Clinical benefit of luspatercept in patients with lower-risk myelodysplastic syndromes and high transfusion burden in the phase 3 MEDALIST study

Abstract Number
EP798

Amer M. Zeidan,[1] Guillermo Garcia-Manero,[2] Amy E. DeZern,[3] Pierre Fenaux,[4] Peter L. Greenberg,[5] Michael R. Savona,[6] Joseph G. Jurcic,[7] Amit Verma,[8] Ghulam J. Mufti,[9] Rena Buckstein,[10] Valeria Santini,[11] Abderrahmane Laadem,[12] Jennie Zhang,[12] Anita Rampersad,[12] Daniel Sinsimer,[12] Chrystal U. Louis,[12] Peter G. Linde,[13] Uwe Platzbecker,[14] Mikkael A. Sekeres[15]

[1]Department of Internal Medicine, Yale School of Medicine and Yale Cancer Center, Yale University, New Haven, CT, USA; [2]Department of Leukemia, University of Texas MD Anderson Cancer Center, Houston, TX, USA; [3]The Sidney Kimmel Comprehensive Cancer Center at Johns Hopkins, Baltimore, MD, USA; [4]Service d'Hématologie Séniors, Hôpital Saint-Louis, Université Paris 7, Paris, France; [5]Stanford University Cancer Center, Stanford, CA, USA; [6]Vanderbilt-Ingram Cancer Center, Vanderbilt University School of Medicine, Nashville, TN, USA; [7]Division of Hematology/Oncology, Herbert Irving Comprehensive Cancer Center, Columbia University Medical Center, New York, NY, USA; [8]Department of Oncology, Albert Einstein College of Medicine, Montefiore Medical Center, New York, NY, USA; [9]Department of Haemato-Oncology, King's College London, London, UK; [10]Odette Cancer Centre, Sunnybrook Health Sciences Centre, Toronto, ON, Canada; [11]MDS Unit, Azienda Ospedaliero Universitaria Careggi, University of Florence, Florence, Italy; [12]Bristol Myers Squibb, Princeton, NJ, USA; [13]Acceleron Pharma, Cambridge, MA, USA; [14]Medical Clinic and Policlinic 1, Hematology and Cellular Therapy, University Hospital Leipzig, Leipzig, Germany; [15]Department of Hematology and Medical Oncology, Cleveland Clinic, Cleveland, OH, USA

Scientific Content on Demand
To request a copy of this poster:
Scan QR code
via a barcode reader application.
This QR code will take you to an external site.
QR codes are valid for 120 days after the congress presentation date.

Introduction

- Patients with lower-risk myelodysplastic syndromes (LR MDS) develop anemia due to ineffective erythropoiesis, leading to dependence on red blood cell (RBC) transfusions[1,2]
- There is a need for effective therapies to reduce RBC transfusion burden, particularly for those patients who have significant transfusion needs[1,2]
- Luspatercept is a first-in-class erythroid maturation agent that binds to select transforming growth factor-β superfamily ligands to diminish Smad2/3 signaling and enhance late-stage erythropoiesis[3]
 - Luspatercept is indicated for the treatment of anemia in adult patients with LR MDS with ring sideroblasts (RS) who require regular RBC transfusions
- MEDALIST is a phase 3, randomized, double-blind, placebo-controlled study evaluating the efficacy and safety of luspatercept in patients with LR MDS with RS who require RBC transfusions[4]

Objective

- To evaluate the clinical benefit of luspatercept in patients with LR MDS with high transfusion burden (HTB) in the MEDALIST trial

Methods

Study design

- MEDALIST is an ongoing phase 3, randomized, double-blind, placebo-controlled trial (NCT02631070)
- Patients were randomized at 65 sites between March 2016 and June 2017
- Eligible patients were:
 - Aged ≥ 18 years
 - Had anemia due to Revised International Prognostic Scoring System-defined Very low-, Low-, or Intermediate-risk MDS with RS
 - Were refractory to, intolerant of, or unlikely to respond to (serum erythropoietin > 200 U/L) erythropoiesis-stimulating agents (ESAs)
 - Required RBC transfusions
- Patients were randomized 2:1 to receive luspatercept (starting dose 1.0 mg/kg, with titration up to 1.75 mg/kg in patients not achieving RBC transfusion independence [RBC-TI]) or placebo subcutaneously every 21 days for ≥ 24 weeks (**Figure 1**)
- Patients receiving transfusion of ≥ 6 RBC units within 8 weeks prior to study were classed as HTB; patients receiving < 6 RBC units were classed as low transfusion burden (LTB)

Study endpoints

- The primary endpoint was achievement of RBC-TI ≥ 8 weeks during Weeks 1–24
- The key secondary endpoint was achievement of RBC-TI ≥ 8 weeks during Weeks 1–48
- Additional secondary endpoints included:
 - ≥ 50% and ≥ 75% reduction in RBC units transfused from baseline over at least 24 weeks
 - Achievement of major hematologic improvement-erythroid (HI-E) (per International Working Group [IWG] 2006 criteria[5]) for ≥ 8 weeks, defined as a reduction of ≥ 4 RBC units from baseline (for patients with baseline RBC transfusion burden of ≥ 4 units/8 weeks) or a mean hemoglobin increase of ≥ 1.5 g/dL from baseline (for patients with baseline RBC transfusion burden of < 4 units/8 weeks)

