Effects of luspatercept on serum ferritin in patients with lower-risk myelodysplastic syndromes with ring sideroblasts in the phase 3 MEDALIST trial

Presentation Number
EP807

Pierre Fenaux,[1] Valeria Santini,[2] Ghulam J. Mufti,[3] María Diez-Campelo,[4] Carlo Finelli,[5] Mikkael A. Sekeres,[6] Bruno Quesnel,[7] Odile Beyne-Rauzy,[8] Guillermo Garcia-Manero,[9] Rami S. Komrokji,[10] Rena Buckstein,[11] Maria Teresa Voso,[12] Dominik Selleslag,[13] Amy E. DeZern,[14] Peter L. Greenberg,[15] Amer M. Zeidan,[16] Lionel Adès,[1] Amit Verma,[17] Michael R. Savona,[18] Abderrahmane Laadem,[19] Rodrigo Ito,[19] Jennie Zhang,[19] Anita Rampersad,[19] Daniel Sinsimer,[19] Jessica Morison,[19] Chrystal Louis,[19] Peter G. Linde,[20] Uwe Platzbecker[21]

[1]Service d'Hématologie Séniors, Hôpital Saint-Louis, Université Paris 7, Paris, France; [2]MDS Unit, Azienda Ospedaliero Universitaria (AOU) Careggi, University of Florence, Florence, Italy; [3]Department of Haemato-Oncology, King's College London, London, UK; [4]Hematology Department, Institute of Biomedical Research of Salamanca, University Hospital of Salamanca, Salamanca, Spain; [5]Department of Oncology and Hematology, S. Orsola-Malpighi University Hospital, Bologna, Italy; [6]Department of Hematology and Medical Oncology, Cleveland Clinic, Cleveland, OH, USA; [7]Service des Maladies du Sang, Hôpital Huriez, Centre Hospitalier Universitaire de Lille, Lille, France; [8]Médicine Interne, Institute Universitaire du Cancer de Toulouse Oncopole, Toulouse, France; [9]Department of Leukemia, University of Texas MD Anderson Cancer Center, Houston, TX, USA; [10]Moffitt Cancer Center, Tampa, FL, USA; [11]Odette Cancer Centre, Sunnybrook Health Sciences Centre, Toronto, ON, Canada; [12]Dipartimento di Biopatologia e Diagnostica per Immagini, University of Rome Tor Vergata, Rome, Italy; [13]Department of Haematology, AZ Sint-Jan, Bruges, Belgium; [14]The Sidney Kimmel Comprehensive Cancer Center at Johns Hopkins, Baltimore, MD, USA; [15]Stanford University Cancer Center, Stanford, CA, USA; [16]Department of Internal Medicine, Yale School of Medicine and Yale Cancer Center, Yale University, New Haven, CT, USA; [17]Department of Oncology, Albert Einstein College of Medicine, Montefiore Medical Center, New York, NY, USA; [18]Vanderbilt-Ingram Cancer Center, Vanderbilt University School of Medicine, Nashville, TN, USA; [19]Bristol Myers Squibb, Princeton, NJ, USA; [20]Acceleron Pharma, Cambridge, MA, USA; [21]Medical Clinic and Policlinic 1, Hematology and Cellular Therapy, University Hospital Leipzig, Leipzig, Germany

Scientific Content on Demand
To request a copy of this poster:
Scan QR code
via a barcode reader application
This QR code will take you to an external site.
QR codes are valid for 120 days after the congress presentation date.

Introduction

- Iron overload resulting from red blood cell (RBC) transfusions is associated with significant morbidity in patients with myelodysplastic syndromes (MDS)[1,2]
- For patients who have MDS with ring sideroblasts (RS), iron overload may also occur prior to receiving RBC transfusions, due to ineffective erythropoiesis[1]
- Luspatercept is a first-in-class erythroid maturation agent, which binds to select transforming growth factor beta superfamily ligands to diminish Smad2/3 signaling and promote late-stage erythropoiesis[3]
- The safety and efficacy of luspatercept has been assessed in the phase 3, randomized, double-blind, placebo-controlled MEDALIST study of patients with lower-risk MDS with RS who require RBC transfusions
 - The primary results showed a significant proportion of luspatercept-treated patients achieved RBC transfusion independence ≥ 8 weeks during Weeks 1–24[4]
- Luspatercept has been approved by the US Food and Drug Administration for the treatment of anemia in adult patients failing an erythropoiesis-stimulating agent and requiring ≥ 2 RBC units over 8 weeks with Revised International Prognostic Scoring System (IPSS-R) Very low- to Intermediate-risk MDS with RS or with myelodysplastic/myeloproliferative neoplasm with RS and thrombocytosis[5]

Objective

- To assess changes from baseline in serum ferritin (SF) and iron chelation therapy (ICT) use in patients in the MEDALIST trial

Methods

Study design

- MEDALIST is an ongoing phase 3, randomized, double-blind, placebo-controlled trial (NCT02631070); patients in the luspatercept arm who have not discontinued from treatment continue to receive the study drug
- Patients were randomized at 65 sites between March 2016 and June 2017
- The MEDALIST study design is shown in **Figure 1**
 - Patients were randomized 2:1 to receive luspatercept (starting dose 1.0 mg/kg with titration up to 1.75 mg/kg, if needed) or placebo, administered subcutaneously every 3 weeks for ≥ 24 weeks
 - Stratification factors were: average baseline RBC transfusion burden (≥ 6 vs < 6 units per 8 weeks) and baseline IPSS-R score

