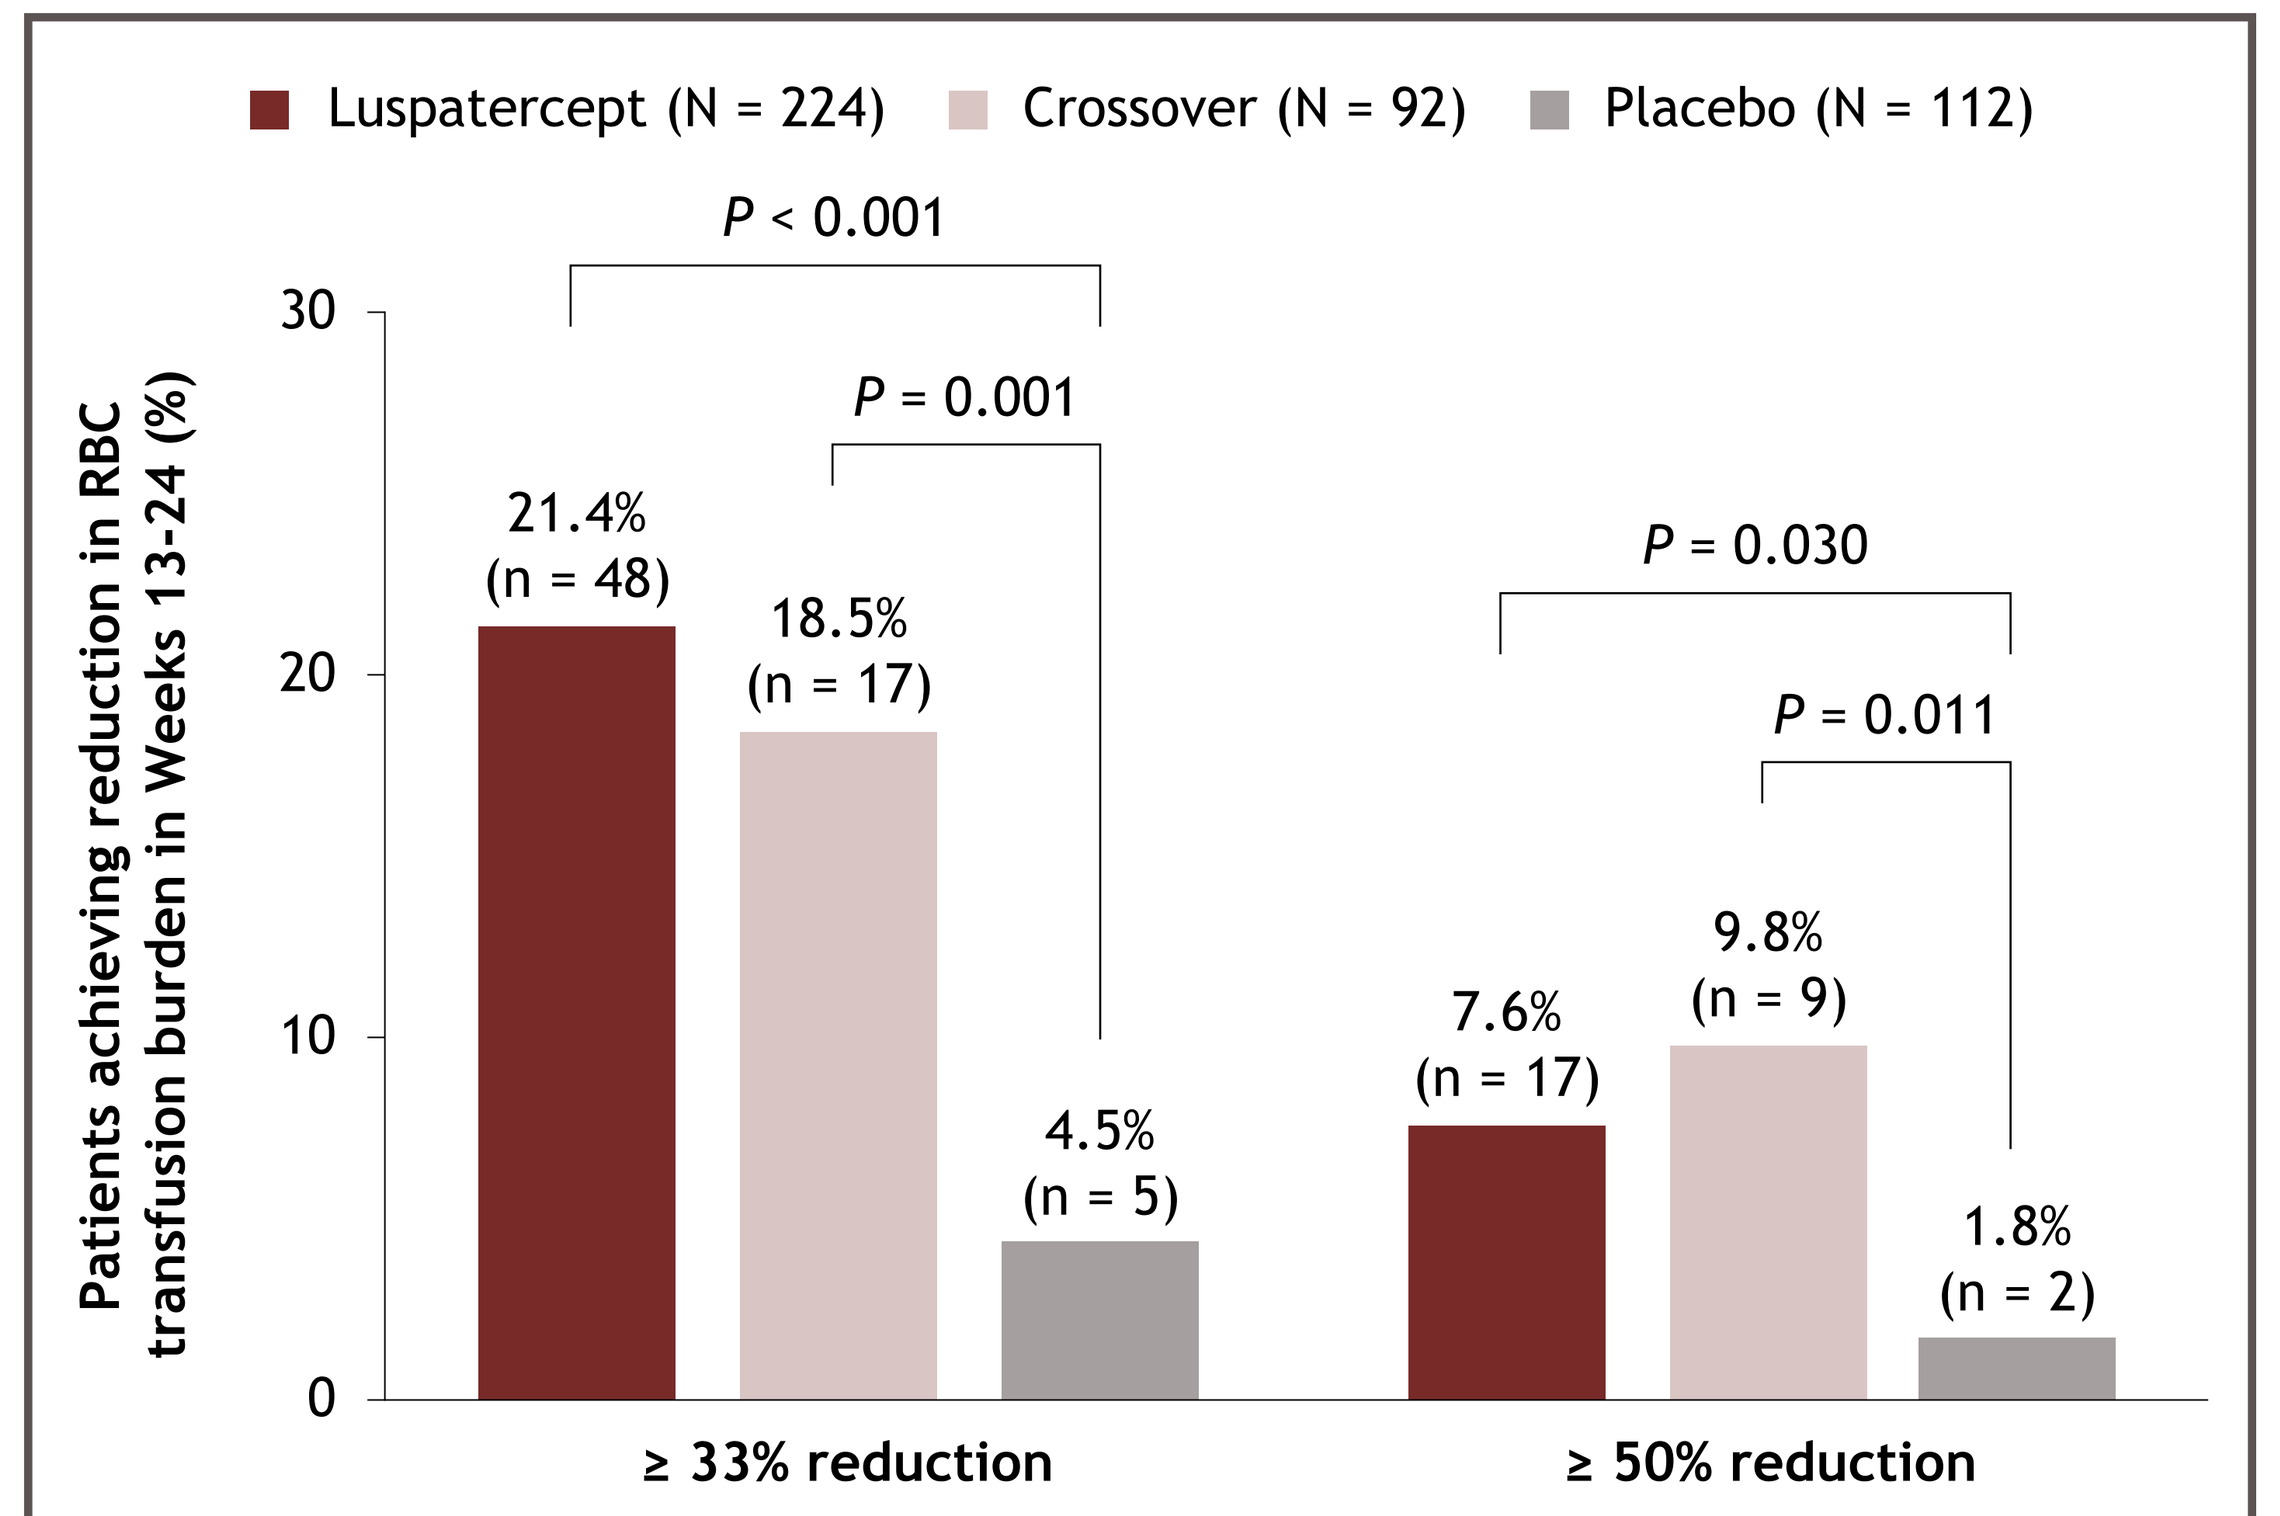
Luspatercept (N = 224)
Crossover (N = 92)
Placebo (N = 112)
Patients achieving reduction in RBC transfusion burden in Weeks 13-24 (%)
30
20
10
0
P < 0.001
P = 0.001
21.4% (n = 48)
18.5% (n = 17)
4.5% (n = 5)
P = 0.030
P = 0.011
7.6% (n = 17)
9.8% (n = 9)
1.8% (n = 2)
≥ 33% reduction
≥ 50% reduction