Figure 1. MEDALIST trial study design

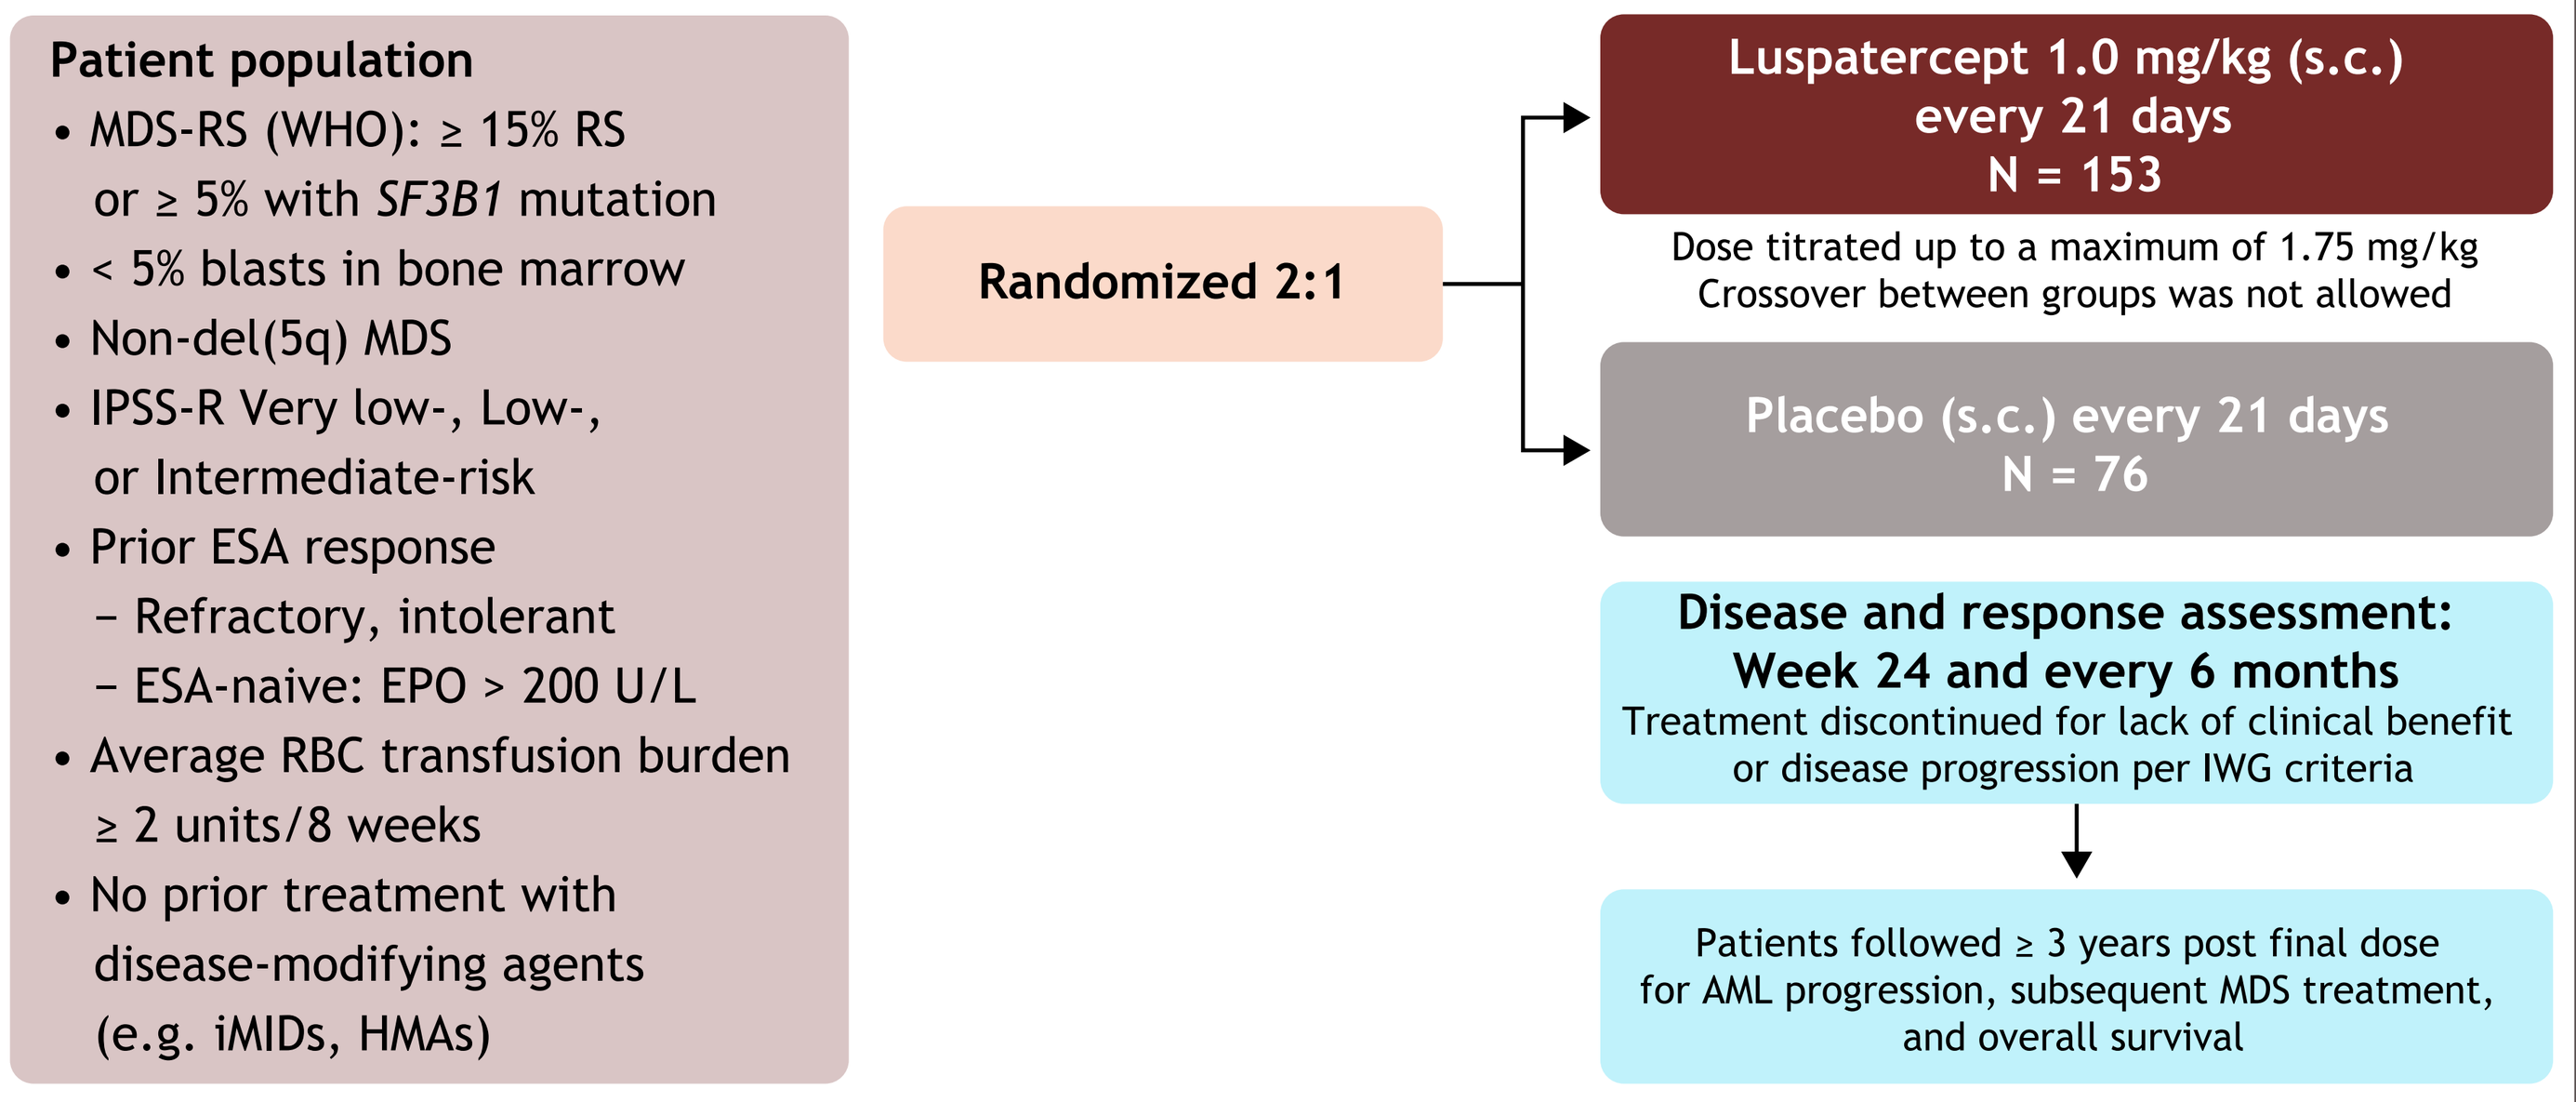

Patient population
• MDS-RS (WHO): ≥ 15% RS or ≥ 5% with *SF3B1* mutation
• < 5% blasts in bone marrow
• Non-del(5q) MDS
• IPSS-R Very low-, Low-, or Intermediate-risk
• Prior ESA response
– Refractory, intolerant
– ESA-naive: EPO > 200 U/L
• Average RBC transfusion burden ≥ 2 units/8 weeks
• No prior treatment with disease-modifying agents (e.g. iMIDs, HMAs)
Randomized 2:1
Luspatercept 1.0 mg/kg (s.c.) every 21 days N = 153
Dose titrated up to a maximum of 1.75 mg/kg
Crossover between groups was not allowed
Placebo (s.c.) every 21 days N = 76
Disease and response assessment: Week 24 and every 6 months
Treatment discontinued for lack of clinical benefit or disease progression per IWG criteria
Patients followed ≥ 3 years post final dose for AML progression, subsequent MDS treatment, and overall survival

Data cutoff: July 1, 2019. Includes last patient randomized + 48 weeks.
AML, acute myeloid leukemia; EPO, erythropoietin; ESA, erythropoiesis-stimulating agent; HMA, hypomethylating agent; iMID, immunomodulatory drug; IPSS-R, Revised International Prognostic Scoring System; IWG, International Working Group; MDS, myelodysplastic syndromes; RBC, red blood cell; RS, ring sideroblast; s.c., subcutaneously; *SF3B1*, splicing factor 3b subunit 1; WHO, World Health Organization.