Figure 1. MEDALIST trial study design

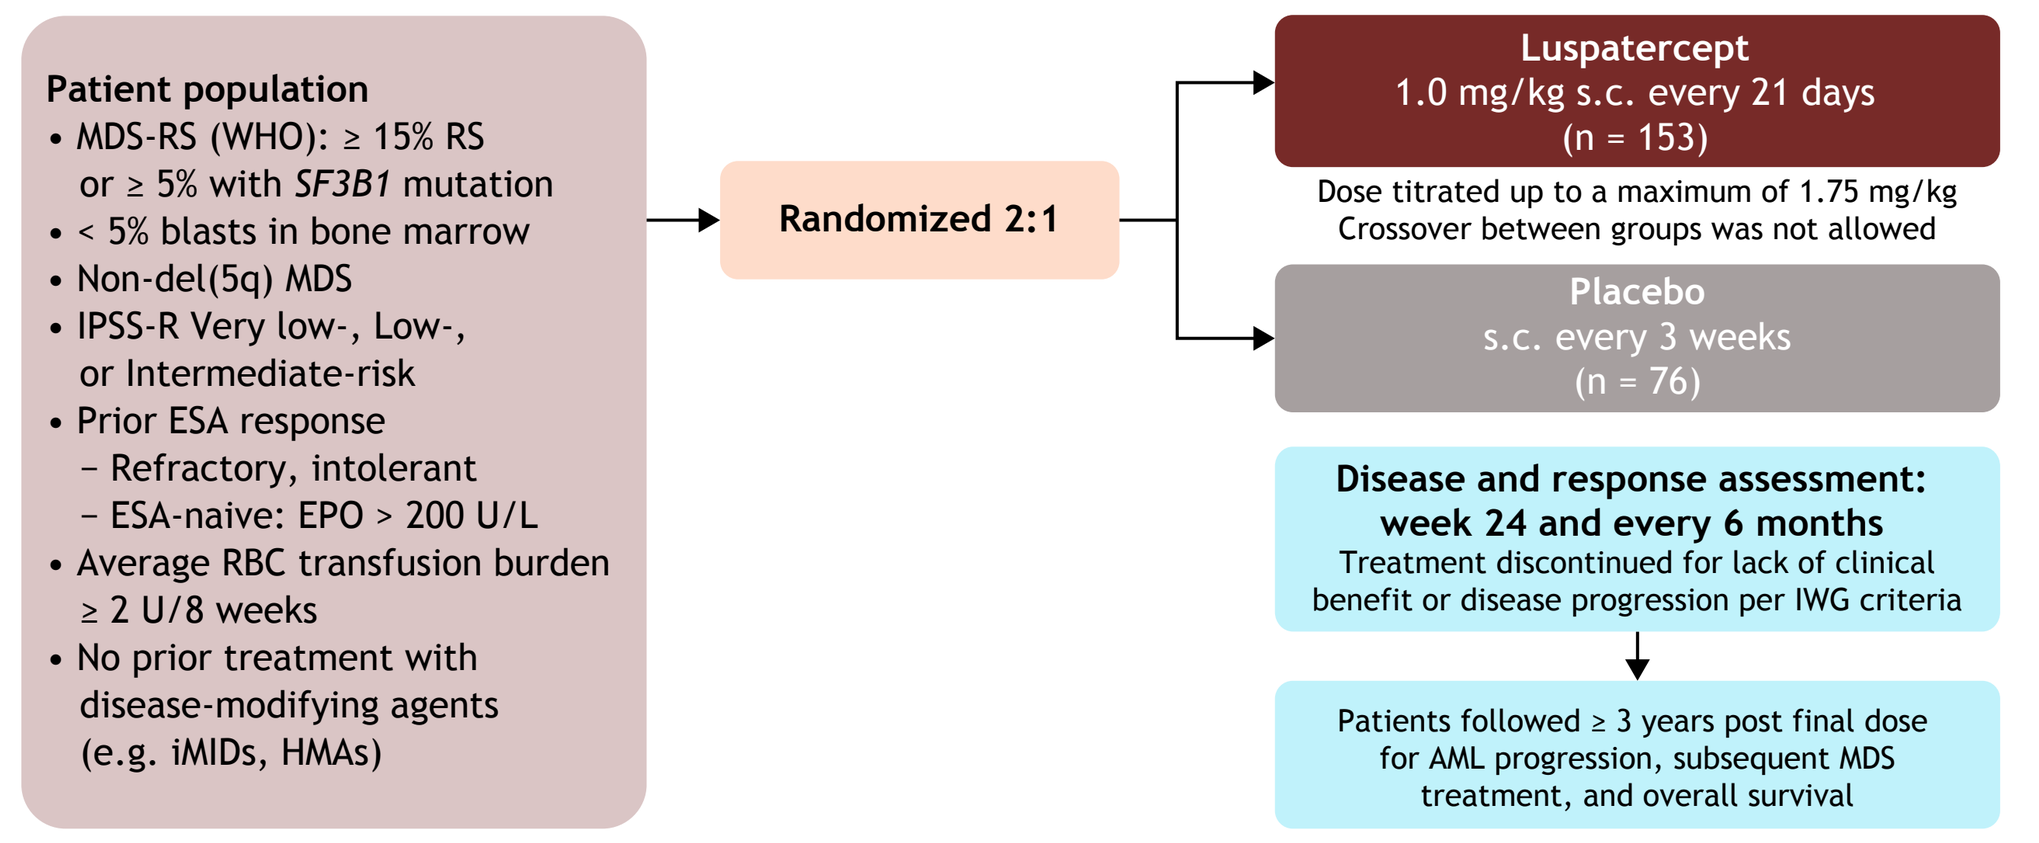
Patient population
• MDS-RS (WHO): ≥ 15% RS or ≥ 5% with SF3B1 mutation
• < 5% blasts in bone marrow
• Non-del(5q) MDS
• IPSS-R Very low-, Low-, or Intermediate-risk
• Prior ESA response
– Refractory, intolerant
– ESA-naive: EPO > 200 U/L
• Average RBC transfusion burden ≥ 2 U/8 weeks
• No prior treatment with disease-modifying agents (e.g. iMIDs, HMAs)
Randomized 2:1
Luspatercept
1.0 mg/kg s.c. every 21 days
(n = 153)
Dose titrated up to a maximum of 1.75 mg/kg
Crossover between groups was not allowed
Placebo
s.c. every 3 weeks
(n = 76)
Disease and response assessment: week 24 and every 6 months
Treatment discontinued for lack of clinical benefit or disease progression per IWG criteria
Patients followed ≥ 3 years post final dose for AML progression, subsequent MDS treatment, and overall survival

AML, acute myeloid leukemia; EPO, erythropoietin; ESA, erythropoiesis-stimulating agent; HMA, hypomethylating agent; iMID, immunomodulatory drug; IPSS-R, Revised International Prognostic Scoring System; IWG, International Working Group; MDS, myelodysplastic syndromes; RBC, red blood cell; RS, ring sideroblasts; s.c., subcutaneously; *SF3B1*, splicing factor 3b subunit 1; WHO, World Health Organization.

Change in mean SF and mean ICT daily dose endpoints and safety analyses

- Changes in mean SF levels and mean daily dose of ICT were analyzed over Weeks 9–24 and Weeks 33–48 and reported as least squares mean (LSM) values; LSMs are means estimated from an analysis of covariance model and adjusted for other factors in the model
- Changes in mean SF levels from baseline over Weeks 9–24 and Weeks 33–48 were analyzed using a post hoc multivariate analysis with baseline SF level, mean ICT daily dose in the 16 weeks prior to first dose, achievement of modified hematologic improvement-erythroid[6] (HI-E) in Weeks 1–24, average baseline RBC transfusion requirement, and baseline IPSS-R score as covariates
- These analyses were measured within the first 48 weeks of the study and are reported as of May 8, 2018
- Baseline RBC transfusion burden subgroups for these analyses included:
 - ≥ 6 RBC units/8 weeks (high transfusion burden; HTB)
 - < 6 RBC units/8 weeks (low transfusion burden; LTB)
- Safety analyses are reported as of July 1, 2019

Results

Patients

- 229 patients were randomized and received luspatercept (n = 153) or placebo (n = 76) (**Table 1**)
- Baseline transfusion burden was ≥ 6 RBC units/8 weeks (HTB) in 66 of 153 (43.1%) luspatercept patients and in 33 of 76 (43.4%) placebo patients (**Table 1**)
- Median baseline SF was 1,306.1 µg/L in HTB luspatercept patients and 1,492.9 µg/L in HTB placebo patients (**Table 1**)
- 50 (75.8%) luspatercept patients received ICT at baseline versus 26 (78.8%) placebo patients (**Table 1**)

Table 1. Baseline patient characteristics

Characteristic	Luspatercept (n = 153)	Placebo (n = 76)
Age, median (range), years	71 (40-95)	72 (26-91)
IPSS-R, n (%)[a]		
Very low	18 (11.8)	6 (7.9)
Low	109 (71.2)	57 (75.0)
Intermediate	25 (16.3)	13 (17.1)
RBC transfusion burden, median (range), RBC U/8 weeks[b]	5 (1-15)	5 (2-20)
≥ 6 RBC U/8 weeks (HTB), n (%)	66 (43.1)	33 (43.4)
< 6 RBC U/8 weeks (LTB), n (%)	87 (56.9)	43 (56.6)
SF, µg/L		
Mean (SD)	1,348.0 (971.2)	1,503.8 (1,242.9)
HTB patients[c]	1,620.3 (1,099.9)	1,808.9 (1,288.6)
LTB patients[d]	1,141.5 (808.0)	1,269.7 (1,168.1)
Median (range)	1,089.2 (64-5,968)	1,122.1 (165-5,849)
HTB patients[c]	1,306.1 (64-5,968)	1,492.9 (256-5,280)
LTB patients[d]	959.2 (184-4,865)	1,013.0 (165-5,849)
ICT use, n (%)	71 (46.4)	40 (52.6)
HTB patients[c]	50/66 (75.8)	26/33 (78.8)
LTB patients[d]	21/87 (24.1)	14/43 (32.6)
Baseline Hb, median (range), g/dL[e]	7.6 (6-10)	7.6 (5-9)

[a]1 patient in the luspatercept arm was classified as IPSS-R High risk. [b]In the 16 weeks prior to randomization. [c]HTB defined as patients with baseline transfusion burden of ≥ 6 RBC units/8 weeks. [d]LTB defined as patients with baseline transfusion burden of < 6 RBC units/8 weeks. [e]Data were missing for 1 patient in the luspatercept arm.
Hb, hemoglobin; HTB, high transfusion burden; ICT, iron chelation therapy; IPSS-R, Revised International Prognostic Scoring System; LTB, low transfusion burden; RBC, red blood cell; SD, standard deviation; SF, serum ferritin.