Luspatercept and placebo analyzed using May 11, 2018 data cutoff. RBC, red blood cell.

- RBC transfusion burden reduction of ≥ 33% during any 12 or any 24 weeks was achieved by 76 (82.6%) and 41 (44.6%) crossover patients, respectively (**Figure 4**)
- RBC transfusion burden reduction of ≥ 50% during any 12 or any 24 weeks was achieved by 36 (39.1%) and 12 (13.0%) crossover patients, respectively (**Figure 4**)

Figure 4. Crossover patients achieving ≥ 33% or ≥ 50% reduction in RBC transfusion burden during any 12 or 24 weeks

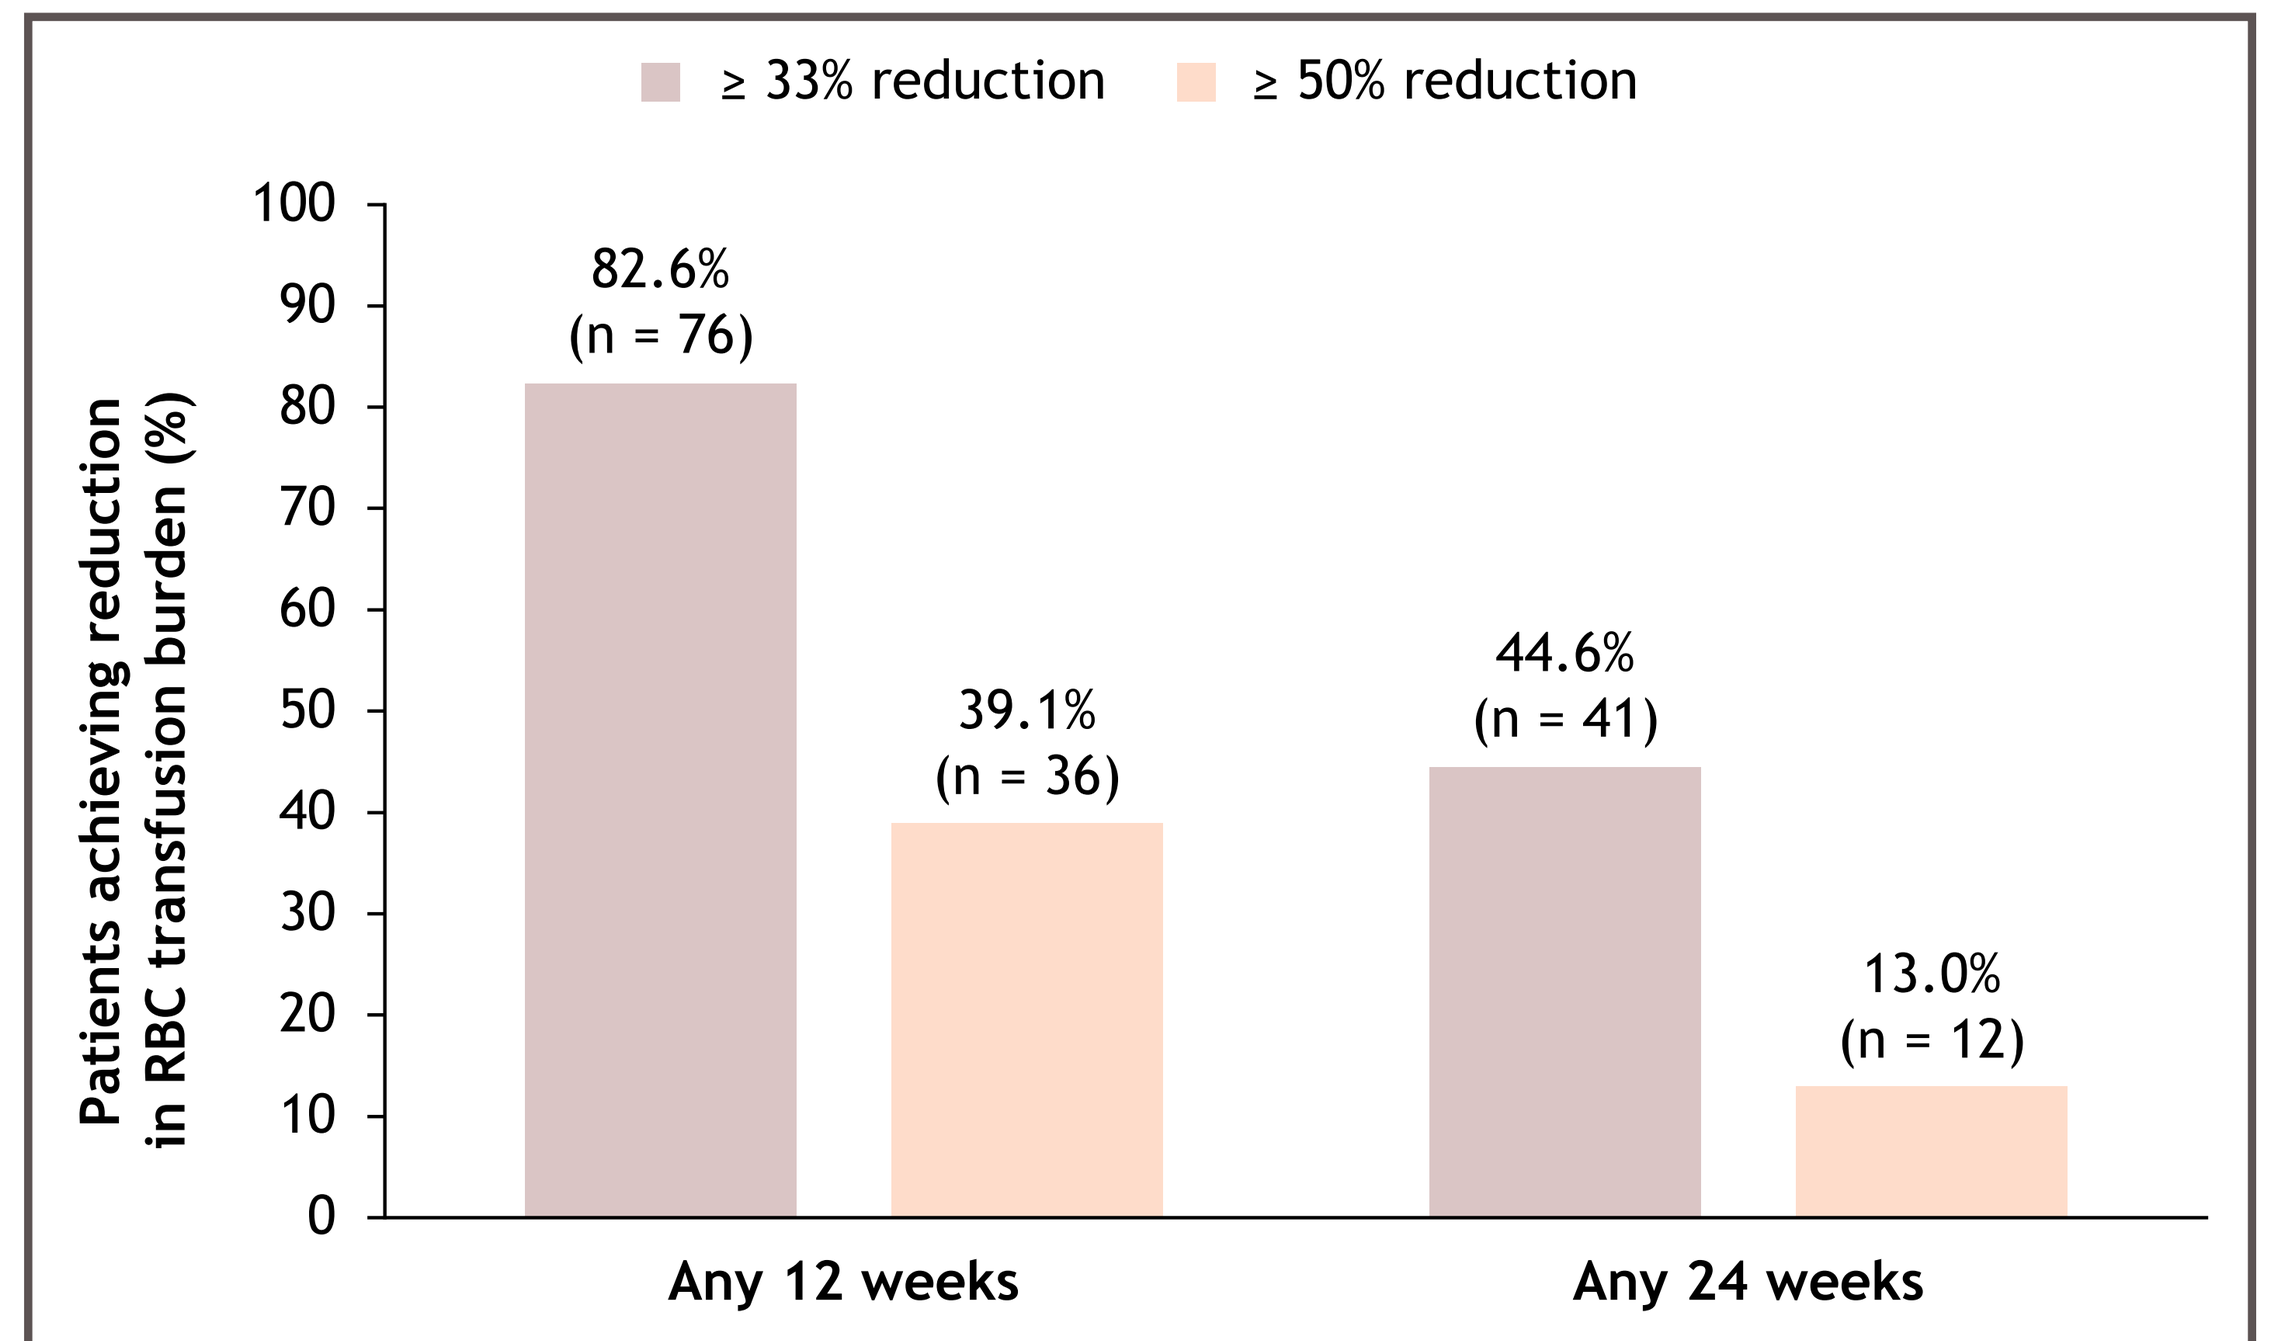
≥ 33% reduction
≥ 50% reduction
Patients achieving reduction in RBC transfusion burden (%)
100
90
80
70
60
50
40
30
20
10
0
82.6% (n = 76)
39.1% (n = 36)
44.6% (n = 41)
13.0% (n = 12)
Any 12 weeks
Any 24 weeks