Results

Patients

- 229 patients were randomized to receive luspatercept (N = 153) or placebo (N = 76) (**Table 1**)
 - 99 (43%) were HTB and 130 (57%) were LTB
- 148 (97%) and 70 (92%) patients in the luspatercept and placebo arms, respectively, had received prior ESAs
- The median duration of treatment received on study was 7.6 and 18.6 months for HTB and LTB patients receiving luspatercept, respectively, versus 5.5 and 5.5 months for patients receiving placebo
- The median (range) transfusion burden at baseline for HTB patients in the luspatercept and placebo arms was 7.5 (6-15) and 8.0 (6-20) RBC units over 8 weeks (in the 16 weeks prior to randomization), respectively
- 68.2% and 66.7% of HTB and LTB patients in the luspatercept arm, respectively, received 1.75 mg/kg as their maximum dose level
- As of data cutoff (July 1, 2019), 41 (26.8%) patients continue to receive luspatercept, whereas all patients receiving placebo discontinued at the end of the treatment phase

Table 1. Baseline characteristics

Characteristic	Luspatercept (N = 153)		Placebo (N = 76)	
	HTB (n = 66)	LTB (n = 87)	HTB (n = 33)	LTB (n = 43)
Age, median (range), years	71.5 (40-85)	71.0 (41-95)	71.0 (37-85)	74.0 (26-91)
Male, n (%)	53 (80.3)	41 (47.1)	23 (69.7)	27 (62.8)
RBC transfusion burden category, n (%)[a]				
≥ 6 units/8 weeks	66 (100)	0	33 (100)	0
≥ 4 to < 6 units/8 weeks	0	41 (47.1)	0	23 (53.5)
< 4 units/8 weeks	0	46 (52.9)	0	20 (46.5)
IPSS-R, n (%)				
Very low	10 (15.2)	8 (9.2)	4 (12.1)	2 (4.7)
Low	43 (65.2)	66 (75.9)	21 (63.6)	36 (83.7)
Intermediate	12 (18.2)	13 (14.9)	8 (24.2)	5 (11.6)
High	1 (1.5)	0	0	0
Baseline Hb, median (range), g/dL[b]	7.6 (6-10)	7.7 (6-10)	7.5 (5-9)	7.8 (6-9)
Baseline EPO level, mean, IU/L	331.94	239.45	407.70	189.98
***SF3B1* mutation, n (%)[c]**	60 (90.9)	81 (93.1)	30 (90.9)	35 (81.4)
Serum EPO, n (%)				
< 200 U/L	36 (54.5)	52 (59.8)	15 (45.5)	35 (81.4)
≥ 200 U/L	30 (45.5)	34 (39.1)	18 (54.5)	8 (18.6)
Missing	0	1 (1.1)	0	0
Prior ESA use, n (%)	63 (95.5)	85 (97.7)	29 (87.9)	41 (95.4)

[a]In the 16 weeks prior to randomization. [b]Baseline Hb was defined as the last value measured on or before the date and time of first dose. [c]Of patients with available baseline gene mutation data: n = 148 in the luspatercept arm; and n = 74 in the placebo arm; no patients with *SF3B1* mutation had RS < 15%. EPO, erythropoietin; ESA, erythropoiesis-stimulating agent; Hb, hemoglobin; HTB, high transfusion burden; IPSS-R, Revised International Prognostic Scoring System; LTB, low transfusion burden; RBC, red blood cell; RS, ring sideroblast; *SF3B1*, splicing factor 3b subunit 1.

Achievement of HI-E per IWG 2006 criteria

- In Weeks 1-48, 37 of 66 (56.1%) HTB patients in the luspatercept arm and 9 of 33 (27.3%) HTB patients in the placebo arm achieved HI-E response per IWG 2006 criteria (P = 0.0071) (**Figure 2**)

Achievement of RBC-TI

- 6 of 66 (9.1%) luspatercept-treated HTB patients and 1 of 33 (3.0%) placebo-treated HTB patients achieved the primary endpoint of RBC-TI ≥ 8 weeks in Weeks 1–24 (P = 0.27)
 - Median time to achieve RBC-TI in HTB patients was 50.0 days (range 1.0-100.0) with luspatercept versus 1.0 day for the 1 patient who achieved RBC-TI with placebo
 - Median duration of RBC-TI in HTB patients was 53.9 weeks (range 8.4-137.1) with luspatercept versus 54.9 weeks for the 1 patient who achieved RBC-TI with placebo
- 12 of 66 (18.2%) luspatercept-treated HTB patients and 2 of 33 (6.1%) placebo-treated HTB patients achieved RBC-TI ≥ 8 weeks in Weeks 1–48 (**Figure 3**)
 - In the placebo arm, as of the data cutoff, all patients had discontinued treatment
- 14 of 66 (21.2%) luspatercept- and 2 of 33 (6.1%) placebo-treated HTB patients achieved RBC-TI ≥ 8 weeks at any point during the entire assessment period

Reduction in transfusion burden

- 23 of 66 (34.8%) and 12 of 66 (18.2%) HTB patients receiving luspatercept achieved ≥ 50% and ≥ 75% reduction in RBC transfusion burden from baseline over at least 24 weeks, respectively (**Figure 4**)
 - Among patients receiving luspatercept, the most common dose level at which a ≥ 50% reduction in RBC transfusion burden over at least 24 weeks was first achieved was 1.0 mg/kg
 - Mean time to first achievement of a ≥ 50% reduction in RBC transfusion burden over at least 24 weeks was 2.7 months for the 23 luspatercept-treated HTB responders and 1.5 months for the 3 placebo-treated HTB responders