Changes in SF over Weeks 9-24 and 33-48

- A decrease from baseline in mean SF was observed with luspatercept versus placebo over Weeks 9–24 (LSM −2.7 vs +226.5 µg/L; *P* = 0.0024) and Weeks 33–48 (LSM −72.0 vs +247.4 µg/L; *P* = 0.0294) (**Figure 2**)
 - In HTB patients, LS mean SF change from baseline in luspatercept versus placebo arms over Weeks 9–24 was −63.5 µg/L versus +271.2 µg/L (*P* = 0.0103) and over Weeks 33–48 was −7.0 µg/L and −19.0 µg/L, although the number of patients in the placebo arm was small (n = 6)
- In a multivariate analysis, mean SF levels were decreased from baseline in luspatercept patients versus placebo over Weeks 9–24 (LSM −26.1 vs +255.2 µg/L) and Weeks 33–48 (LSM −132.0 vs +705.8 µg/L) (**Figure 3**)
 - In luspatercept HTB patients, the LSM change from baseline in mean SF was +0.6 µg/L over Weeks 9–24 and −178.0 µg/L over Weeks 33–48

Figure 2. LSM changes from baseline in SF over Weeks 9-24 (A) and Weeks 33-48 (B)

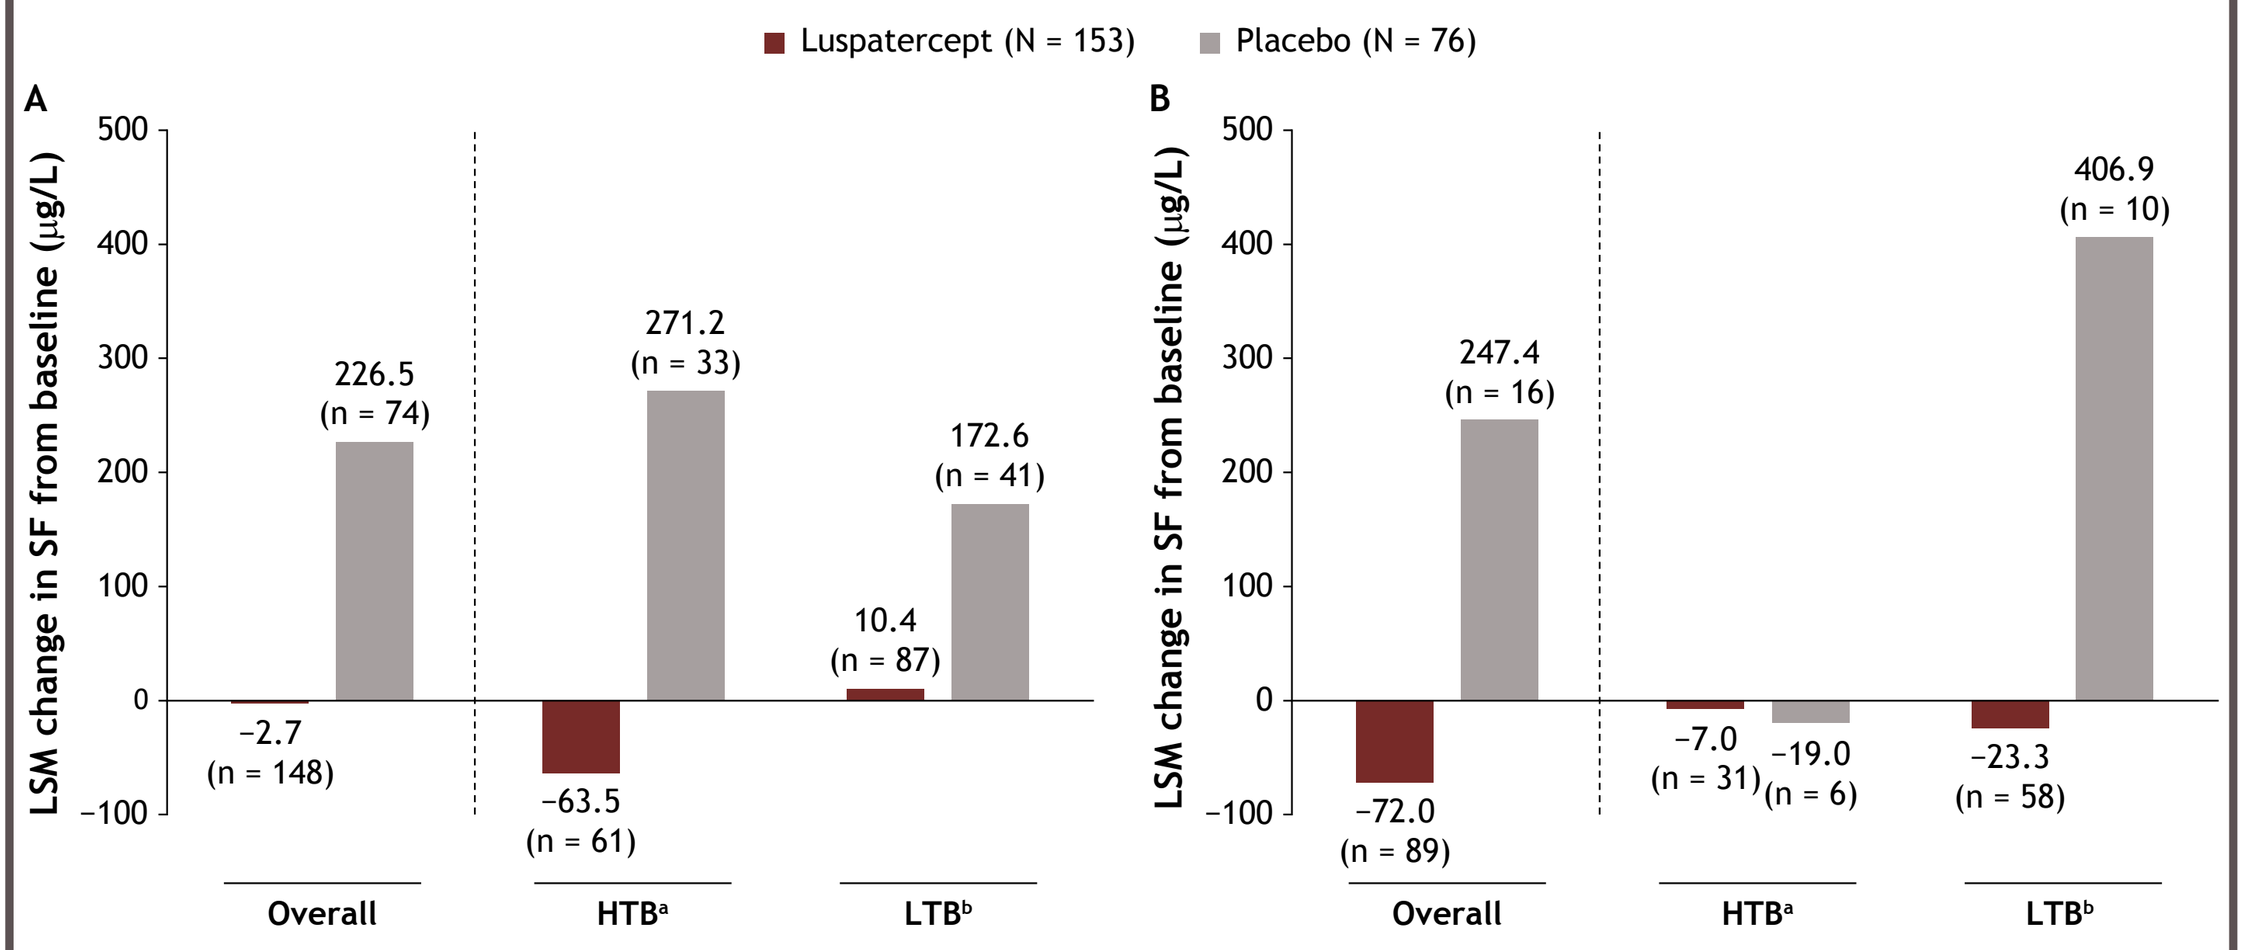
Luspatercept (N = 153)
Placebo (N = 76)
A
B
LSM change in SF from baseline (µg/L)
−2.7 (n = 148)
226.5 (n = 74)
−63.5 (n = 61)
271.2 (n = 33)
10.4 (n = 87)
172.6 (n = 41)
−72.0 (n = 89)
247.4 (n = 16)
−7.0 (n = 31)
−19.0 (n = 6)
−23.3 (n = 58)
406.9 (n = 10)
Overall
HTB[a]
LTB[b]