Crossover population N = 92. RBC, red blood cell.

Safety

- Overall, ≥ 1 TEAE (grade ≥ 3) was reported in 77 of 223 (34.5%), 14 of 92 (15.2%), and 19 of 109 (17.4%) patients in the luspatercept, crossover, and placebo arms, respectively (**Table 4**)
- Of the 128, 53, and 62 patients in the luspatercept, crossover, and placebo arms, respectively, who had a prior splenectomy, 11 (8.6%), 2 (3.8%), and 1 (1.6%) patients reported ≥ 1 thromboembolic event

Table 4. Summary of incidence of TEAEs in safety and crossover patient populations

Parameter, n (%)	Luspatercept (N = 223)	Placebo crossover to luspatercept (N = 92)	Placebo[a] (N = 109)
Grade ≥ 3 TEAEs (≥ 1 event)	77 (34.5)	14 (15.2)	19 (17.4)
Arthralgia	51 (22.9)	15 (16.3)	15 (13.8)
Bone pain	47 (21.1)	16 (17.4)	9 (8.3)
Dizziness	28 (12.6)	4 (4.3)	5 (4.6)
Thromboembolic or thrombophlebitis events (≥ 1 event)	12 (5.4)	2 (2.2)	1 (0.9)
Serious AEs	48 (21.5)	11 (12.0)	8 (7.3)
TEAEs leading to dose reduction	135 (60.5)	36 (39.1)	52 (47.7)

Median treatment duration of luspatercept, crossover, and placebo patients was 119.1, 45.9, and 74.7 weeks, respectively.
[a]Patients in the placebo arm are evaluated only prior to the open-label phase. TEAE, treatment-emergent adverse event.

Conclusions

- Although direct comparisons cannot be made because of different treatment durations, prolonged periods of clinically meaningful reductions in RBC transfusion burden were observed in patients in the luspatercept arm and in patients who crossed over from the placebo arm
- The safety profile of crossover patients did not substantially change for the most frequent TEAEs compared with the primary safety results[6]
 - The incidence of TEAEs leading to dose reductions was similar in crossover and placebo arms, despite differences in median treatment duration
 - The incidence of grade ≥ 3 TEAEs was similar in crossover and placebo patients, except for bone pain; new onset of bone pain has been previously observed to resolve after initial doses[7]
- The increase of thromboembolic events in patients with splenectomies in this study is consistent with previously reported risk
- The BEYOND study is an ongoing phase 2 trial to determine the efficacy and safety of luspatercept in patients with non-transfusion-dependent β-thalassemia (NCT03342404)
- A phase 2a study is evaluating the safety and pharmacokinetics of luspatercept in pediatric patients with transfusion-dependent β-thalassemia (NCT04143724)

References

1. Taher AT, et al. *Lancet* 2018;391:155-167.
2. Cappellini MD, et al. Guidelines for the management of transfusion dependent thalassemia (TDT). 3rd ed. Nicosia: Thalassaemia International Federation; 2014.
3. Attie KM, et al. *Am J Hematol* 2014;89:766-770.
4. Reblozyl® (luspatercept-aamt) [prescribing information]. Summit, NJ: Celgene Corporation; April 2020.
5. Suragani RN, et al. *Nat Med* 2014;20:408-414.
6. Cappellini MD, et al. *N Engl J Med* 2020;382:1219-1231.
7. Viprakasit V, et al. *Blood* 2019;134:abstract 3545.