Figure 2. Patients achieving HI-E per IWG 2006 criteria during Weeks 1-48

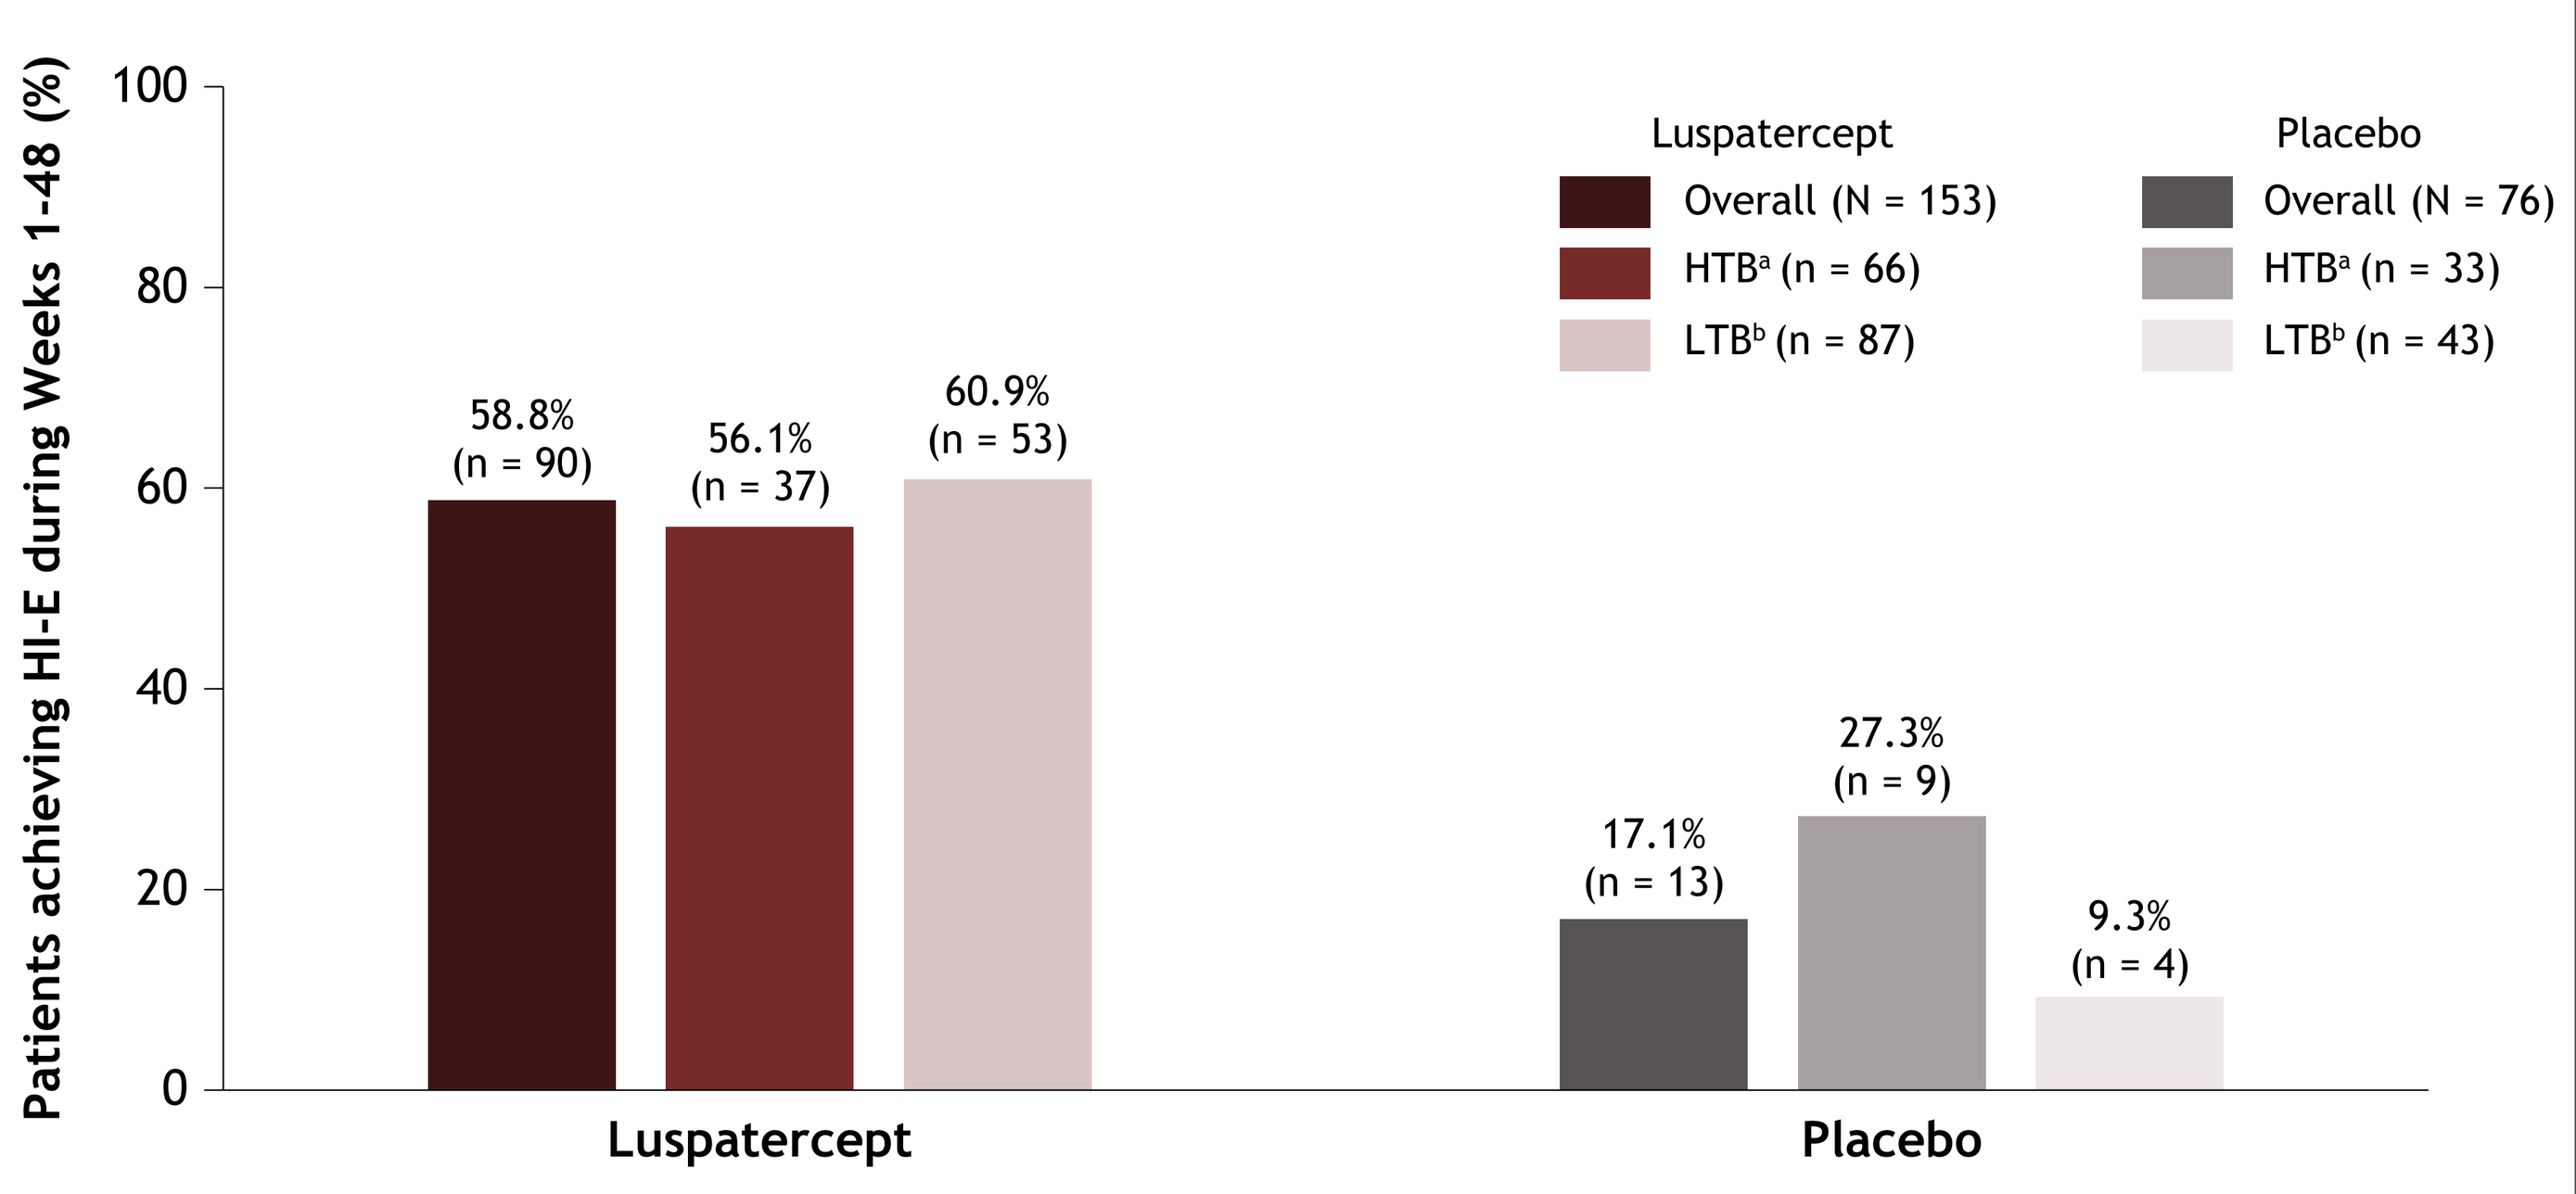

Patients achieving HI-E during Weeks 1-48 (%)
100
80
60
40
20
0
Luspatercept
Overall (N = 153)
HTB[a] (n = 66)
LTB[b] (n = 87)
Placebo
Overall (N = 76)
HTB[a] (n = 33)
LTB[b] (n = 43)
58.8% (n = 90)
56.1% (n = 37)
60.9% (n = 53)
17.1% (n = 13)
27.3% (n = 9)
9.3% (n = 4)
Luspatercept
Placebo

[a]HTB defined as patients receiving transfusion of ≥ 6 RBC units within 8 weeks prior to study. [b]LTB defined as patients receiving < 6 RBC units within 8 weeks prior to study. HI-E, hematologic improvement-erythroid; HTB, high transfusion burden; IWG, International Working Group; LTB, low transfusion burden; RBC, red blood cell.