ANCOVA was used to compare the treatment difference between groups, with the change in value as the dependent variable, treatment group (2 levels) as a factor, and baseline SF value as covariates.
[a]HTB defined as patients with baseline transfusion burden of ≥ 6 RBC units/8 weeks. [b]LTB defined as patients with baseline transfusion burden of < 6 RBC units/8 weeks.
ANCOVA, analysis of covariance; HTB, high transfusion burden; LSM, least squares mean; LTB, low transfusion burden; RBC, red blood cell; SF, serum ferritin.

Figure 3. Multivariate analysis of LSM changes from baseline in SF levels over Weeks 9-24 (A) and Weeks 33-48 (B)

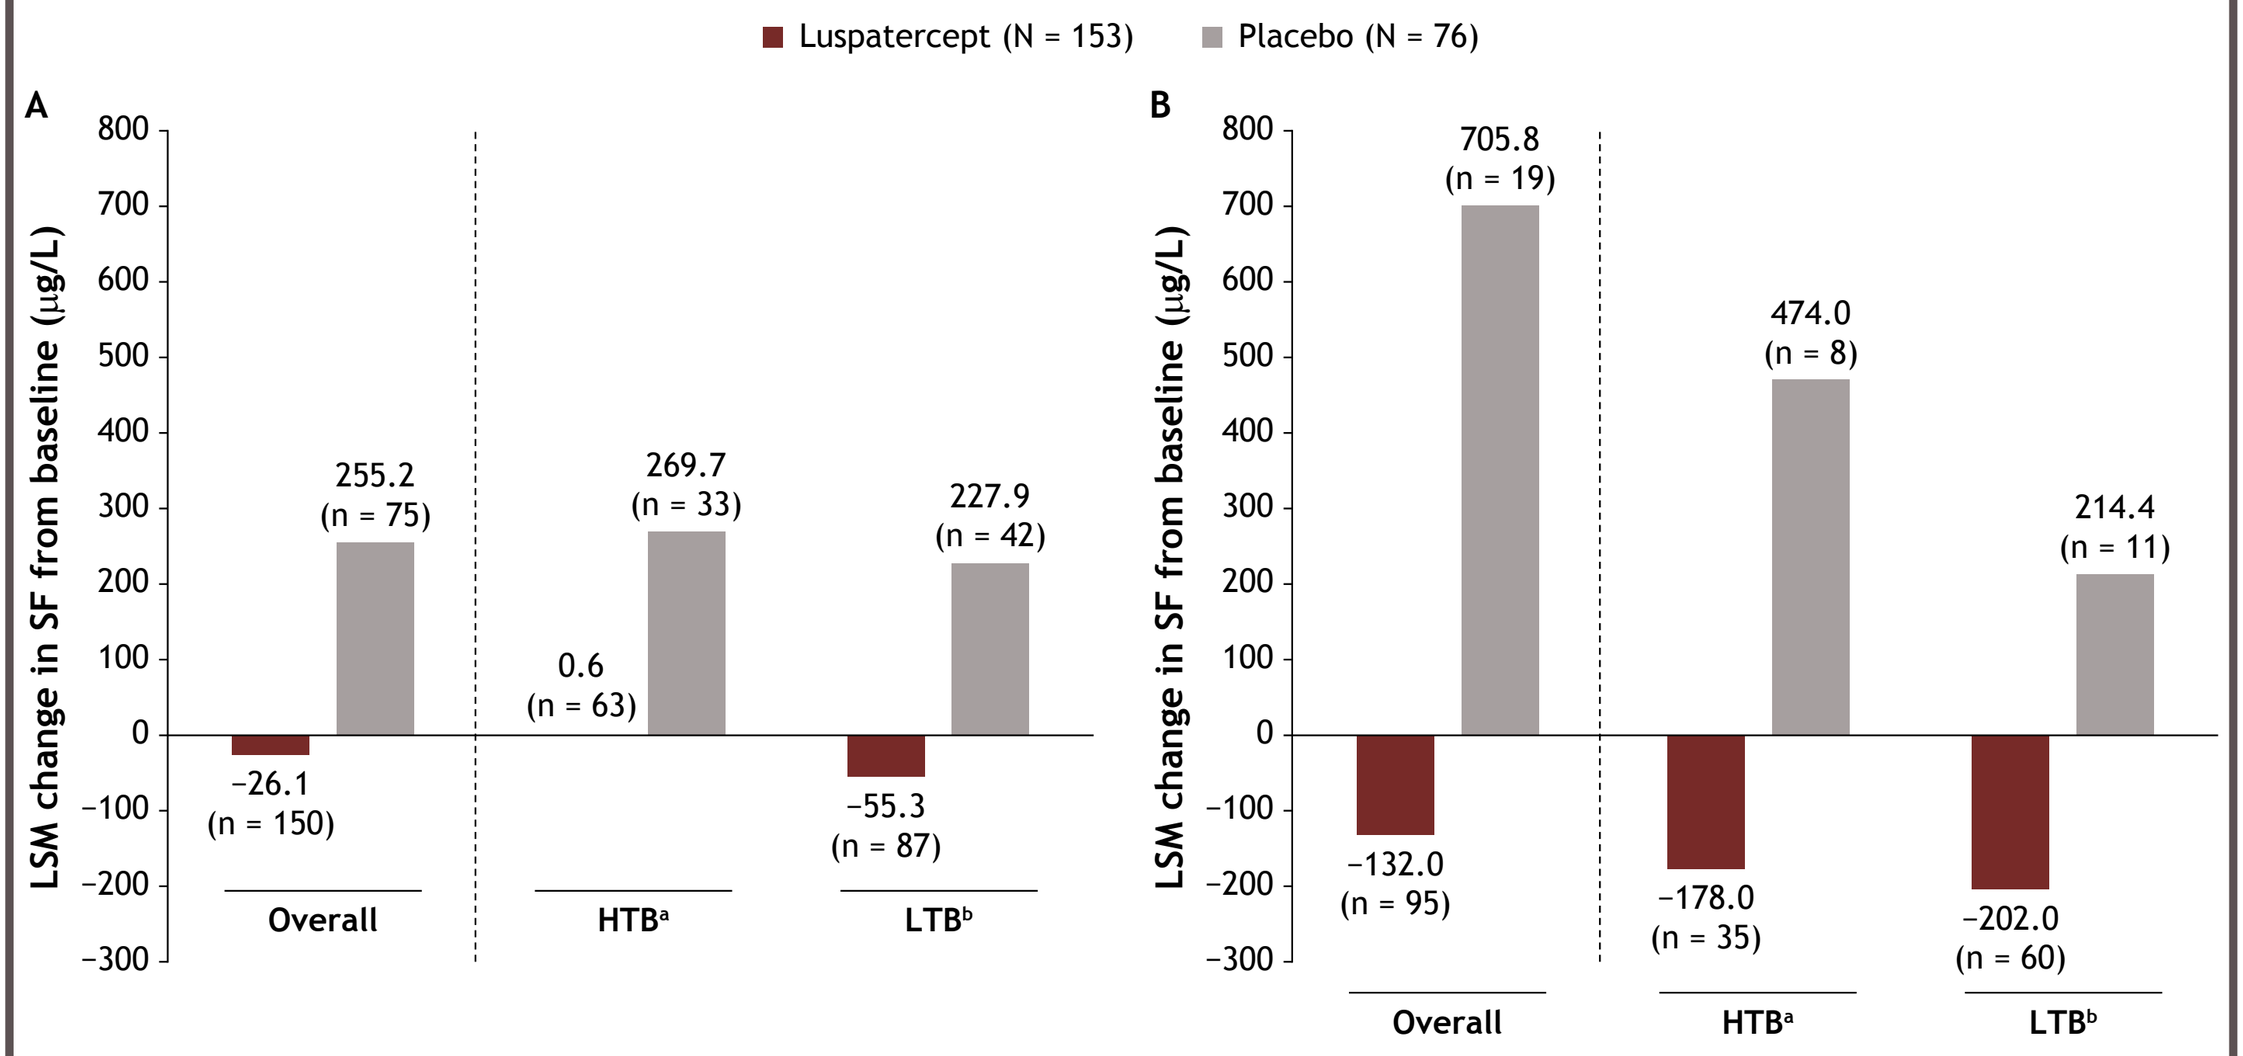
Luspatercept (N = 153)
Placebo (N = 76)
A
B
LSM change in SF from baseline (µg/L)
−26.1 (n = 150)
255.2 (n = 75)
0.6 (n = 63)
269.7 (n = 33)
−55.3 (n = 87)
227.9 (n = 42)
−132.0 (n = 95)
705.8 (n = 19)
−178.0 (n = 35)
474.0 (n = 8)
−202.0 (n = 60)
214.4 (n = 11)
Overall
HTB[a]
LTB[b]