Acknowledgments

- The study was supported by Celgene, a Bristol-Myers Squibb Company, in collaboration with Acceleron Pharma
- All authors contributed to and approved the presentation; writing and editorial assistance were provided by Jacqueline Moy, PhD, of Excerpta Medica, funded by Bristol-Myers Squibb Company

Disclosures

A.T.T.: Novartis - consultancy, honoraria, research funding; BMS - research funding; La Jolla Pharmaceuticals, Protagonist Therapeutics - consultancy, research funding; Abfero Pharmaceuticals, Inc., Ionis Pharmaceuticals - consultancy. V.V.: BMS, Novartis - consultancy, honoraria, research funding, speakers bureau; Agios Pharmaceuticals, Ionis Pharmaceuticals, La Jolla Pharmaceuticals, Protagonist Therapeutics, Vifor Pharma - consultancy, research funding. M.D.C.: BMS - honoraria; Genzyme/Sanofi - honoraria, membership on board of directors or advisory committee; CRISPR Therapeutics, Novartis, Vifor Pharma - membership on board of directors or advisory committee. O.H.: AB Science - consultancy, equity ownership, honoraria, research funding; BMS, Novartis - research funding. P.G.: Alnylam - consultancy. K.H.M.K.: Agios Pharmaceuticals, Apellis, Bluebird Bio, BMS, Pfizer - consultancy; Alexion, Novartis - consultancy, honoraria; Bioverativ - data safety monitoring board. T.C.: Apo Pharma, Vifor Pharma - consultancy, honoraria, speakers bureau; Agios Pharmaceuticals - consultancy, honoraria; BMS - consultancy, honoraria, steering committee. E.V.: Acceleron Pharma, BMS, Genesis Pharmaceuticals - consultancy, research funding; Protagonist Therapeutics - research funding; Addmedica - membership on board of directors or advisory committee. G.L.F.: Novartis, BMS, Roche - research funding; Bluebird Bio - consultancy. S.P.: Novartis - honoraria, research funding; Acceleron Pharma - research funding. A.Lal: Bluebird Bio, BMS, La Jolla Pharmaceutical, Insight Magnetics, Novartis, Terumo Corporation - research funding; La Jolla Pharmaceutical - consultancy. A.Ka.: Ionis Pharmaceuticals, Novartis Oncology, Vifor Pharma - membership on board of directors or advisory committee, research funding; Celgene Corporation - honoraria, membership on board of directors or advisory committee; Vertex - membership on board of directors or advisory committee. F.S.: Novartis Silence Therapeutics, Roche - membership on a board of directors or advisory committee, Abfero Pharmaceuticals - clinical safety committee, BMS - clinical trial steering committee. J.P.: Agios Pharmaceutials, Bluebird Bio, BMS - consultancy, honoraria; La Jolla Pharmaceuticals, Protagonist Therapeutics, Silence Therapeutics, VifoPharma - honoraria. A.Laa., J.K.S., W-L.K., D.S.: BMS - employment, equity ownership. J.Z.: BMS - employment, stock and other ownership interests. D.M., T.Z.: BMS - employment. C.U.L.: BMS - employment, stock and other ownership interests, patents, royalties, other intellectual property; Cell Medica, Merrimack - patents, royalties, other intellectual property. P.G.L.: Acceleron Pharma - employment, stock and other ownership interests, patents, royalties, other intellectual property; Abbott, AbbVie, FibroGen - stock and other ownership interests. A.P.: BMS, Novartis - consultancy, research funding; Acceleron Pharma - research funding. H.K.L., I.P-K., A.Kh.: no conflicts of interest to disclose.

Presented at the 25th Congress of the European Hematology Association (EHA), June 11–14, 2020.

Contact: http://www.globalbmsmedinfo.com

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from EHA® and the author of this poster.