Figure 3. Patients achieving RBC-TI ≥ 8 weeks during Weeks 1-48

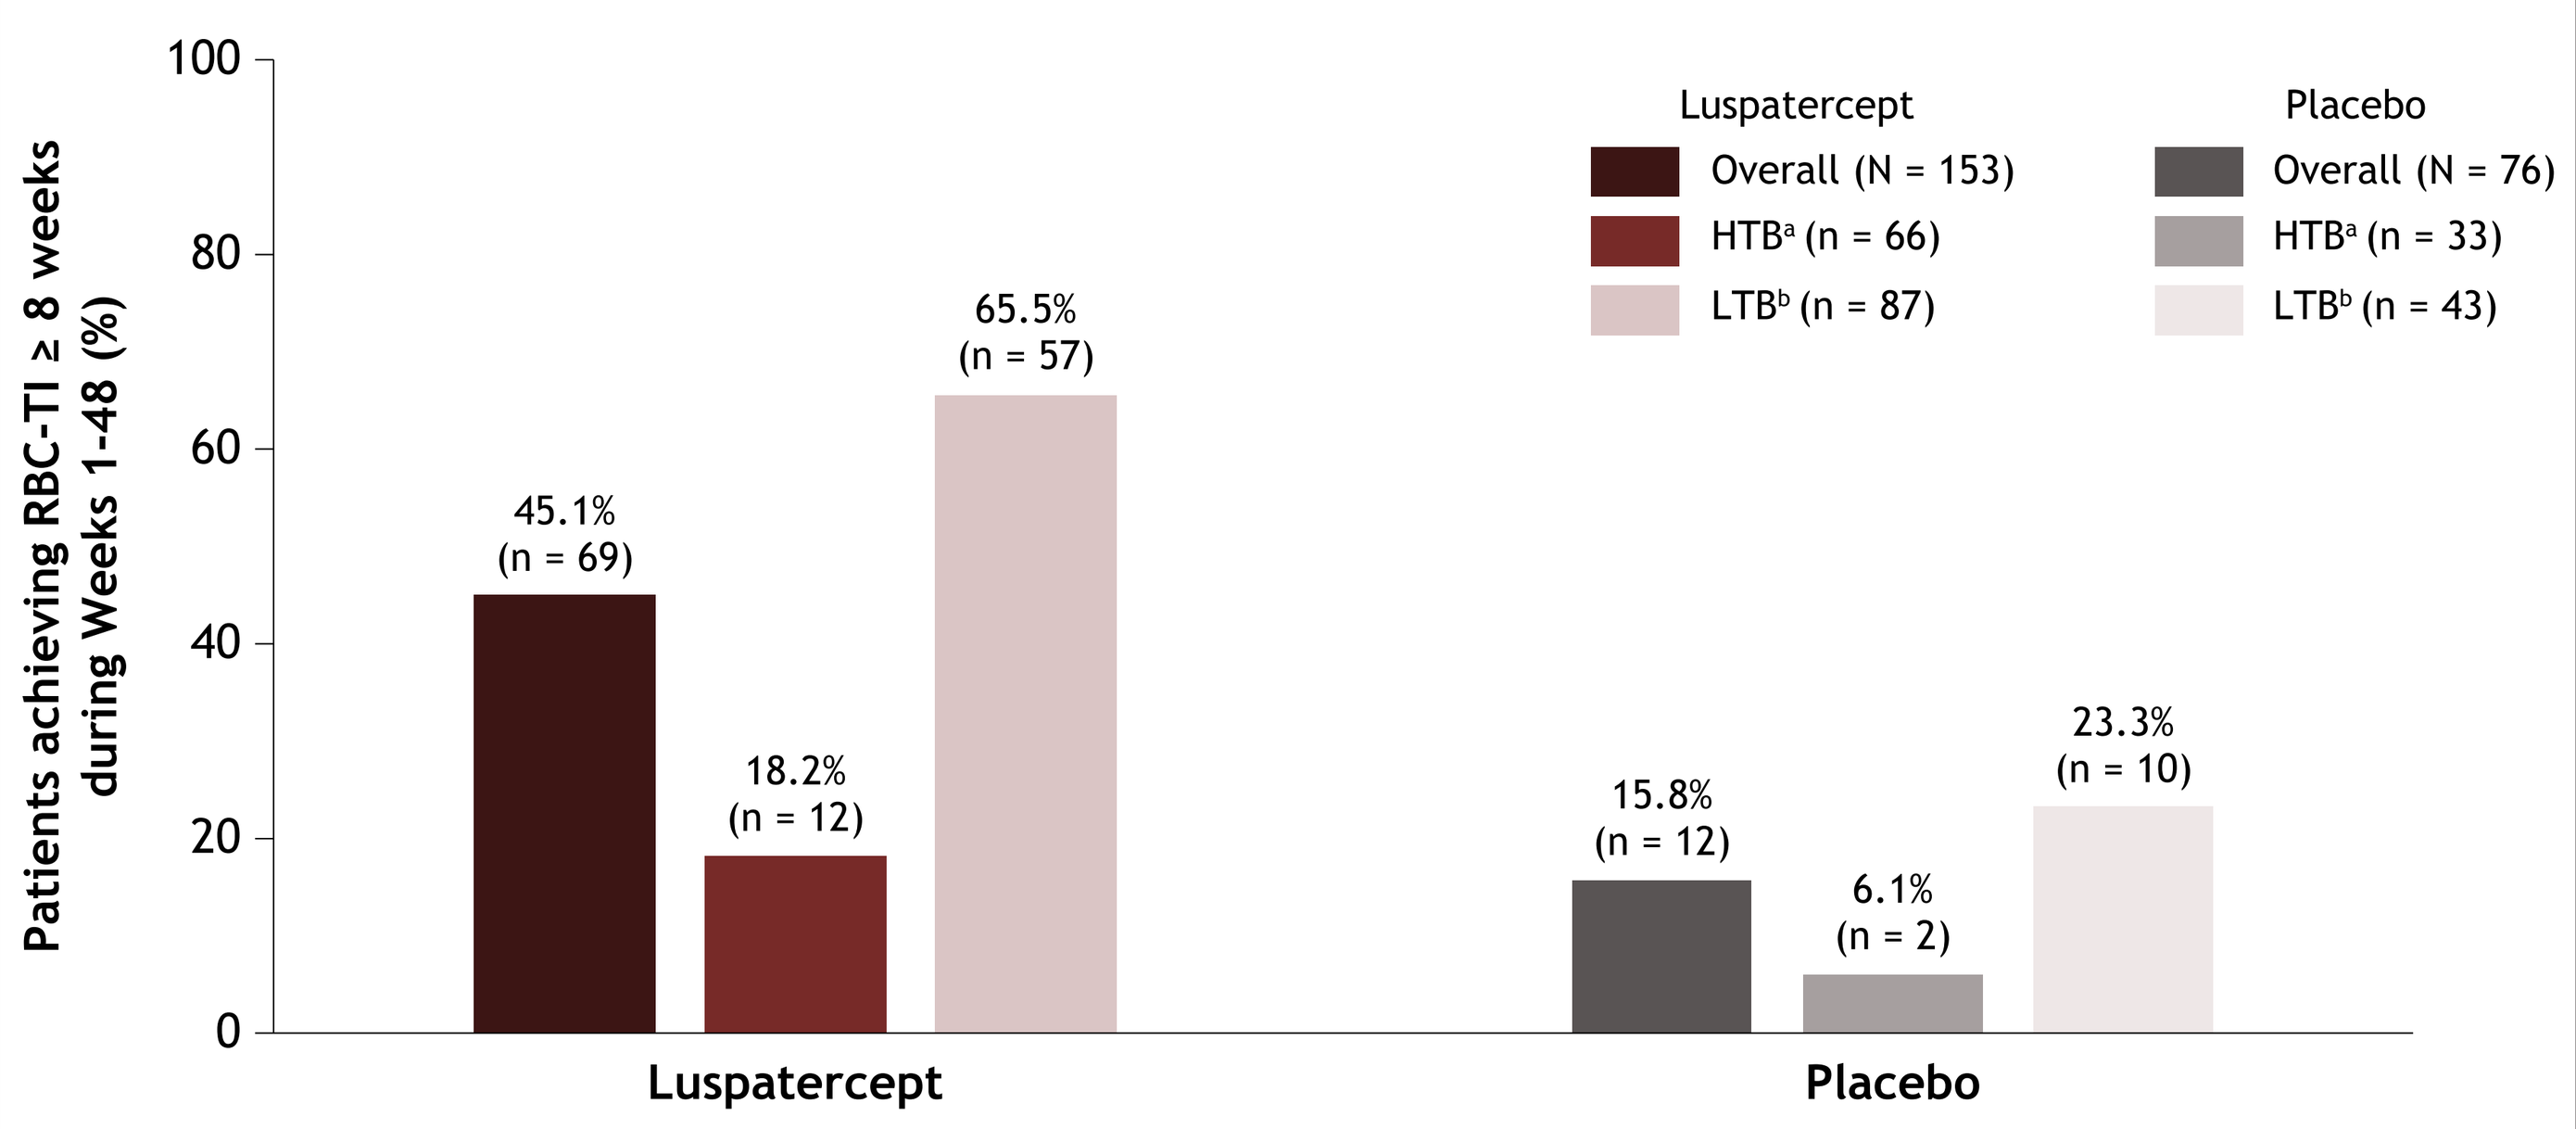

Patients achieving RBC-TI ≥ 8 weeks during Weeks 1-48 (%)
100
80
60
40
20
0
Luspatercept
Overall (N = 153)
HTB[a] (n = 66)
LTB[b] (n = 87)
Placebo
Overall (N = 76)
HTB[a] (n = 33)
LTB[b] (n = 43)
45.1% (n = 69)
18.2% (n = 12)
65.5% (n = 57)
15.8% (n = 12)
6.1% (n = 2)
23.3% (n = 10)
Luspatercept
Placebo

[a]HTB defined as patients receiving transfusion of ≥ 6 RBC units within 8 weeks prior to study. [b]LTB defined as patients receiving < 6 RBC units within 8 weeks prior to study. HTB, high transfusion burden; LTB, low transfusion burden; RBC-TI, red blood cell transfusion independence.

- Kaplan-Meier estimate of median longest duration of ≥ 50% reduction in RBC transfusion burden over at least 24 weeks was 72.3 weeks (95% confidence interval [CI] 29.6-not estimable [NE]) for the 23 luspatercept-treated HTB responders and NE (95% CI 27.7-NE) for the 3 placebo-treated HTB responders
- The mean number of transfusion events for HTB patients in Weeks 1–24 was 9.2 and 12.4 in the luspatercept and placebo arms, respectively (hazard ratio 0.794, 95% CI 0.660–0.956)

Safety