Estimates are based on an ANCOVA model with treatment (luspatercept vs placebo) as the main factor, and baseline SF level, mean daily dose of ICT during the 16 weeks immediately prior to the first dose, HI-E in Weeks 1-24, average baseline RBC transfusion requirement (≥ 6 vs < 6 RBC units/8 weeks), and baseline IPSS-R score (Very low or Low, vs Intermediate) as covariates. [a]HTB defined as patients with baseline transfusion burden of ≥ 6 RBC units/8 weeks. [b]LTB defined as patients with baseline transfusion burden of < 6 RBC units/8 weeks.
ANCOVA, analysis of covariance; HI-E, hematologic improvement-erythroid; HTB, high transfusion burden; ICT, iron chelation therapy; IPSS-R, Revised International Prognostic Scoring System; LSM, least squares mean; LTB, low transfusion burden; RBC, red blood cell; SF, serum ferritin.

Changes in SF over Weeks 9-24 by achievement of HI-E (Weeks 1-24)

- The LSM change from baseline in SF averaged over Weeks 9–24 in HI-E responders (Weeks 1–24) was −102.3 µg/L versus 60.4 µg/L in luspatercept versus placebo arms (*P* = 0.3442) (**Figure 4**)
 - In HTB HI-E responders (Weeks 1-24), LSM change from baseline in SF was −111.4 µg/L and +8.6 µg/L in luspatercept and placebo arms
- A difference in mean SF change from baseline was observed in luspatercept HI-E non-responders (Weeks 1–24) versus placebo HI-E non-responders (LSM +73.4 vs +244.2 µg/L; *P* = 0.0777) (**Figure 4**)
 - A difference in mean SF change from baseline was observed in luspatercept HTB HI-E non-responders (Weeks 1–24) versus placebo HTB HI-E non-responders (LSM +3.0 vs +344.3 µg/L; *P* = 0.0452)

Changes in ICT use

- During the 12 weeks immediately prior to the first dose, the mean daily dose of ICT in luspatercept patients was 1,408.9 mg versus 973.7 mg during the 12 weeks prior to study discontinuation (**Table 2**)
- LSM change from baseline in mean daily ICT dose in luspatercept patients was 10.0 mg/d over Weeks 9–24 and −148.8 mg/day over Weeks 33–48 (**Figure 5**)
 - In luspatercept HTB patients, LSM change from baseline in mean daily ICT dose was −68.5 mg/day over Weeks 9–24 and −327.1 mg/day over Weeks 33–48

Figure 4. LSM changes from baseline in SF over Weeks 9–24 by HI-E response[a] (Weeks 1-24)