- The most common treatment-emergent adverse events (TEAEs) of any grade with luspatercept were fatigue (30.1% of patients), diarrhea (28.1%), and asthenia (25.5%)
- 11 of 66 (16.7%) HTB patients receiving luspatercept and 3 of 33 (9.1%) patients receiving placebo reported ≥ 1 TEAE leading to discontinuation
- 35 of 66 (53.0%) HTB patients receiving luspatercept and 18 of 33 (54.5%) patients receiving placebo reported ≥ 1 grade 3–4 TEAE
- 28 of 66 (42.4%) luspatercept-treated HTB patients and 15 of 33 (45.5%) placebo-treated patients reported ≥ 1 serious TEAE
 - 3 (4.5%) patients receiving luspatercept reported ≥ 1 suspected-related serious TEAE
- Of all patients, 3 of 153 (2.0%) receiving luspatercept progressed to acute myeloid leukemia (AML) versus 2 of 76 (2.6%) receiving placebo
 - Of those patients receiving luspatercept who progressed to AML, 2 had HTB
 - Of those receiving placebo who progressed to AML, 0 had HTB
 - Time from MDS diagnosis to AML progression ranged from 57.2-223.6 months in the 3 patients receiving luspatercept versus 30.1-32.7 months in the 2 patients receiving placebo

Figure 4. Patients achieving ≥ 50% (A) and ≥ 75% (B) reduction in RBC transfusion burden from baseline over at least 24 weeks

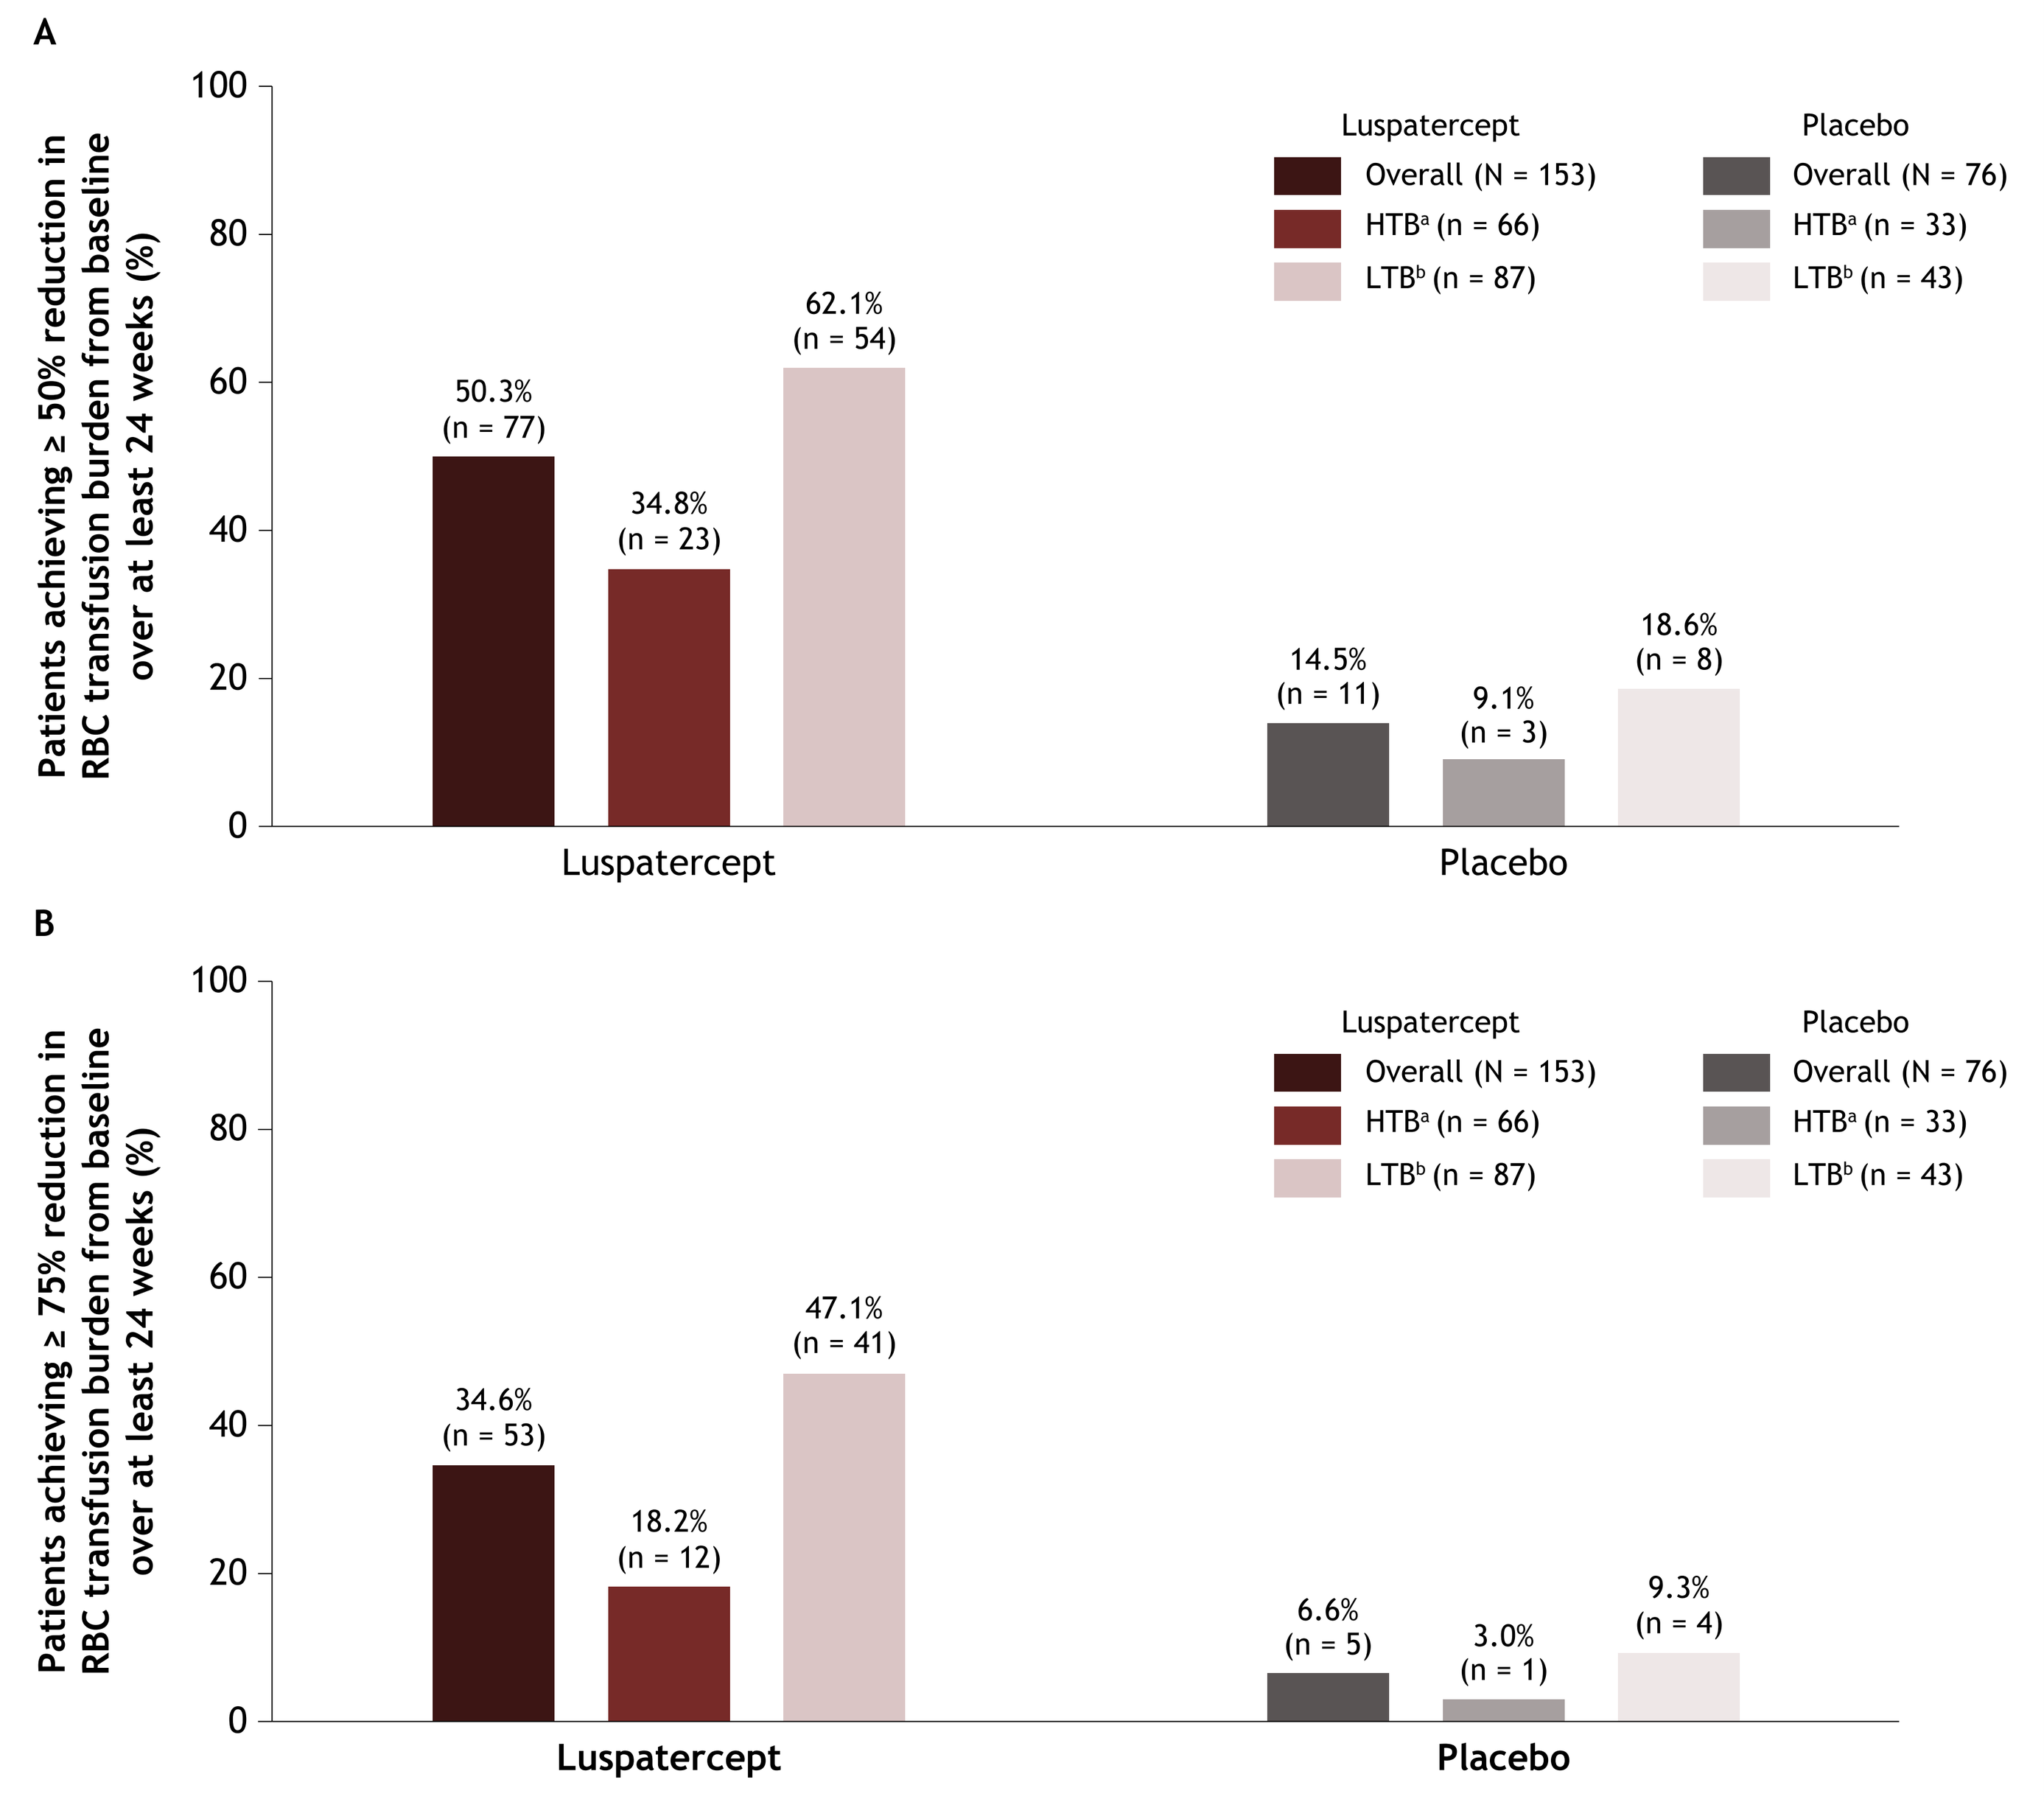

A
Patients achieving ≥ 50% reduction in RBC transfusion burden from baseline over at least 24 weeks (%)
100
80
60
40
20
0
Luspatercept
Overall (N = 153)
HTB[a] (n = 66)
LTB[b] (n = 87)
Placebo
Overall (N = 76)
HTB[a] (n = 33)
LTB[b] (n = 43)
50.3% (n = 77)
34.8% (n = 23)
62.1% (n = 54)
14.5% (n = 11)
9.1% (n = 3)
18.6% (n = 8)
Luspatercept
Placebo
B
Patients achieving ≥ 75% reduction in RBC transfusion burden from baseline over at least 24 weeks (%)
100
80
60
40
20
0
Luspatercept
Overall (N = 153)
HTB[a] (n = 66)
LTB[b] (n = 87)
Placebo
Overall (N = 76)
HTB[a] (n = 33)
LTB[b] (n = 43)
34.6% (n = 53)
18.2% (n = 12)
47.1% (n = 41)
6.6% (n = 5)
3.0% (n = 1)
9.3% (n = 4)
Luspatercept
Placebo

[a]HTB defined as patients receiving transfusion of ≥ 6 RBC units within 8 weeks prior to study. [b]LTB defined as patients receiving < 6 RBC units within 8 weeks prior to study. HTB, high transfusion burden; LTB, low transfusion burden; RBC, red blood cell.

Conclusions

- In this study of patients with LR MDS with RS, luspatercept treatment resulted in clinically significant reductions in RBC transfusion burden and achievement of HI-E, including in HTB patients
- Although rates of RBC-TI ≥ 8 weeks were low in HTB patients receiving luspatercept, there were reductions in number of transfusion events, and HTB patients had longer periods of transfusion burden reduction

References

1. Adès L, et al. *Lancet* 2014;383:2239-2252.
2. Fenaux P, Adès L. *Blood* 2013;121:4280-4286.
3. Suragani RN, et al. *Nat Med* 2014;20:408-414.
4. Fenaux P, et al. *N Engl J Med* 2020;382:140-151.
5. Cheson BD, et al. *Blood* 2006;108:419-425.

Acknowledgments

- The study was supported by Celgene, a Bristol-Myers Squibb Company in collaboration with Acceleron Pharma
- All authors contributed to and approved the presentation; writing and editorial assistance were provided by Daniel Gilmartin, PhD, of Excerpta Medica, funded by Bristol-Myers Squibb Company

Disclosures

A.M.Z.: AbbVie, Otsuka, Pfizer, BMS, Jazz Pharmaceuticals, Incyte, Agios, Boehringer Ingelheim, Novartis, Acceleron Pharma, Astellas Pharma, Daiichi Sankyo, Cardinal Health, Taiho Pharmaceutical, Seattle Genetics, BeyondSpring Pharmaceuticals, Trovagene, Takeda, Ionis Pharmaceuticals, Epizyme – honoraria, consulting or advisory role; BMS, AbbVie, Astex Pharmaceuticals, Pfizer, AstraZeneca/MedImmune, Boehringer Ingelheim, Trovagene, Incyte, Takeda, Novartis, Aprea Therapeutics, ADC Therapeutics – research funding; Pfizer, Novartis, BMS, Trovagene – travel, accommodations, expenses. G.G-M.: BMS, Astex Pharmaceuticals, Acceleron Pharma, Jazz Pharmaceuticals, Helsinn Therapeutics – consulting or advisory role; BMS, Astex