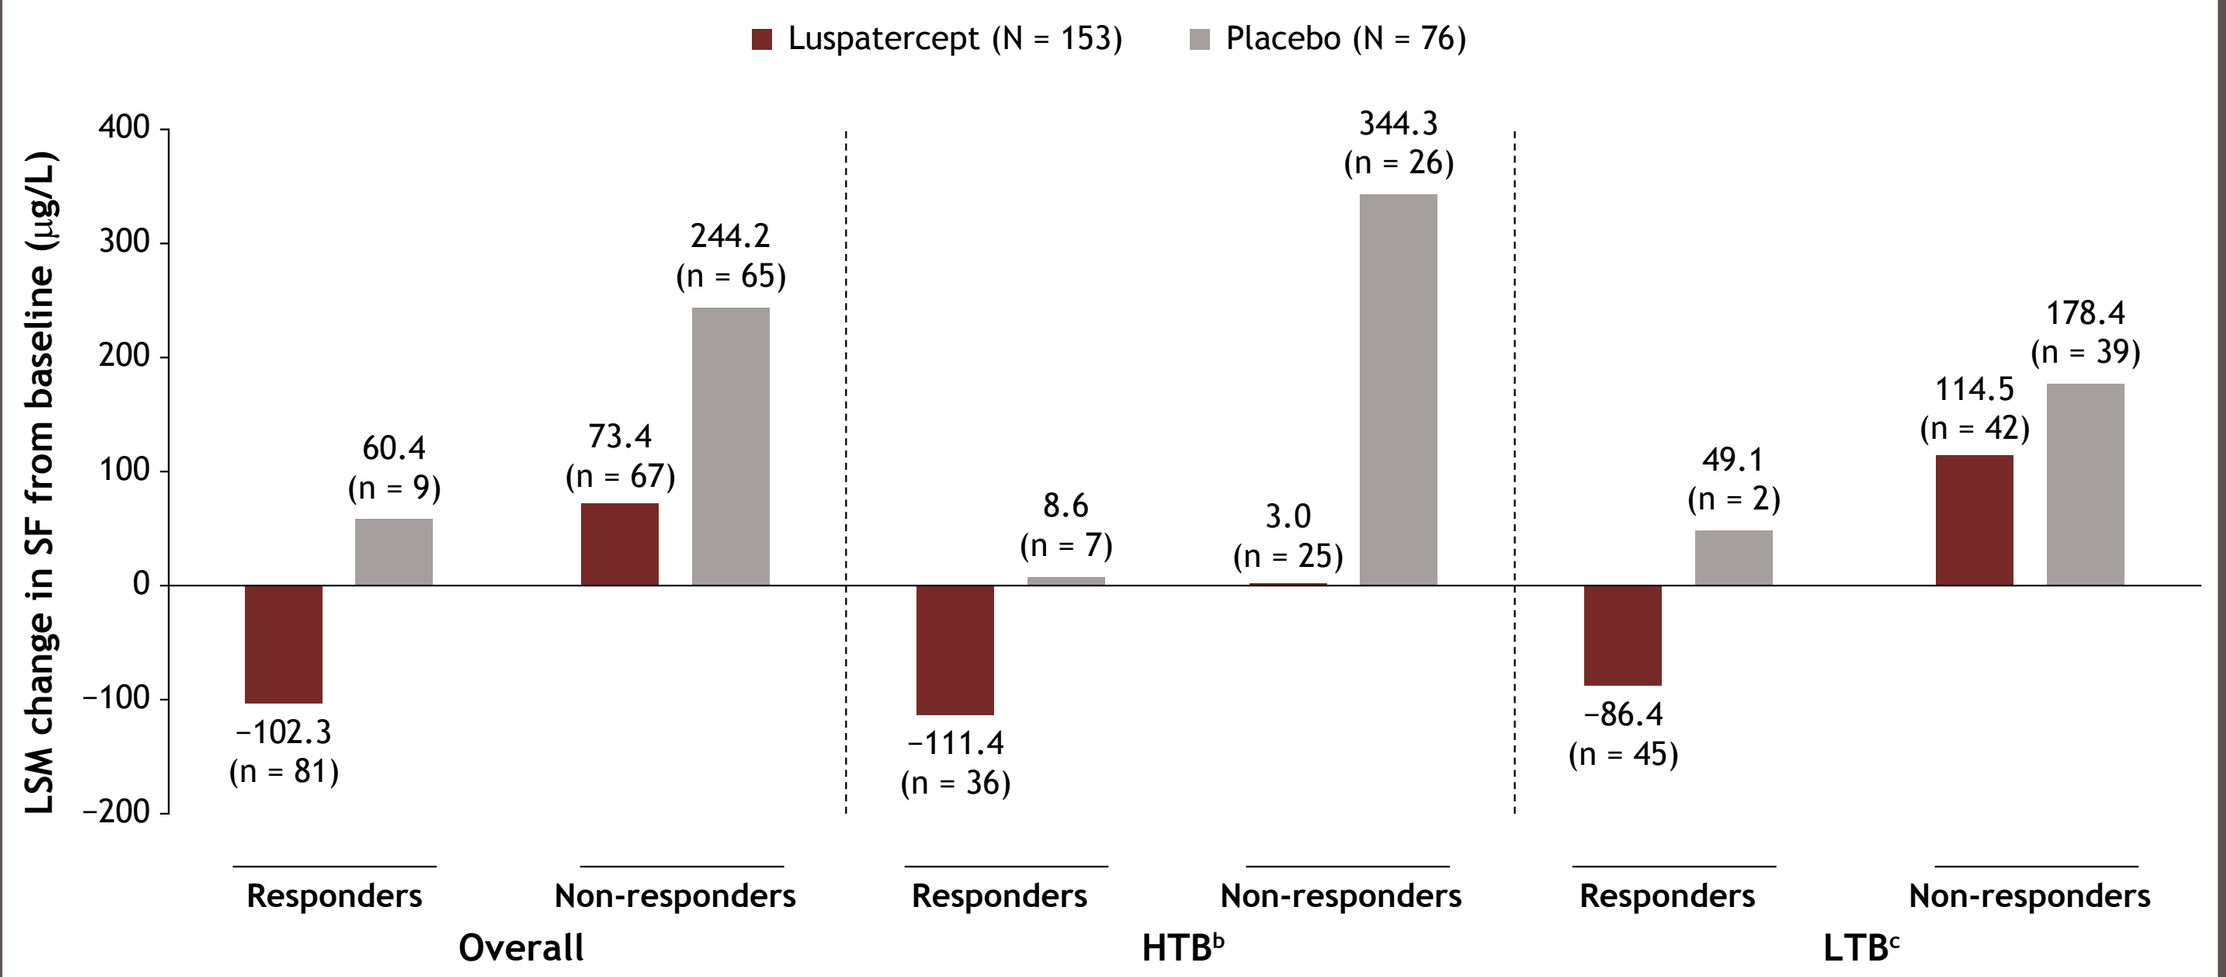
Luspatercept (N = 153)
Placebo (N = 76)
LSM change in SF from baseline (µg/L)
−102.3 (n = 81)
60.4 (n = 9)
73.4 (n = 67)
244.2 (n = 65)
−111.4 (n = 36)
8.6 (n = 7)
3.0 (n = 25)
344.3 (n = 26)
−86.4 (n = 45)
49.1 (n = 2)
114.5 (n = 42)
178.4 (n = 39)
Responders
Non-responders
Overall
HTB[b]
LTB[c]

ANCOVA was used to compare the treatment difference between groups, with the change in value as the dependent variable, treatment group (2 levels) as a factor, and baseline SF value as covariates.
[a]Defined as the proportion of patients who met the modified HI-E criteria per the IWG 2006 criteria[6] sustained over any consecutive 56-day (8-week) period during Weeks 1-24: for subjects with baseline transfusion burden ≥ 4 RBC units/8 weeks, a reduction ≥ 4 RBC units/8 weeks; for subjects with baseline transfusion burden < 4 RBC units/8 weeks, mean Hb increase ≥ 1.5 g/dL in the absence of transfusions for ≥ 8 weeks. [b]HTB defined as patients with baseline transfusion burden of ≥ 6 RBC units/8 weeks. [c]LTB defined as patients with baseline transfusion burden of < 6 RBC units/8 weeks.
ANCOVA, analysis of covariance; Hb, hemoglobin; HI-E, hematologic improvement-erythroid; HTB, high transfusion burden; IWG, International Working Group; LSM, least squares mean; LTB, low transfusion burden; RBC, red blood cell; SF, serum ferritin.

Table 2. Summary of mean daily dose of ICT

Parameter	Luspatercept (n = 153)	Placebo (n = 76)
Mean daily dose of ICT,[a] mg		
During the 12 weeks immediately prior to first dose, n	66	34
Mean (SD), mg	1,408.9 (1,244.41)	1,067.3 (591.47)
During the 12 weeks prior to study discontinuation, n	40	5
Mean (SD), mg	973.7 (795.41)	712.5 (396.43)
Patients who began ICT[a] after first dose and before date of study discontinuation, n (%)	11 (7.2)	5 (6.6)

[a]ICT included deferasirox, deferiprone, or deferoxamine mesylate/deferoxamine.
ICT, iron chelation therapy; SD, standard deviation.