Pharmaceuticals, Acceleron Pharma, Helsinn Therapeutics, AbbVie – honoraria; BMS, Astex Pharmaceuticals, Amphivena Therapeutics, Helsinn Therapeutics, Novartis, AbbVie, Onconova Therapeutics, H3 Biomedicine, Merck – research funding. A.E.D.: Acceleron Pharma, Syros Pharmaceuticals, Otsuka US – consulting or advisory role. P.F.: BMS, Janssen, Jazz Pharmaceuticals, AbbVie – honoraria, consulting or advisory role; Jazz Pharmaceuticals – travel, accommodations, expenses. P.L.G.: BMS, Amgen – stock and other ownership interests; Novartis – consulting or advisory role; BMS, Novartis, H3 Biomedicine, Aprea Therapeutics, Notable Labs, Genentech – research funding. M.R.S.: Karyopharm Therapeutics, BMS, TG Therapeutics, Merck, Ryvu – consulting or advisory role; Boehringer Ingelheim – patents, royalties, other intellectual property (property license); Karyopharm Therapeutics – stock and other ownership interests; Sunesis Pharmaceuticals, TG Therapeutics, Astex Pharmaceuticals, Incyte, Takeda – research funding. J.G.J.: Novartis, AstraZeneca, Boston Biomedical, BMS, Daiichi Sankyo, AbbVie – consulting or advisory role; BMS, Astellas Pharma, Actinium Pharmaceuticals, Daiichi Sankyo, FORMA Therapeutics, Seattle Genetics, Kura, Syros Pharmaceuticals, Genentech, AbbVie – research funding. A.V.: Stelexis Therapeutics – stock and other ownership interests; Stelexis therapeutics, BMS, Acceleron Pharma – honoraria; Janssen Oncology, BMS – research funding. G.J.M.: Novartis, AbbVie – consulting or advisory role; Novartis, BMS – research funding. R.B.: BMS – honoraria, consulting or advisory role, research funding; Otsuka – research funding. V.S.: BMS, Johnson & Johnson, Novartis – honoraria; Acceleron Pharma, Amgen, BMS, Menarini, Novartis – consulting or advisory role. A.L., J.Z., D.S.: BMS – employment, stock and other ownership interests. A.R.: BMS – employment, travel, accommodations, expenses, and stock and other ownership interests. C.U.L.: BMS – employment, stock and other ownership interests, patents, royalties, other intellectual property; Cell Medica, Merrimack – patents, royalties, other intellectual property. P.G.L.: Acceleron Pharma – employment, stock and other ownership interests, patents, royalties, other intellectual property; FibroGen, Abbott, AbbVie – stock and other ownership interests. U.P.: AbbVie, BMS, Novartis – consultancy, honoraria. M.A.S.: BMS, Millennium, Syros Pharmaceuticals – consulting or advisory role; Takeda, Pfizer – research funding.

Presented at the 25th Congress of the European Hematology Association (EHA), June 11-14, 2020.

Contact: http://www.globalbmsmedinfo.com

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from EHA® and the author of this poster.