Figure 5. LSM changes from baseline in ICT use during Weeks 9-24 (A) and Weeks 33-48 (B)

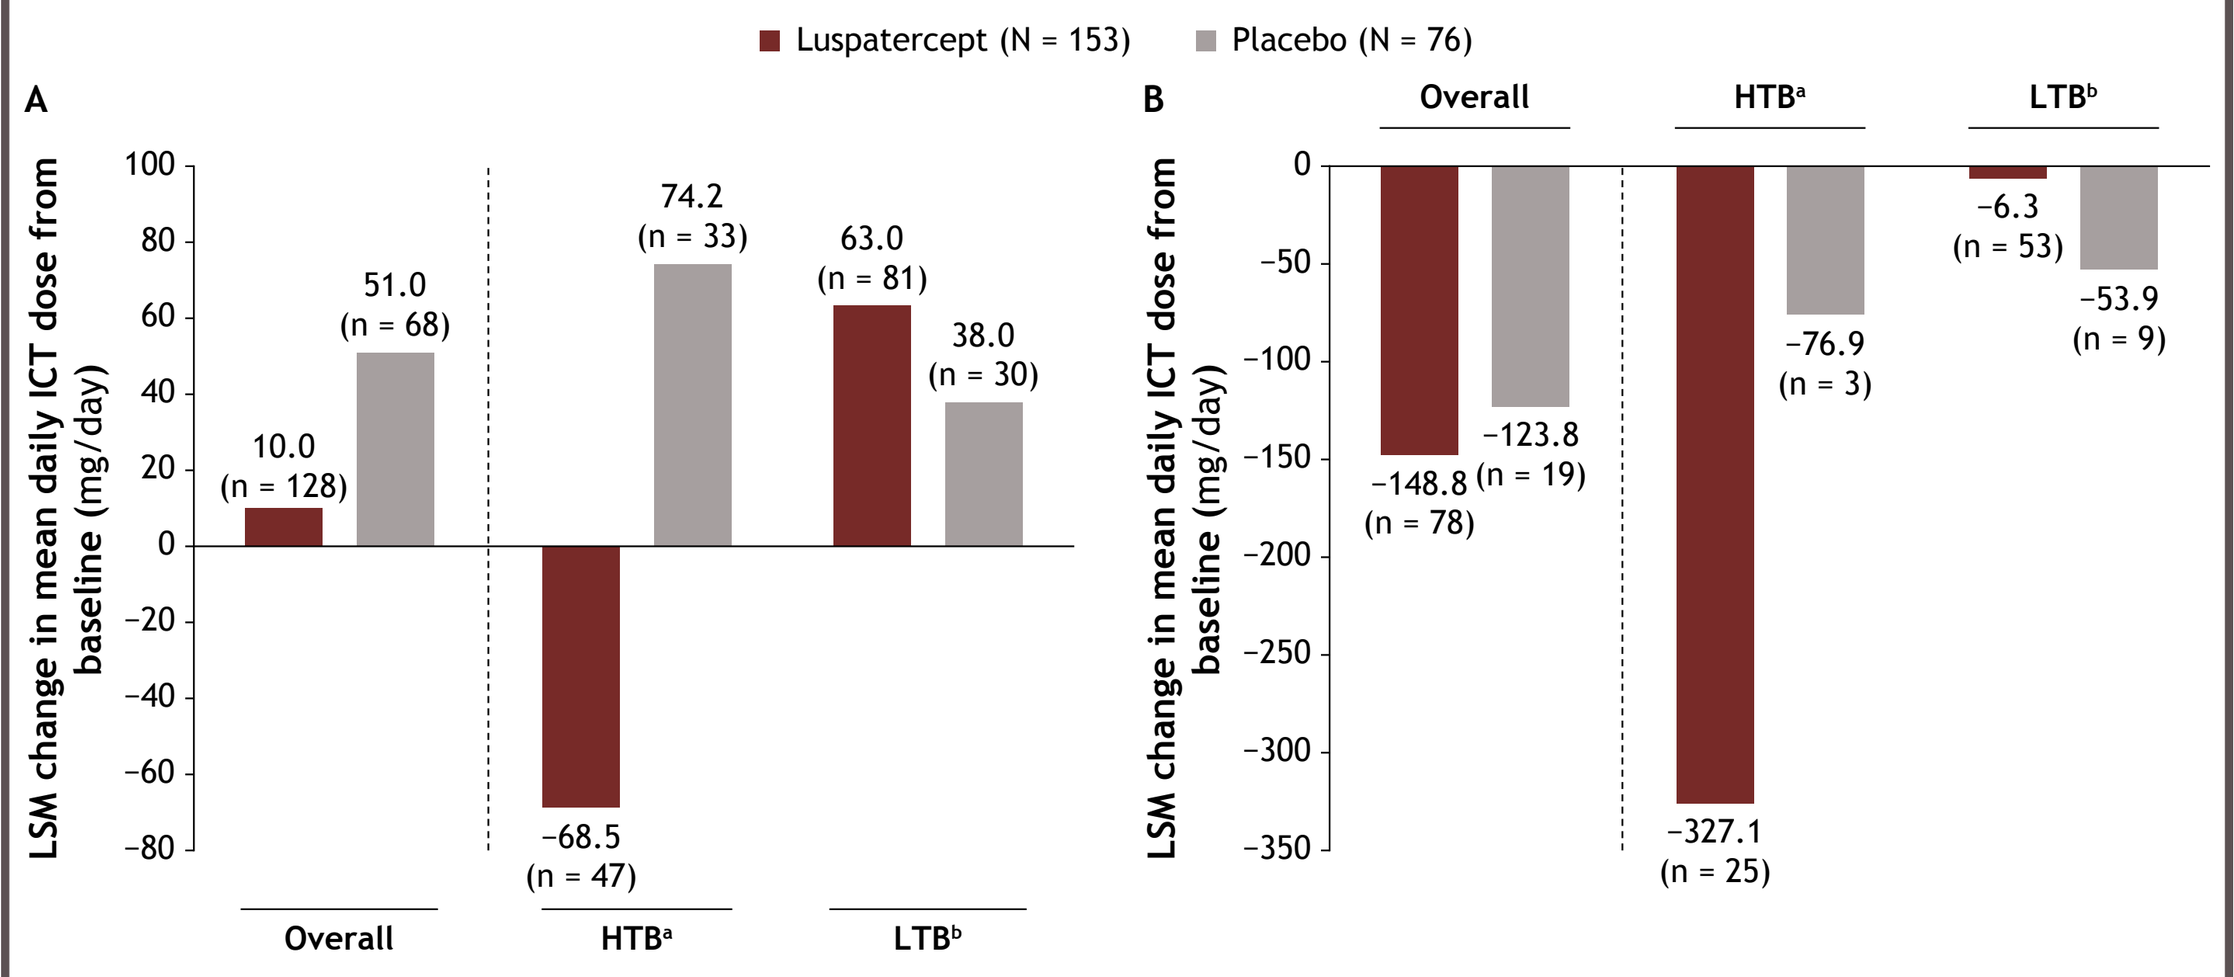
Luspatercept (N = 153)
Placebo (N = 76)
A
B
LSM change in mean daily ICT dose from baseline (mg/day)
10.0 (n = 128)
51.0 (n = 68)
−68.5 (n = 47)
74.2 (n = 33)
63.0 (n = 81)
38.0 (n = 30)
−148.8 (n = 78)
−123.8 (n = 19)
−327.1 (n = 25)
−76.9 (n = 3)
−6.3 (n = 53)
−53.9 (n = 9)
Overall
HTB[a]
LTB[b]

ANCOVA was used to compare the treatment difference between groups, with the change in value as the dependent variable, treatment group (2 levels) as a factor, and baseline ICT value as covariates, stratifying by baseline IPSS-R (Very low or Low, vs Intermediate).
[a]HTB defined as patients with baseline transfusion burden of ≥ 6 RBC units/8 weeks. [b]LTB defined as patients with baseline transfusion burden of < 6 RBC units/8 weeks.
ANCOVA, analysis of covariance; HTB, high transfusion burden; ICT, iron chelation therapy; IPSS-R, Revised International Prognostic Scoring System; LSM, least squares mean; LTB, low transfusion burden; RBC, red blood cell.

Safety

- Of patients in the luspatercept arm who received ICT at baseline, 100% had ≥ 1 treatment-emergent adverse event (TEAE) (any grade) and 56.3% had ≥ 1 grade 3 or 4 TEAE (**Table 3**)
- Of patients in the luspatercept arm who did not receive ICT at baseline, 97.6% had ≥ 1 TEAE (any grade) and 53.7% had ≥ 1 grade 3 or 4 TEAE (**Table 3**)

Table 3. Safety summary by ICT use at baseline

Parameter, n/N (%)	Luspatercept (n = 153)	Placebo (n = 76)
Patients with ≥ 1 TEAE (any grade)		
Received ICT at baseline	71/71 (100.0)	35/40 (87.5)
Did not receive ICT at baseline	80/82 (97.6)	35/36 (97.2)
Patients with ≥ 1 TEAE (Grade 3 or 4)		
Received ICT at baseline	40/71 (56.3)	17/40 (42.5)
Did not receive ICT at baseline	44/82 (53.7)	17/36 (47.2)

TEAEs include adverse events that started on or after the date of first dose and on or before 42 days after the date of last dose of study treatment. If a patient experienced multiple events under the same System Organ Class and Preferred Term, then the patient is counted only once. Medical Dictionary for Regulatory Activities version 20.0 was used for coding. Common Terminology Criteria for Adverse Events version 4.0 was used.
ICT, iron chelation therapy; TEAE, treatment-emergent adverse event.

Conclusions

- Luspatercept treatment resulted in SF decreases, particularly in patients with higher baseline RBC transfusion burden and in those with high baseline SF who experienced a significant reduction in RBC transfusion burden
 - Luspatercept had a positive effect on serum ferritin regardless of HI-E response
 - This suggests that the effect of luspatercept on SF may be due not only to a reduction in RBC transfusion burden, but also potentially to a more direct effect of the drug on ineffective erythropoiesis and/or iron metabolism
- Overall, ICT use was reduced with luspatercept

References

1. Fenaux P, Rose C. *Blood Rev* 2009;23 (Suppl 1):S15-S19.
2. Greenberg PL. *J Natl Compr Canc Netw* 2006;4:91-96.
3. Suragani RN, et al. *Nat Med* 2014;20:408-414.
4. Fenaux P, et al. *N Engl J Med* 2020;382:140–151.
5. Reblozyl® (luspatercept-aamt) [prescribing information]. Summit, NJ, USA; Celgene Corporation. https://www.accessdata.fda.gov/drugsatfda_docs/label/2020/761136orig2lbl.pdf; Accessed May 21, 2020.
6. Cheson BD, et al. *Blood* 2006;108:419-425.

Acknowledgments

- This study was sponsored by Celgene, a Bristol-Myers Squibb Company in collaboration with Acceleron Pharma
- The patients and families who are making this study possible
- The clinical study teams who participated in the study and the protocol managers for this study
- All authors contributed to and approved the presentation; editorial assistance was provided by Emily Poulin, PhD, of Excerpta Medica, funded by Bristol-Myers Squibb Company

Disclosures

P.F.: AbbVie, Bristol Myers Squibb, Janssen, Jazz Pharmaceuticals – honoraria, research funding; Jazz Pharmaceuticals – travel accommodations. V.S.: Bristol Myers Squibb, Johnson & Johnson, Novartis – honoraria; Acceleron Pharma, Amgen, Bristol Myers Squibb, Menarini, Novartis – consulting or advisory role. G.J.M.: AbbVie, Novartis – consulting or advisory role; Bristol Myers Squibb, Novartis – research funding. M.D-C.: Bristol Myers Squibb, Novartis – consulting or advisory role, membership on an entity's board of directors or advisory committee, research funding. C.F.: Bristol Myers Squibb, Janssen, Novartis – consulting or advisory role, speakers bureau; Bristol Myers Squibb – research funding. M.A.S.: Millennium/Takeda – consulting or advisory role, honoraria, research funding. B.Q., O.B-R., G.G-M.: No conflicts of interest to disclose. R.S.K.: AbbVie, Agios, Bristol Myers Squibb, Daiichi Sankyo, Jazz Pharmaceuticals, Pfizer – consulting or advisory role; AbbVie, Agios, Jazz Pharmaceuticals – speakers bureau; AbbVie, Agios, Bristol Myers Squibb, Daiichi Sankyo, Jazz Pharmaceuticals, Pfizer – travel accommodations and expenses. R.B.: Bristol Myers Squibb – consulting or advisory role, honoraria, research funding; Otsuka – research funding. M.T.V.: Bristol Myers Squibb – research funding. D.S.: Novartis – consulting or advisory role, honoraria, speakers bureau. A.E.D.: AbbVie – travel accommodations; Bristol Myers Squibb – consulting or advisory role, research funding. P.L.G.: Amgen, Bristol Myers Squibb – stock and other ownership interests; Novartis – consulting or advisory role; Aprea Therapeutics, Bristol Myers Squibb, Genentech, H3 Biomedicine, Notable Labs, Novartis – research funding. A.M.Z.: AbbVie, Acceleron Pharma, Agios, Astellas Pharma, BeyondSpring Pharmaceuticals, Bristol Myers Squibb, Boehringer Ingelheim, Cardinal Health, Daiichi Sankyo, Epizyme, Incyte, Ionis Pharmaceuticals, Jazz Pharmaceuticals, Novartis, Otsuka, Pfizer, Seattle Genetics, Taiho Pharmaceutical, Takeda, Trovagene –consulting or advisory role, honoraria; AbbVie, ADC Therapeutics, Aprea Therapeutics, Astex Pharmaceuticals, AstraZeneca/MedImmune, Bristol Myers Squibb, Boehringer Ingelheim, Incyte, Novartis, Pfizer, Takeda, Trovagene – research funding; Bristol Myers Squibb, Novartis, Pfizer, Trovagene – travel accommodations and expenses. L.A.: Bristol Myers Squibb – research funding. A.V.: Stelexis Therapeutics – stock and other ownership interests; Acceleron Pharma, Bristol Myers Squibb, Stelexis Therapeutics – honoraria; Bristol Myers Squibb, Janssen Oncology – research funding.M.R.S.: AbbVie, Bristol Myers Squibb, Incyte, Karyopharm Therapeutics, Selvita, Takeda, TG Therapeutics – membership on an entity's board of directors or advisory board; Karyopharm Therapeutics – stock/other ownership; Karyopharm Therapeutics – consulting or advisory role; Incyte, Sunesis, Takeda, TG Therapeutics – research funding; Boehringer Ingelheim – intellectual property, patent, or royalties. A.L.: BMS – employment, stock and other ownership interests. R.I.: BMS – employment; Bayer – employment (immediate family member); BMS – stock and other ownership interests. J.Z.: BMS – employment, stock and other ownership interests. A.R.: BMS – employment, expenses, research funding, stock and other ownership interests, travel accommodations. D.S.: BMS – employment, stock and other ownership interests. J.M.: BMS – employment. C.L.: BMS – employment, stock or other ownership. P.G.L.: Acceleron Pharma – employment, stock and other ownership interests, patents, royalties, other intellectual property; FibroGen, Abbott, AbbVie – stock and other ownership interests. U.P.: AbbVie, Bristol Myers Squibb, Novartis – consultancy, honoraria.

Presented at the 25th Congress of the European Hematology Association (EHA), June 11–14, 2020.

http://www.globalbmsmedinfo.com

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from EHA® and the author of this poster.